A Phase 2 Study of Onvansertib (PCM-075) in Combination with Abiraterone and Prednisone in Patients with Metastatic Castration-Resistant Prostate Cancer

Cancer Center

Beth Israel Deaconess Medical Center

HARVARD MEDICAL SCHOOL TEACHING HOSPITAL

David J. Einstein, MD[1]; Atish Choudhury, MD, PhD[2]; Philip Saylor, MD[3]; Lillian Werner, MS[2]; Mark Erlander, PhD[4]; Maya Ridinger, PhD[4]; Glenn Bubley, MD[1]

[1]Beth Israel Deaconess Medical Center, Boston, MA [2]Dana-Farber Cancer Institute, Boston, MA [3]Massachusetts General Hospital, Boston, MA [4]Trovagene, Inc., San Diego, CA

Background

- Polo-like kinase 1 (PLK1) is a serine-threonine kinase and is a master regulator of mitotic progression
 - PLK1 required for G2/M transition
 - Overexpressed in prostate cancer
 - AR and PLK1 regulate each other

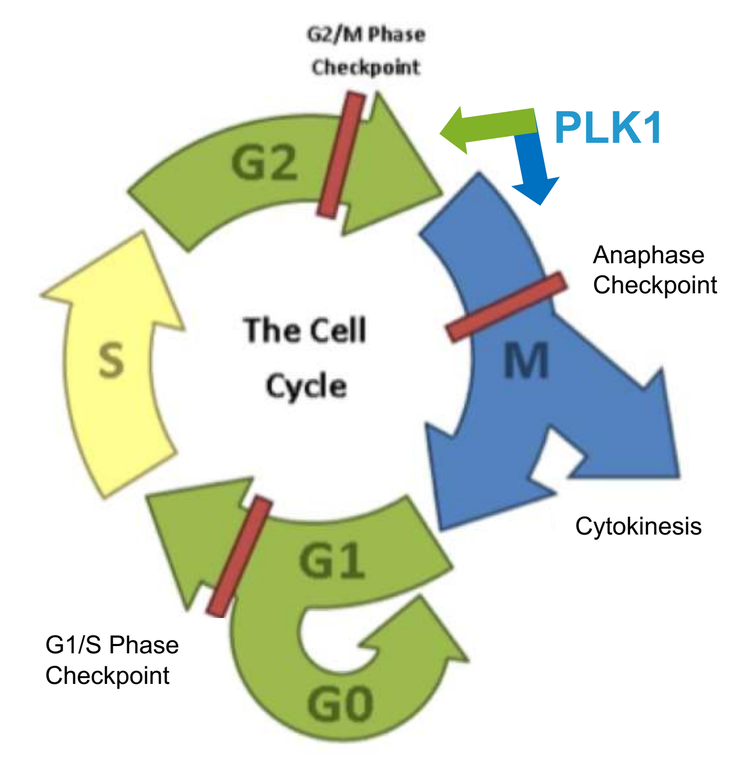
G2/M Phase Checkpoint
PLK1
G2
Anaphase Checkpoint
S
The Cell Cycle
M
Cytokinesis
G1
G1/S Phase Checkpoint
G0

- PLK1 synergy with abiraterone (Abi) identified in a novel in vivo castration-resistant prostate cancer (CRPC) assay

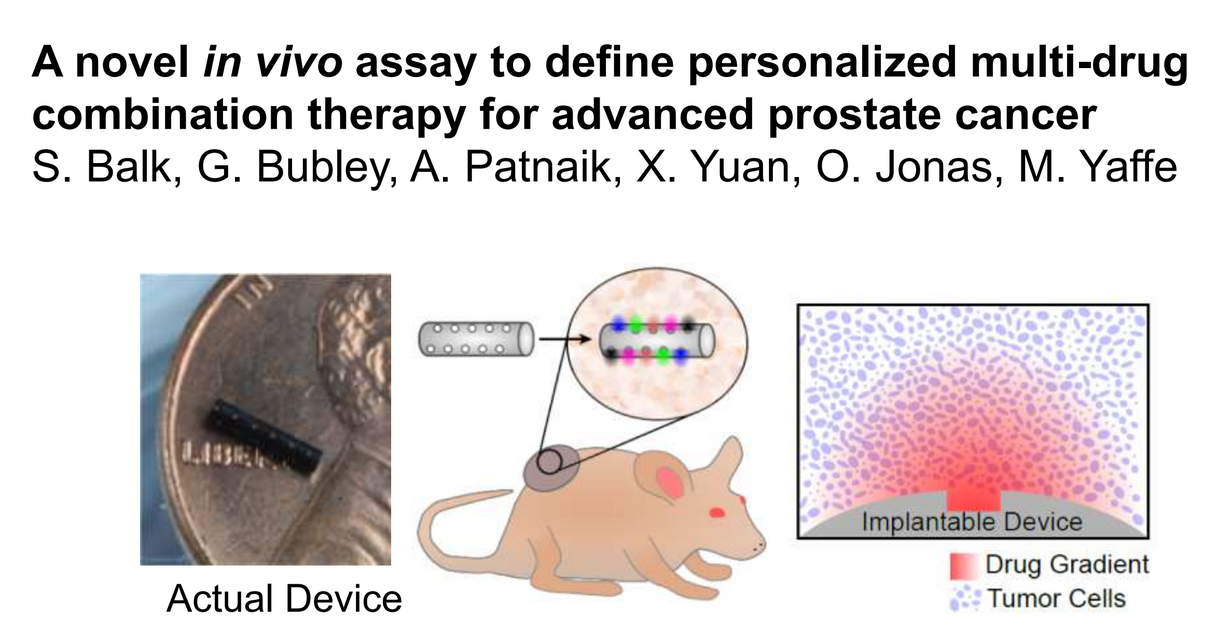
A novel *in vivo* assay to define personalized multi-drug combination therapy for advanced prostate cancer
S. Balk, G. Bubley, A. Patnaik, X. Yuan, O. Jonas, M. Yaffe
Actual Device
Implantable Device
Drug Gradient
Tumor Cells

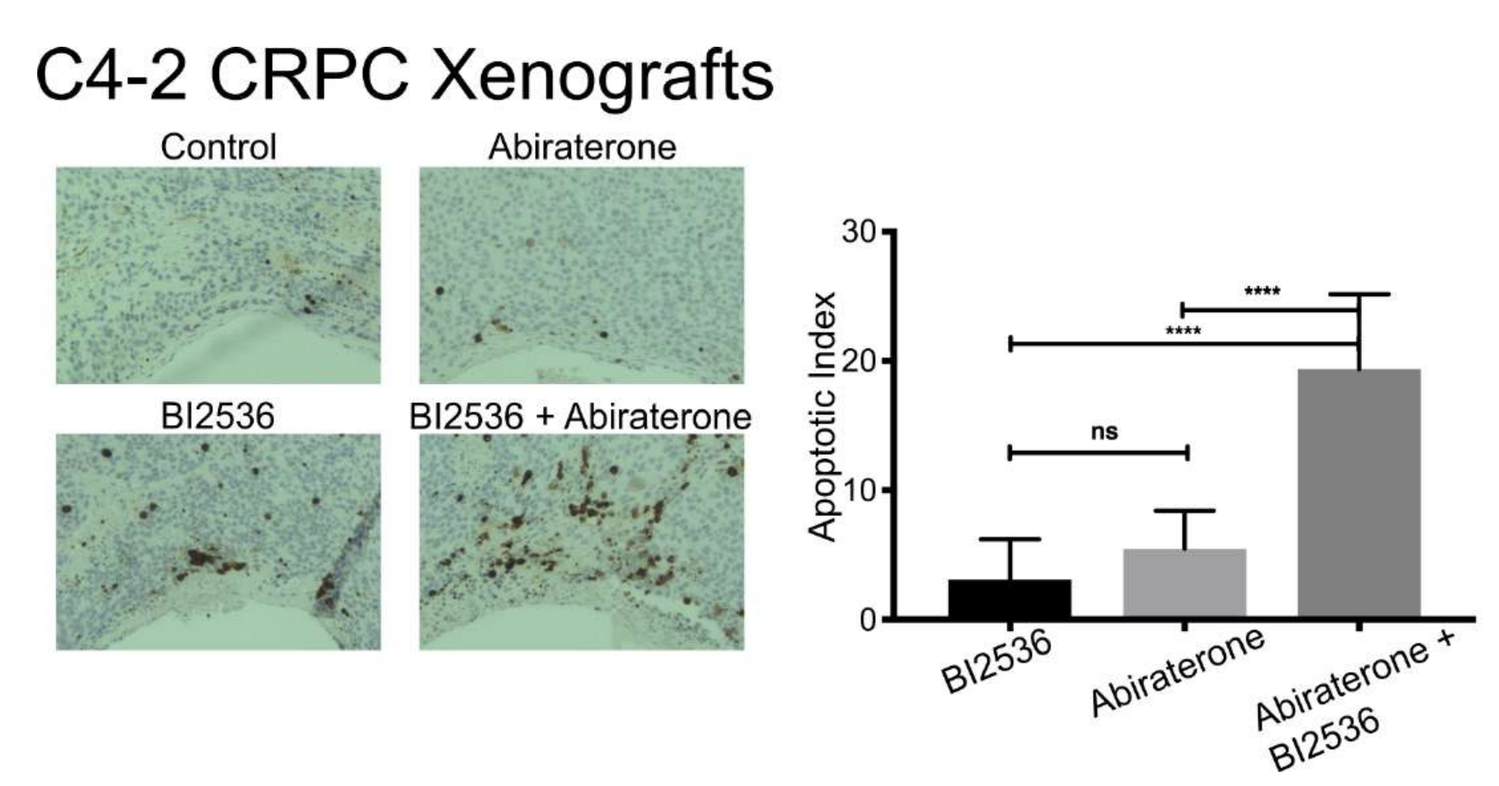
C4-2 CRPC Xenografts
Control
Abiraterone
BI2536
BI2536 + Abiraterone
Apoptotic Index
ns

BI2536
Abiraterone
Abiraterone + BI2536

- Mechanism may be AR-independent

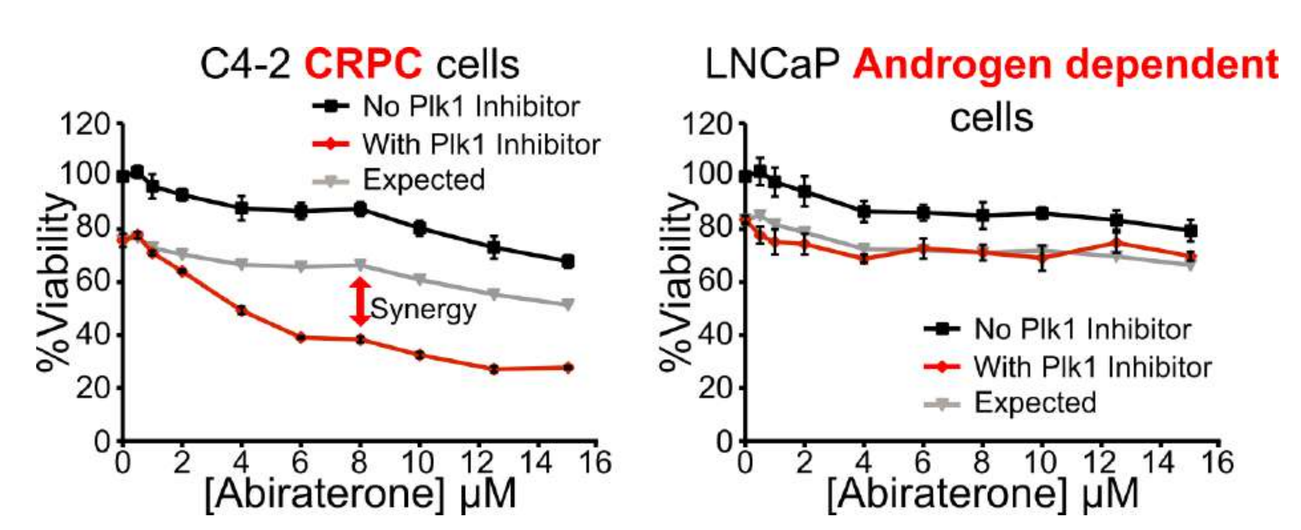
C4-2 CRPC cells
No Plk1 Inhibitor
With Plk1 Inhibitor
Expected
Synergy
%Viability
[Abiraterone] μM
LNCaP Androgen dependent cells
No Plk1 Inhibitor
With Plk1 Inhibitor
Expected
%Viability
[Abiraterone] μM

- Onvansertib (PCM-075) is a first-in-class selective inhibitor for PLK1
 - Orally available, short half-life (24 hours)
 - Primary toxicity in phase 1 testing was myelosuppression
 - Recommended phase 2 dose established

PLK Member	PCM-075 IC50 (nM)1
PLK1	2
PLK2	> 10,000
PLK3	> 10,000

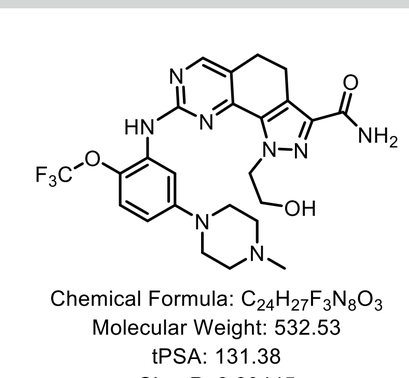
Chemical Formula: $C_{24}H_{27}F_3N_8O_3$
Molecular Weight: 532.53
tPSA: 131.38
CLogP: 2.29445

Objectives

Primary

- Observe effects of onvansertib in combination with Abi and prednisone on disease control rate (PSA decline or stabilization) in patients with mCRPC and early Abi resistance

Secondary

- Determine change in PSA after onvansertib addition relative to baseline
- Determine time to PSA progression and radiographic progression after onvansertib addition
- Determine radiographic response after onvansertib addition
- Determine disease control rate in per-protocol analysis
- Assess safety/toxicity of onvansertib in combination with Abi

Key Eligibility Criteria

Inclusion:

- mCRPC on first-line Abi
 - Abi can have been started in castration-sensitive or castration-resistant setting
 - Prior Abi response
- Early Abi resistance
 - Two rising PSAs on Abi: one rise of ≥0.3 ng/mL and one confirmatory value not showing decline, separated by 1 week

Exclusion:

- Prior enzalutamide or apalutamide
- Rapidly progressive disease or significant symptoms related to disease progression

Accrual

mCRPC with rising PSA on Abi → Abi + Plk1i: 3-patient safety lead-in → Abi + Plk1i: 29-patient expansion cohort

Schema

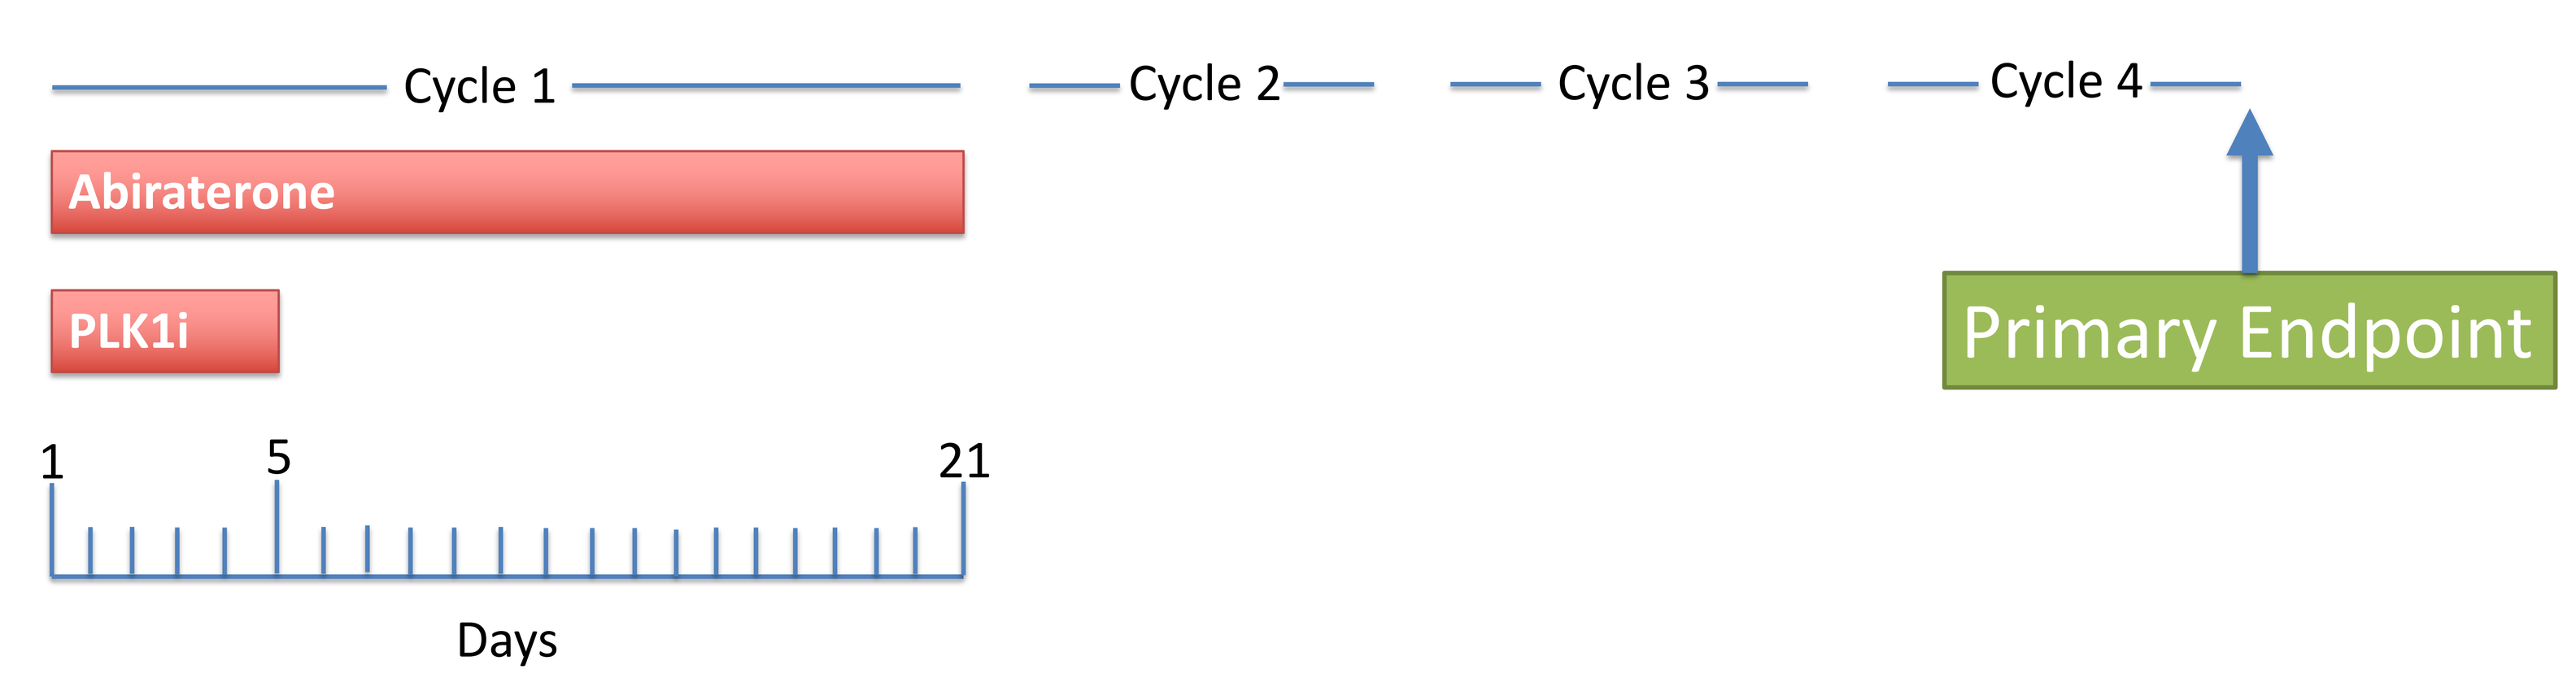
Cycle 1
Cycle 2
Cycle 3
Cycle 4
Abiraterone
PLK1i
Primary Endpoint
1
5
21
Days

Design

- Single-arm Simon's 2-stage design
- Power: 90% to detect improvement in disease control rate from 10% to 30%
- Futility analysis: early stopping if <2 out of the first 13 patients achieve disease control at 12 weeks

Enrollment

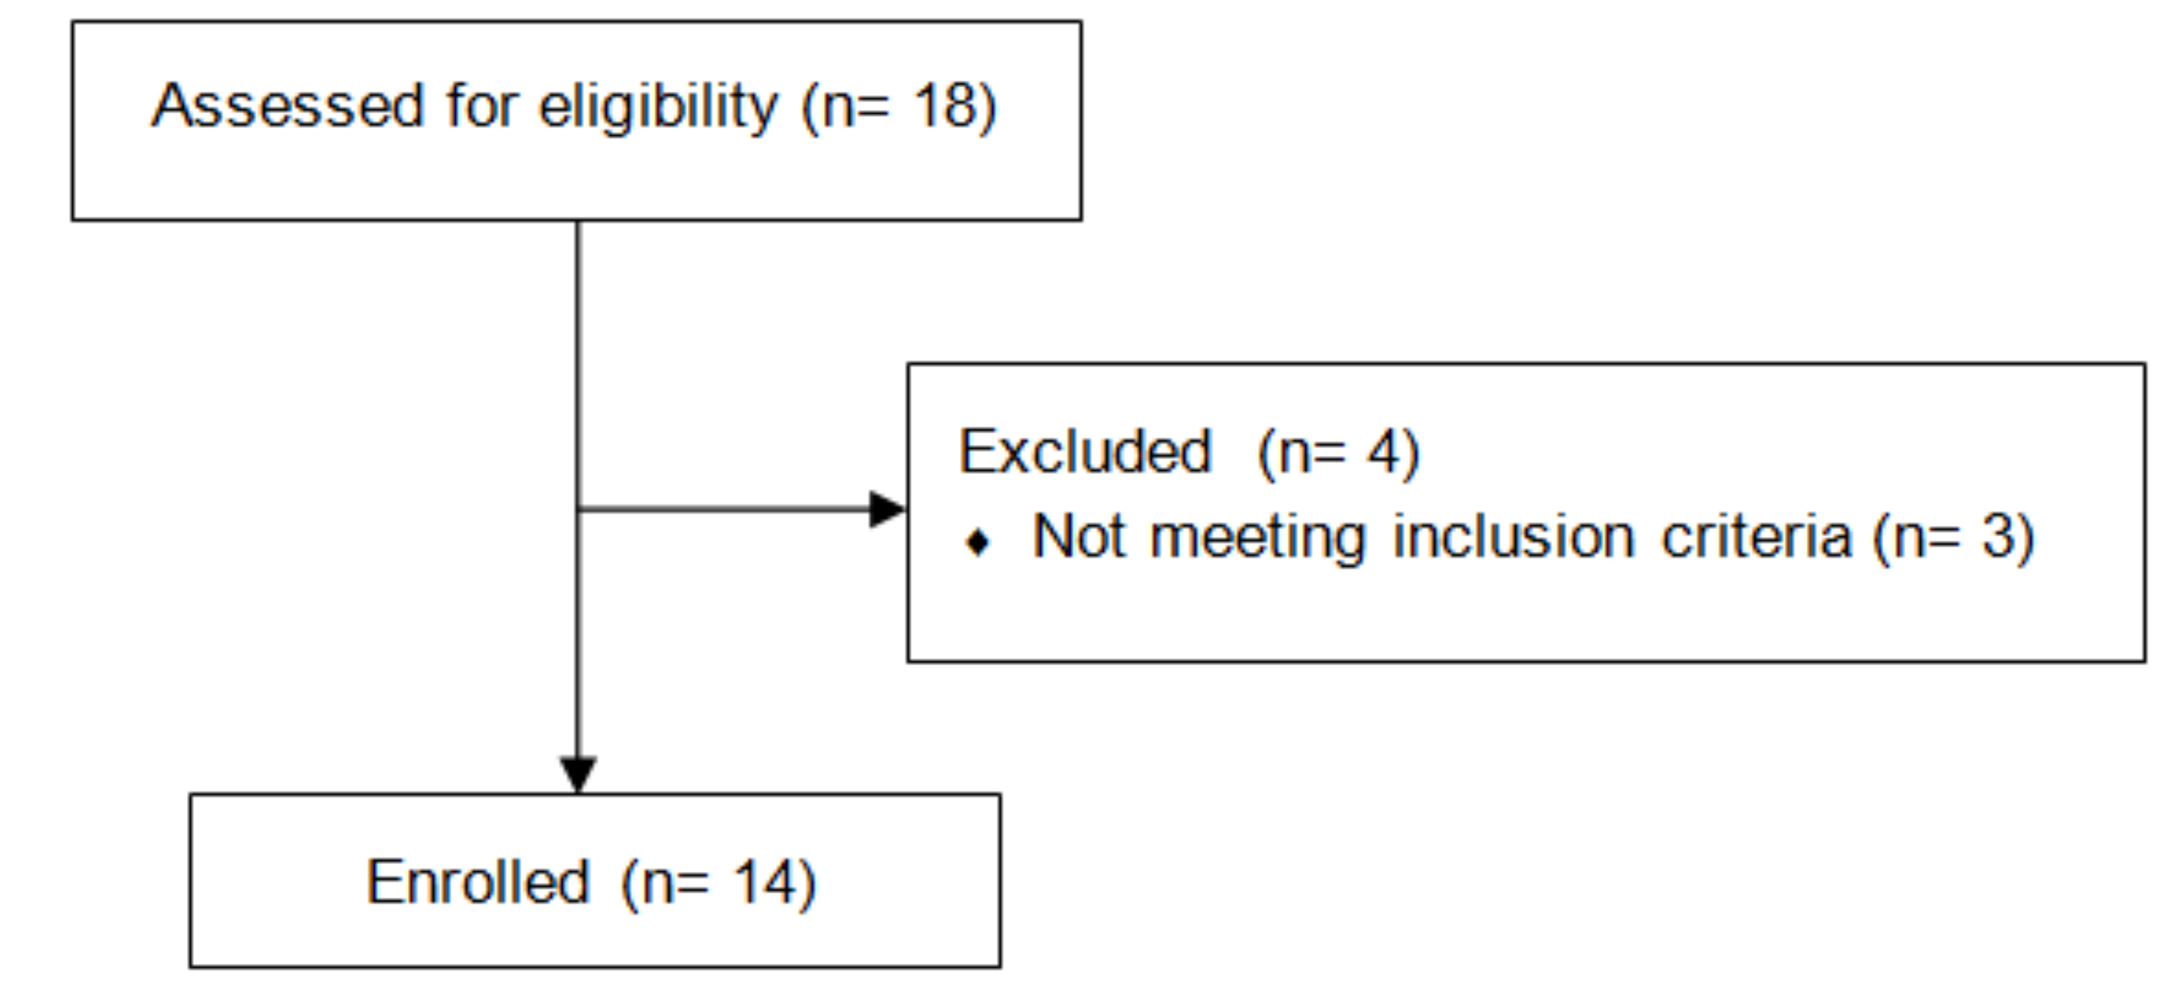
Assessed for eligibility (n= 18)
Excluded (n= 4)
Not meeting inclusion criteria (n= 3)
Enrolled (n= 14)

- Safety lead-in completed, no overlapping toxicities noted
- Now enrolling to expansion cohort

Correlative Objectives

1. Assess target inhibition of PLK1 in peripheral blood mononuclear cells (PBMCs) and circulating tumor cells (CTCs)
2. Evaluate potential biomarkers of response in CTCs and circulating tumor DNA (ctDNA)
 - Genomic alterations
 - Variant androgen receptor splicing (AR-V7)

Future Directions

Clinical Trial

- Plan to open a second arm using a dose-intensified schedule: onvansertib days 1-5 of a 14-day cycle.
 - ***Rationale:*** Transient PSA declines are seen in some patients, so goal is to maximize exposure to drug without causing drug accumulation

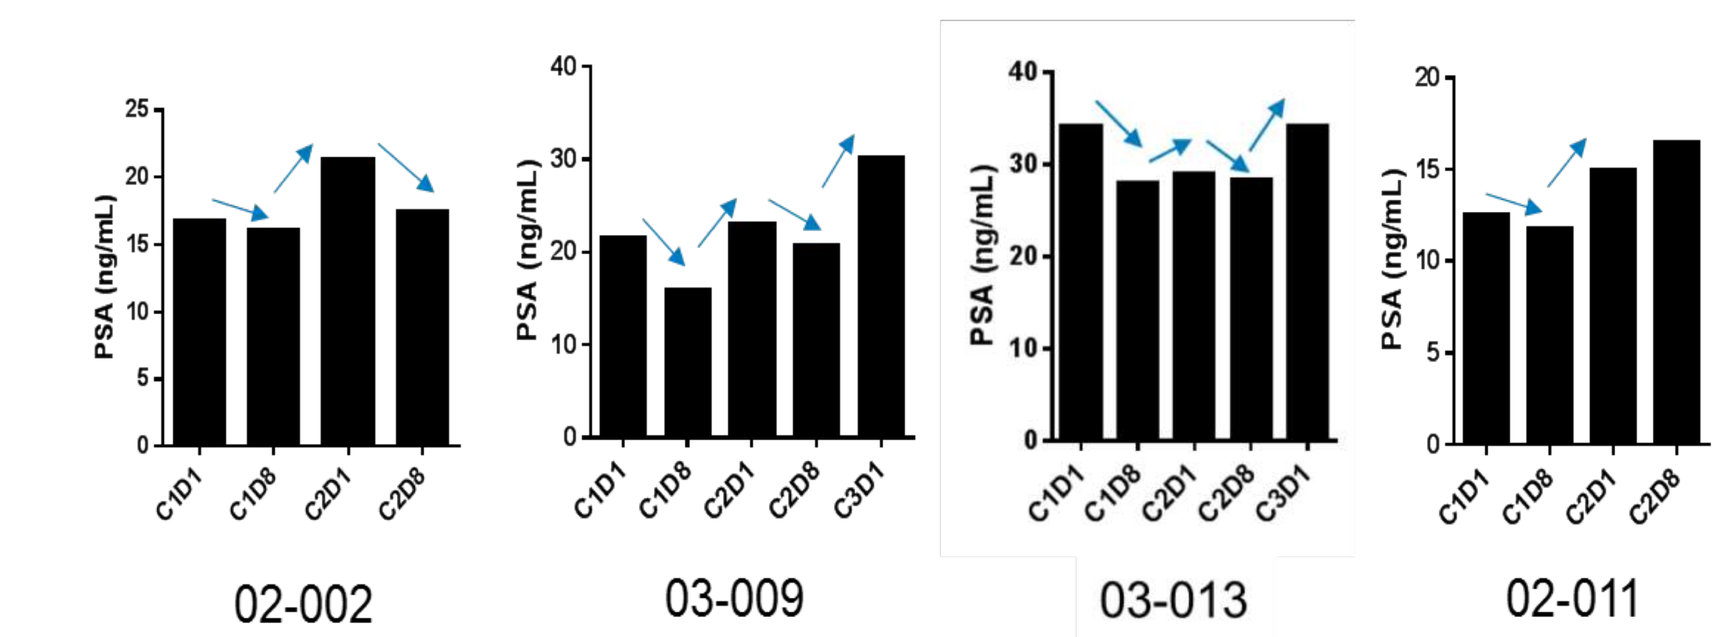
PSA (ng/mL)
C1D1
C1D8
C2D1
C2D8
C3D1
02-002
03-009
03-013
02-011

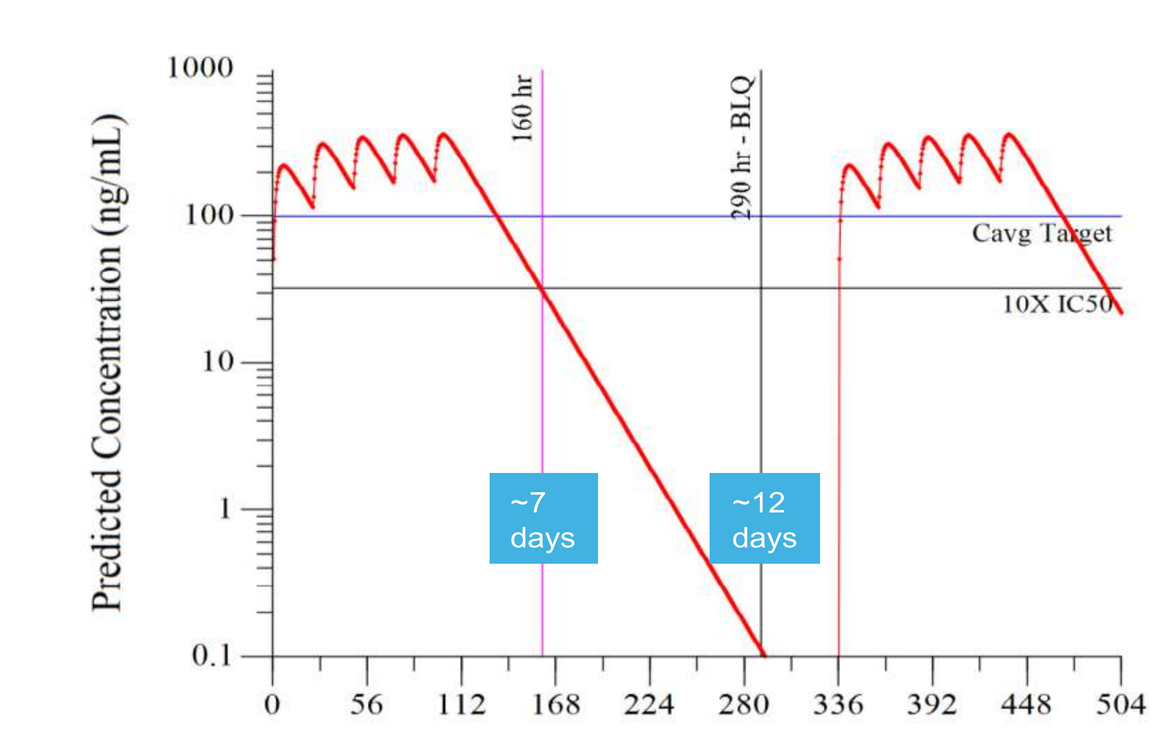
Predicted Concentration (ng/mL)
160 hr
290 hr - BLQ
Cavg Target
10X IC50
~7 days
~12 days
Time (h)

Exploratory Studies

- Co-clinical trial in PDX models to assess predictive biomarkers of synergy between PLK1 inhibition and abiraterone
- Identification of mechanisms of PLK1 synergy to expand utility of PLK1 combination therapy to other cancer types

NCT03414034
Abstract: TPS336

THE BRIDGE PROJECT
KOCH INSTITUTE FOR INTEGRATIVE CANCER RESEARCH AT MIT

Funding: Trovagene, Koch Institute Bridge Grant

deinstei@bidmc.harvard.edu @david.j.einstein