Polo-like Kinase 1 inhibitor Onvansertib Synergizes with Paclitaxel in Breast Cancer Carrying p53 Mutation

Abstract # 5818

Antonio Giordano, MD[1;] Yueying Liu[1;] Christiana Kappler[2]; Yeonhee Park[3]; Elizabeth Yeh[4]; Mark Erlander, PhD[5]; Stephen Ethier[2]

[1]Department of Medicine, Division of Hematology & Oncology, Medical University of South Carolina, Charleston SC; [2]Department of Pathology and Laboratory Medicine, Medical University of South Carolina, Charleston SC;
[3]Department of Public Health Sciences, Hollings Cancer Center, Medical University of South Carolina, Charleston SC; [4]Department of Cell and Molecular Pharmacology & Experimental Therapeutics, Medical University of South Carolina, Charleston SC;
[5]Trovagene Oncology, San Diego CA

MUSC Health
HOLLINGS CANCER CENTER
A National Cancer Institute Designated Cancer Center

Background

- Somatic mutation in TP53 gene (mutp53) is a strong prognostic marker in breast cancer
- Triple negative breast cancer (TNBC) is characterized by up to 80% mutp53 and the greatest overall genomic instability among subtypes
- Polo-like kinase 1 (PLK1) regulates progression of cells through the G2 phase of the cell cycle
- We hypothesize that mutp53 in the context of breast cancer can predict synergy to paclitaxel [p] plus onvansertib [o], an orally available highly selective PLK1 inhibitor

Methods

- **Growth Assay:** cells were plated in 6-well plates at a density of 100,000 cells per well and treated in triplicate with a wide range of drug concentrations or DMSO. After a 3-day exposure to drug, cell number was determined by using a Celigo Imaging Cytometer (Nexcelom, USA).
- **Immunoblotting, Apoptosis and DNA Content Analysis:** cells were synchronized with double thymidine block. For synchronization, SUM149 and SUM159, were treated with 2 mmol/L thymidine, incubated for 24 hours, then washed with PBS before adding fresh media. After an 18-hour release, cells were again treated with thymidine for an additional 24 hours of incubation. Finally, cells were released for 2 hours in fresh media and treated with GSK461364, GSK461364 + docetaxel at the IC50 concentrations and DMSO.
- **In Vivo:** five million SUM159 cells were injected in the mammary fat pad of NOD-scid-IL2 receptor gamma null female mice (Jackson Laboratory). When tumors reached a volume higher than 40 mm3, 35 mice were randomized to receive onvansertib 120 mg/kg day 1-2 every week by oral gavage (P.O.), paclitaxel 10 mg/kg day 1 every week by intraperitoneal injection (I.P.), the combination of the two drugs at the same dose of single agents, or control vehicles. In each arm, P.O. and I.P. vehicle was given as per the control ar

Objectives

- Somatic mutation in TP53 gene (mutp53) is a strong prognostic marker in breast cancer. Triple negative breast cancer (TNBC) is characterized by up to 80% mutp53 and carry the worse prognosis among all subtypes of breast cancer
- In SUM149, a basal-like 2 (BL2) TNBC line isolated from an African American patient with Inflammatory Breast Cancer (IBC), Plk1 is a gene necessary for growth and survival
- Polo-like kinase 1 (PLK1) regulates progression of cells through the G2 phase of the cell cycle.
- In the early pre-clinical development of Plk1 targeted drugs, it was observed that cancer cells with mutp53 were more responsive and had lower IC50 than cell lines with wild type (wtp53)
- Our central hypothesis is that onvansertib will increase the sensitivity to nab-paclitaxel in metastatic breast cancer affecting the ability of cancer cells with mutp53 to progress through mitosis (**Fig. 1**)

Cell Lines with mutp53
1. SUM149 (TNBC)
2. SUM159 (TNBC)
3. SUM52 (Luminal)
4. T47D (Luminal)

Cell Lines with wtp53
1. SUM1315 (TNBC)
2. MCF7 (Luminal)
3. MCF10A (Normal Breast)

Drugs
1. Paclitaxel (microtubules)
2. Docetaxel (microtubules)
3. Onvansertib (PLK1 inhibitor)
4. GSK461364 (PLK1 inhibitor)

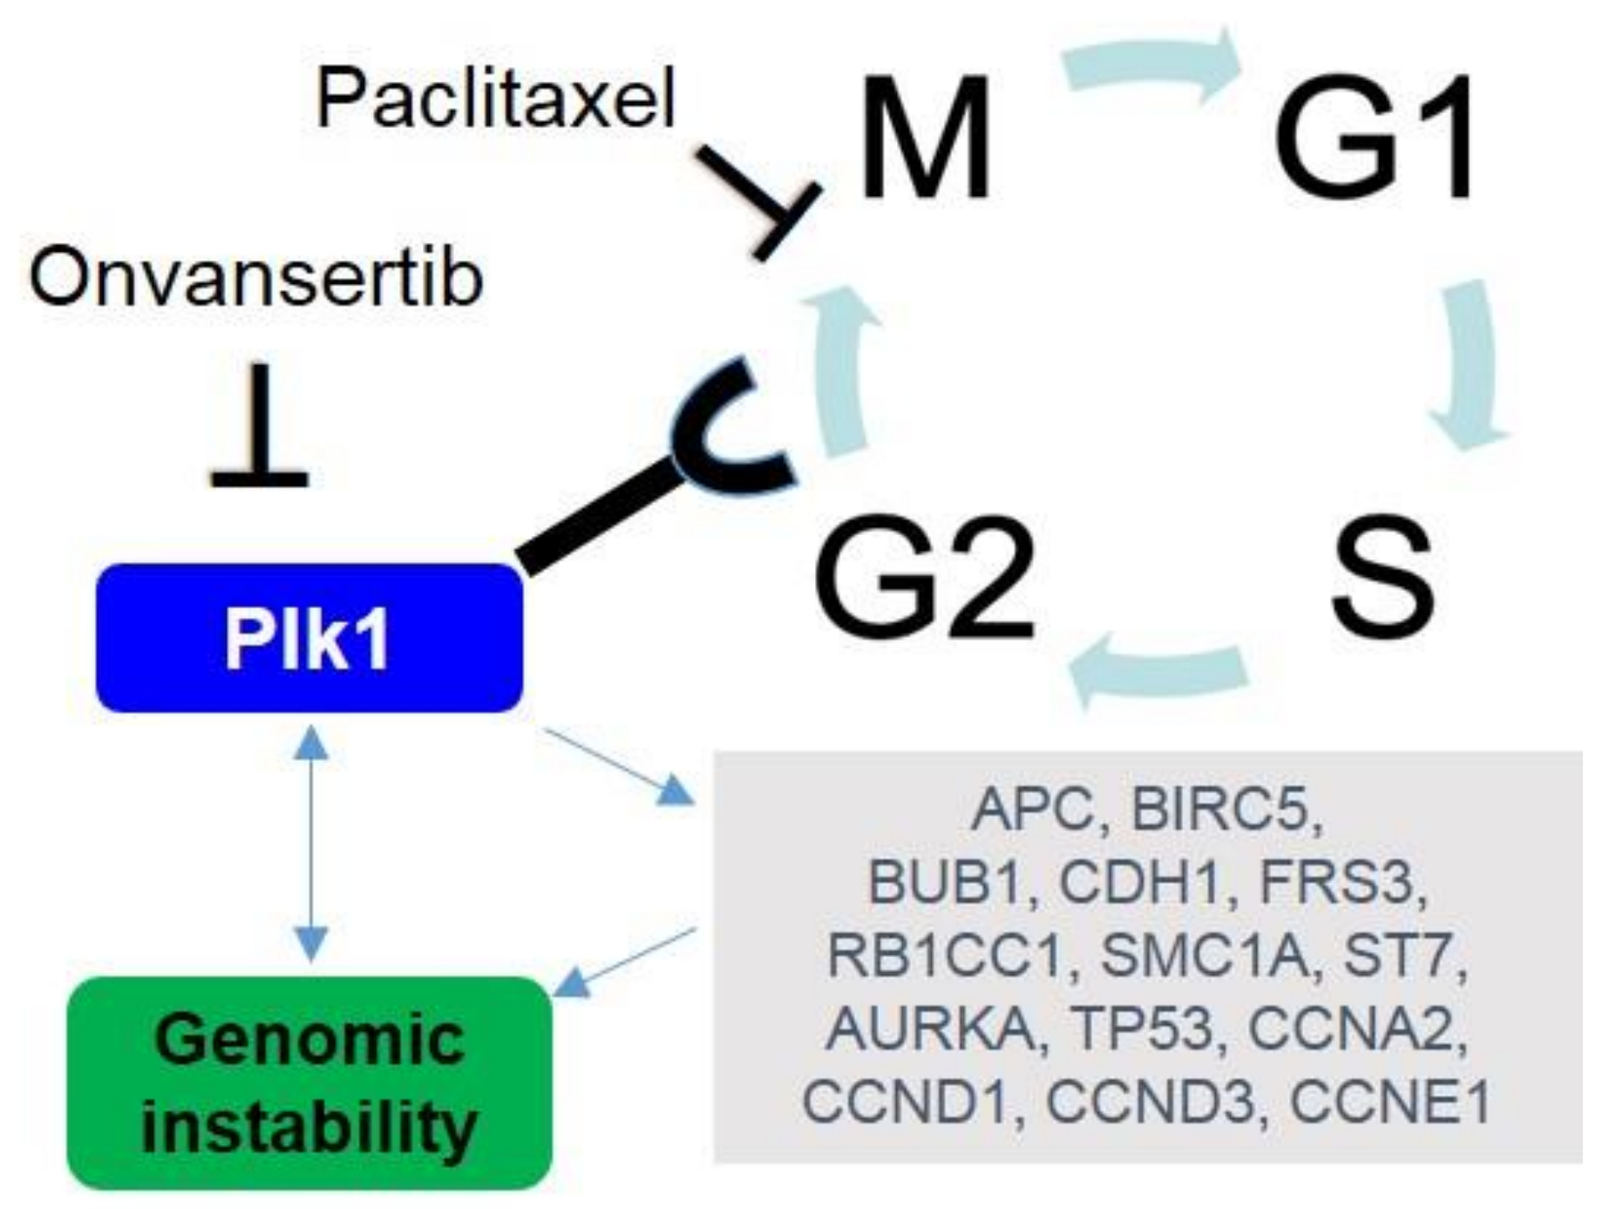
Paclitaxel
Onvansertib
M
G1
G2
S
Plk1
Genomic instability
APC, BIRC5, BUB1, CDH1, FRS3, RB1CC1, SMC1A, ST7, AURKA, TP53, CCNA2, CCND1, CCND3, CCNE1

Figure 1. Working hypothesis

- Cell lines undergo G2–M arrest following PLK1 inhibition. Agents like paclitaxel will synergize with PLK1i and induce apoptosis of cells in G2-M block
- The basal-like subtype, identified for the first time by Sorlie and colleagues, is characterized by up to 80% mutations in the TP53 gene (mutp53) and the greatest overall genomic instability among subtypes.
- At the moment, a targeted therapeutic approach for the treatment of breast cancer patients with TP53 mutation does not exist, and patients receive standard chemotherapy with anthracycline, taxane and/or platinum compounds.

Results

Figure 2. Cell Proliferation Assay of Single Drug Onvansertib and Paclitaxel

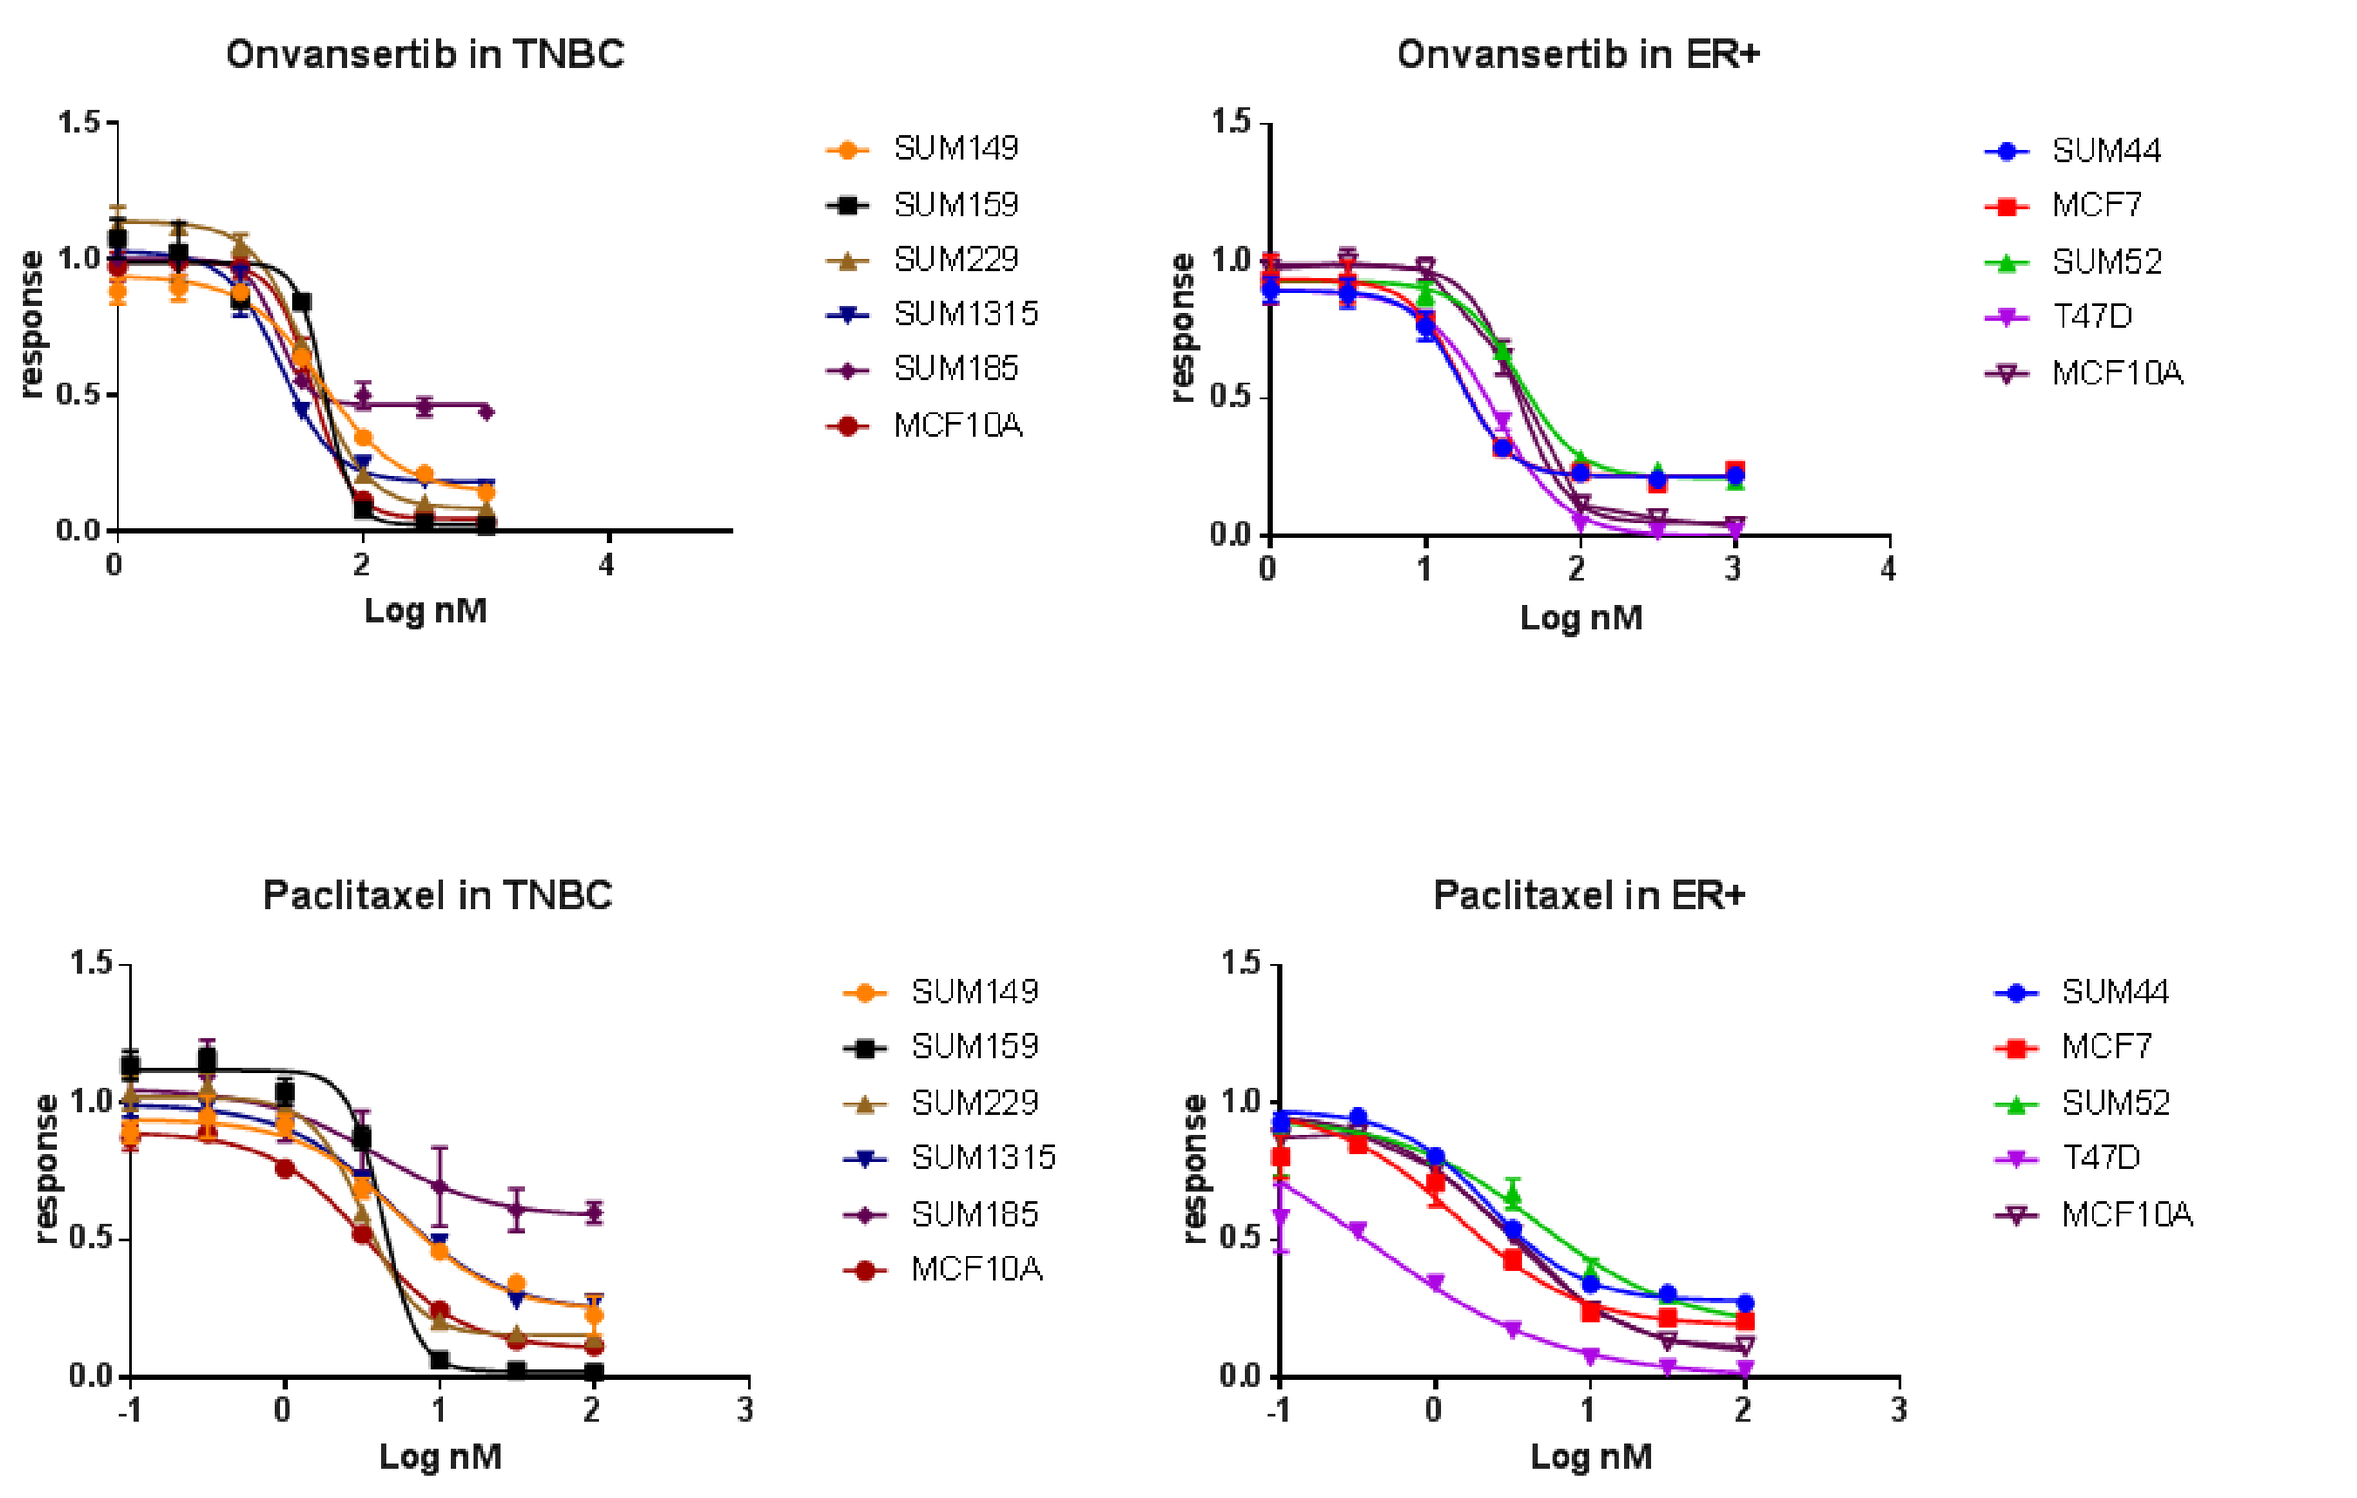
Onvansertib in TNBC
Onvansertib in ER+
Paclitaxel in TNBC
Paclitaxel in ER+
response
Log nM
SUM149
SUM159
SUM229
SUM1315
SUM185
MCF10A
SUM44
MCF7
SUM52
T47D
MCF10A

IC50 nM	SUM149	SUM159	SUM229	SUM1315	SUM185		SUM44	MCF7	SUM52	MCF10A
onvansertib	48.46	49.4	35.01	17.11	19.35		16.61	16.16	38.77	37.32
paclitaxel	5.503	4.141	3.004	4.912	3.605		2.262	2.051	5.153	3.253

- Most of selected TNBC and luminal cells were equally sensitive to single agent Onvansertib (top two figures)
- At different degree and IC_{50}, breast cancer cell lines are sensitive to Paclitaxel (bottom two figures)

Figure 3. Combination Index (CI) of onvansertib plus paclitaxel in SUM149 and SUM159 cells

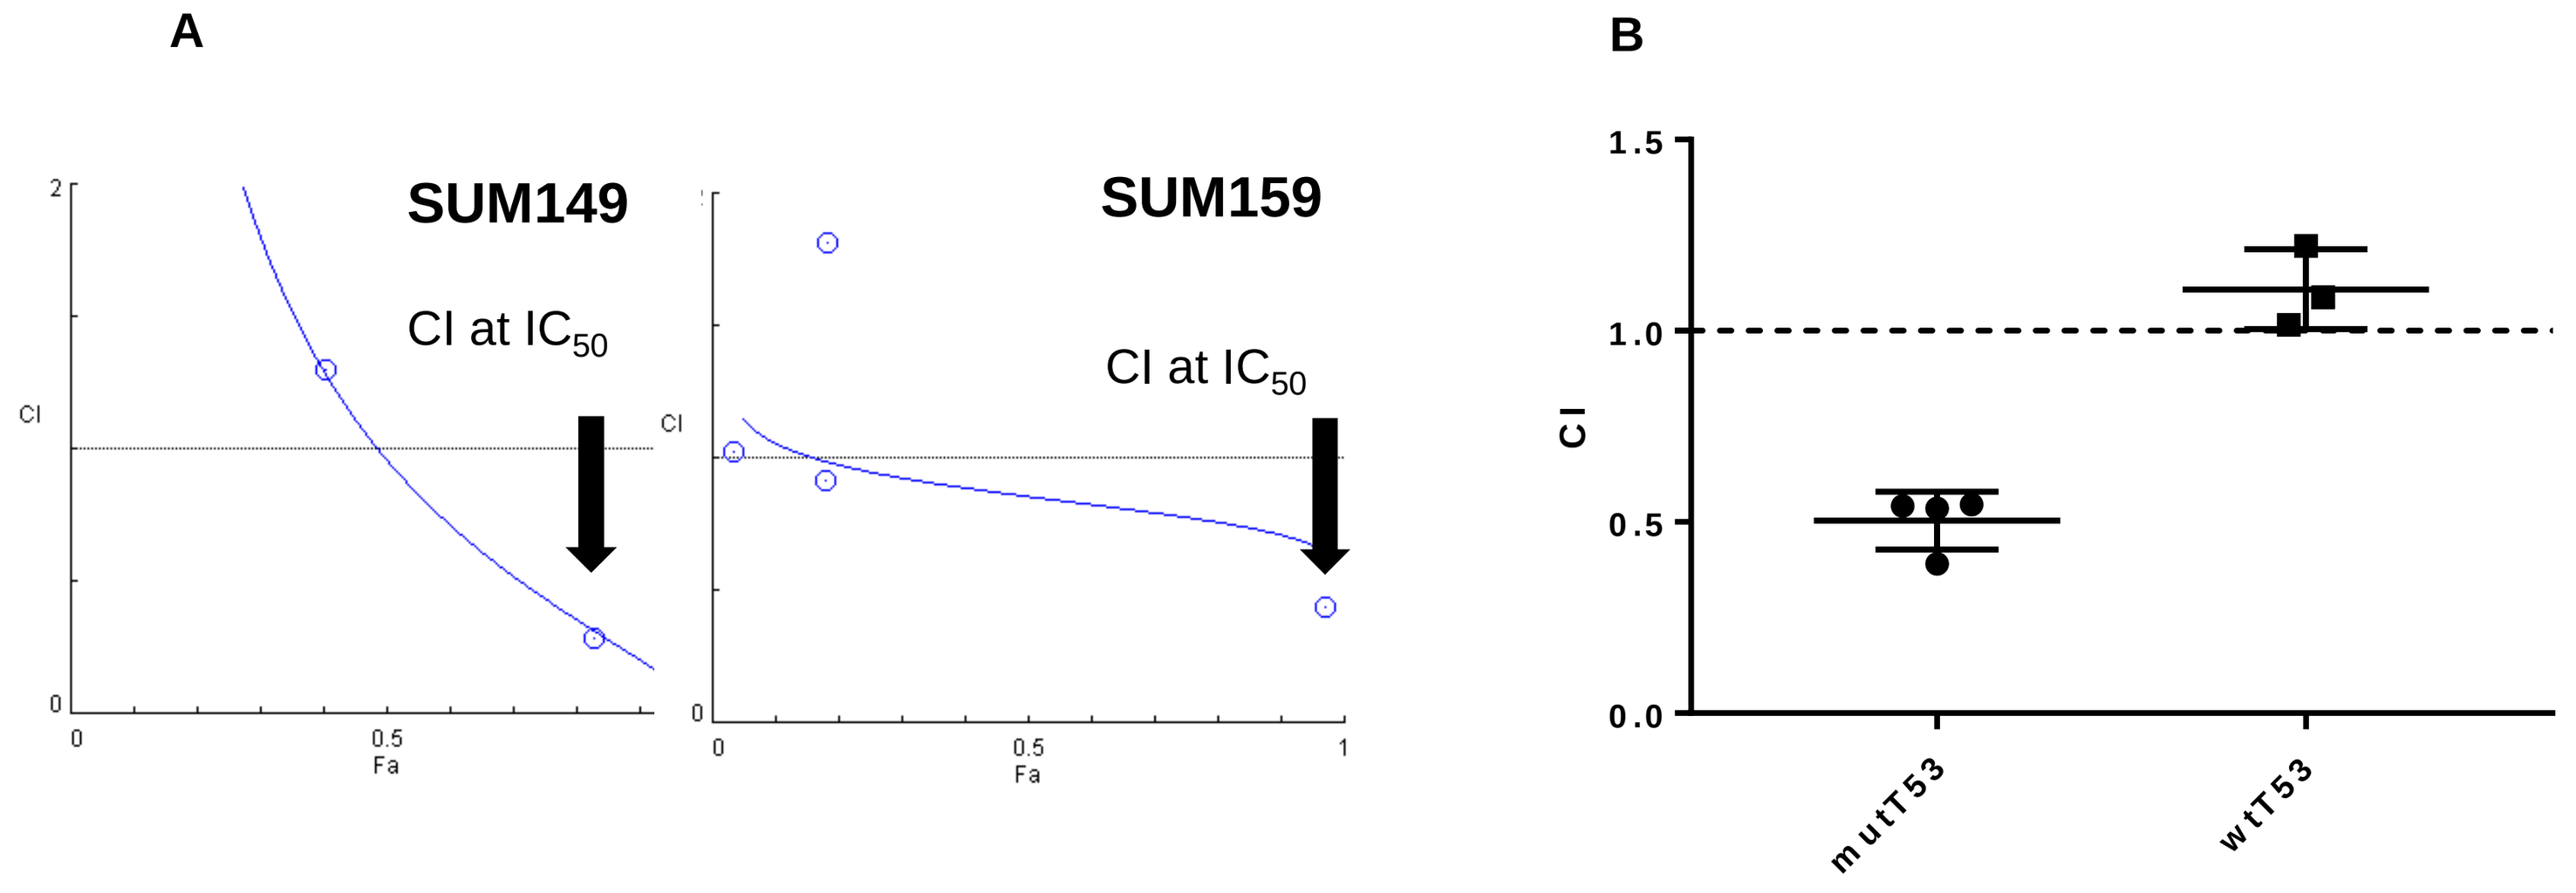
A
B
SUM149
SUM159
CI at IC_{50}
CI at IC_{50}
CI
Fa
mutT53
wtT53

	CI values at:						CI values at:				
Combo	ED50	ED75	ED90	ED95			Combo	ED50	ED75	ED90	ED95
149O+P	0.95773	0.43494	0.20377	0.12365			159O+P	0.85586	0.77729	0.71061	0.67115

- Figure 3A shows synergy of Onvansertib plus Paclitaxel in two cell lines of TNBC, SUM149 and SUM159; Combination Index (CI) in SUM149 = 0.54, and SUM159 CI = 0.54
- Figure 3B shows CI for the combination Paclitaxel-Onvansertib in cell lines with p53 mutated (SUM149, SUM159, SUM52 and T47D) and wild type p53 (SUM1315, MCF7, and MCF10A)
- Synergy observed is not dependent on IC50's or importantly not dependent on TNBC vs ER+/luminal classification but rather p53 mutational status

Figure 4. Cell Cycle Immunoblotting, Apoptosis and DNA Content Analysis in Cells Treated with PLK1 Inhibition Plus Docetaxel

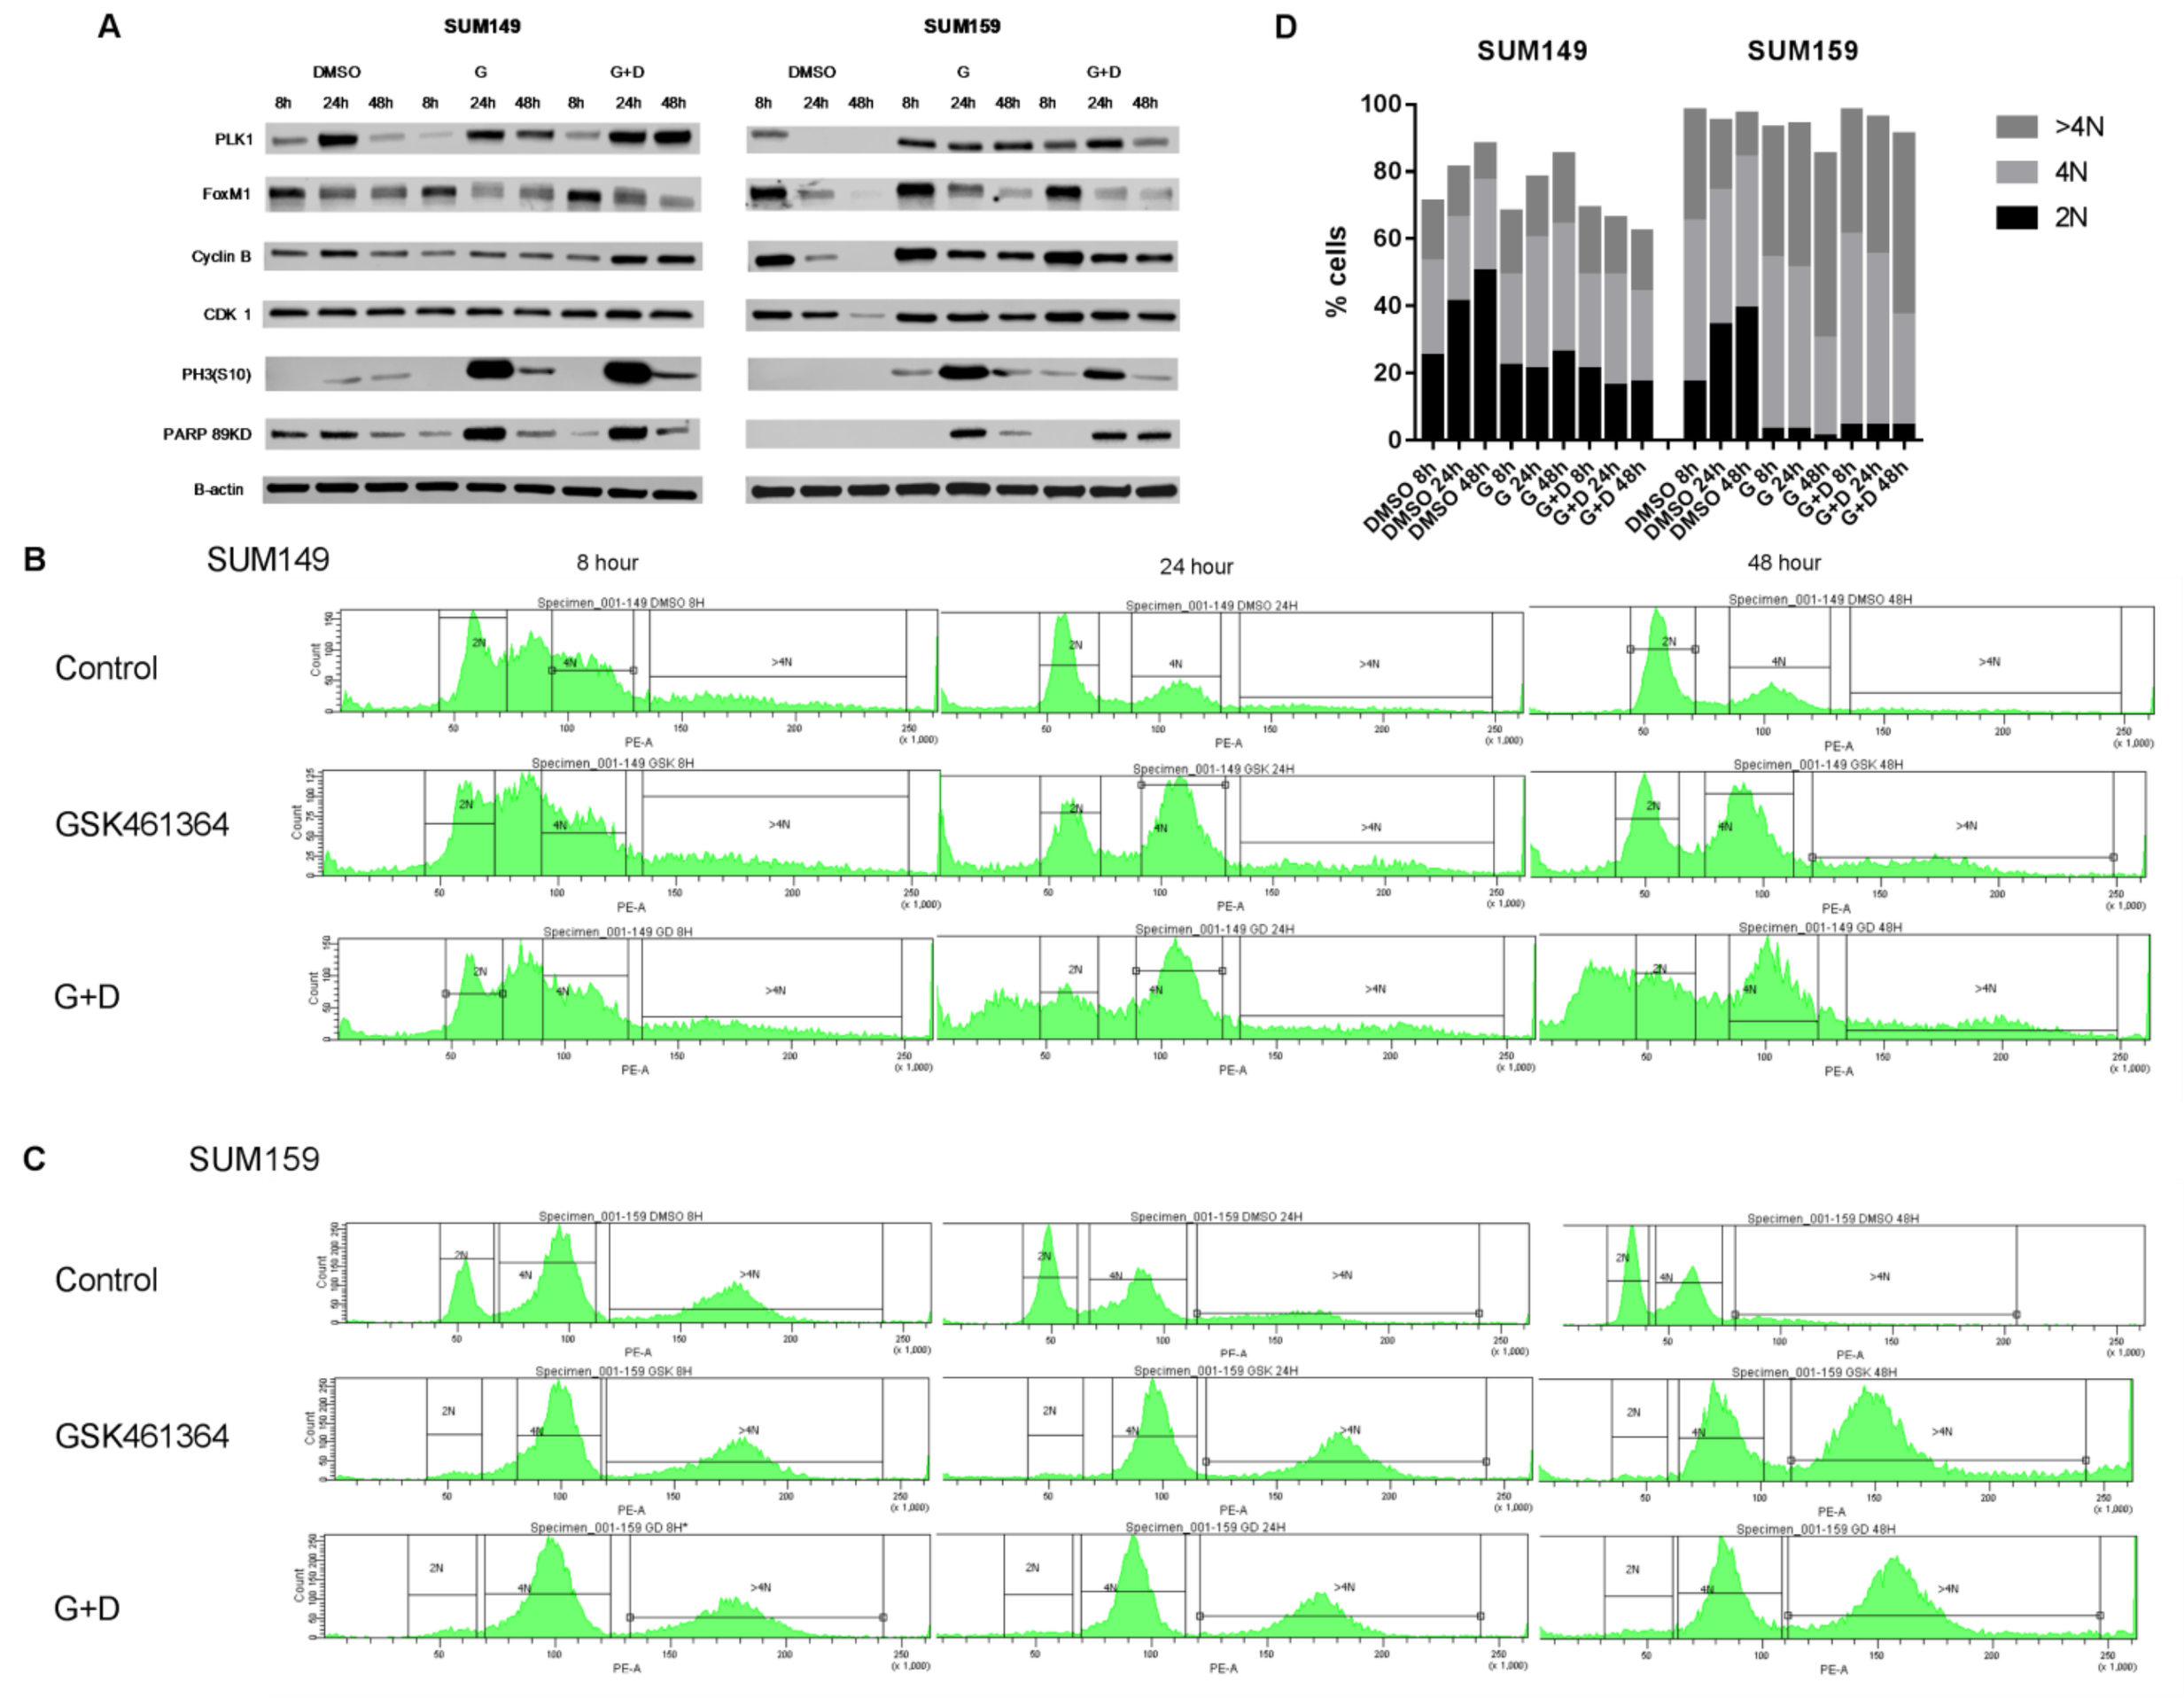
A
SUM149
SUM159
DMSO
G
G+D
PLK1
FoxM1
Cyclin B
CDK 1
PH3(S10)
PARP 89KD
B-actin
D
% cells
>4N
4N
2N
B
SUM149
8 hour
24 hour
48 hour
Control
GSK461364
G+D
C
SUM159
Control
GSK461364
G+D

- PLK1 inhibitor GSK461364 induced mitotic arrest (Cyclin B1 and PLK1 accumulation followed by pH3 accumulation) earlier in SUM159 compared to SUM149.
- This difference could be explained by the fast doubling time and earlier mitosis entry by SUM159 compared to SUM149.
- Treatment with single PLK1 inhibitor induced apoptosis after 24 hours in both cell lines (Fig 6A). In the DNA content analysis with propidium iodide, the percentage of aneuploid SUM149 (Fig 6B) and SUM159 cells (Fig 6C) increased over time (24-48 hours) after treatment with GSK461364 and after combination of GDK461364 with docetaxel. Indeed, aberrant mitotic exit was observed when SUM149 and SUM159 cells were treated with single agent and the combination of Plk1 inhibitor plus docetaxel (Fig 6D).

Figure 5. In vivo efficacy of onvansertib alone or in combination with paclitaxel against SUM159 xenografts

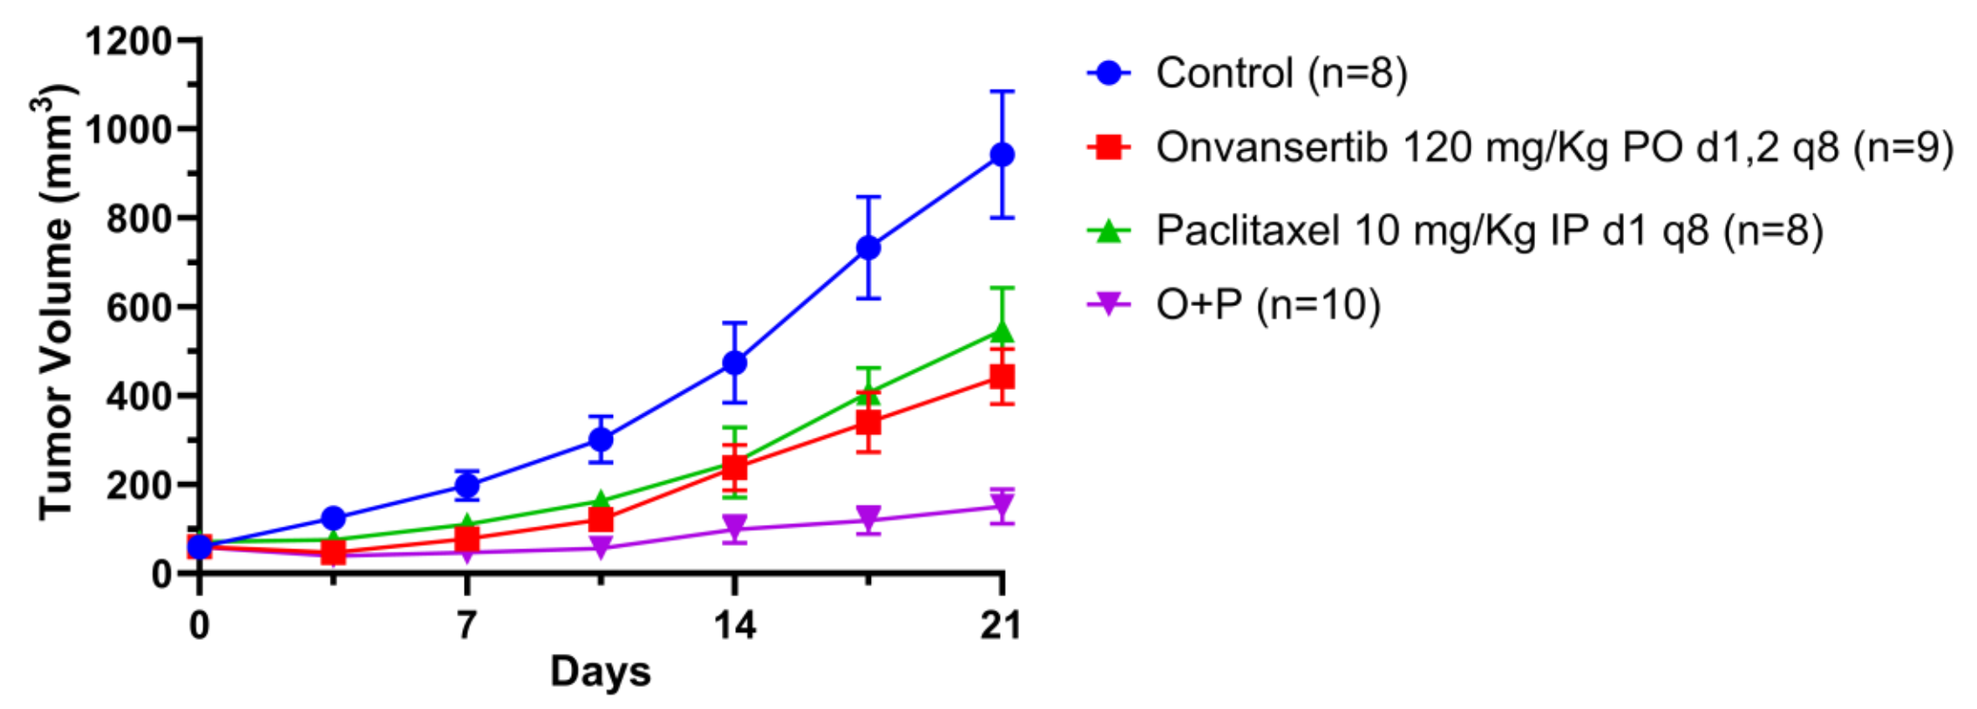
Tumor Volume (mm^3)
Days
Control (n=8)
Onvansertib 120 mg/Kg PO d1,2 q8 (n=9)
Paclitaxel 10 mg/Kg IP d1 q8 (n=8)
O+P (n=10)

SUM159 cells were implanted in the mammary fat pad of NOD-scid-IL2 receptor gamma null female mice, and treatments began 14-21 days later when tumors were well established (tumor volume ≥ 40 mm3). Onvansertib was given by oral gavage (PO) on two consecutive days every week; paclitaxel was given intraperitoneally (IP) once per week; controls received PO vehicle on two consecutive days every week and IP vehicle once per week. Tumor volume was assessed twice per week and mice treated for 3 weeks. Mean of tumor volume with standard errors of the mean were plotted.

- The LME regression model provides all fixed effects for experimental condition (onvansertib, paclitaxel, onvansertib plus paclitaxel and control), time (3, 7, 10, 14, 18 and 21 days), and their interaction are significant (p-value < 0.0001 in all effects).
- Onvansertib and Paclitaxel demonstrated similar tumor growth inhibition when compared to controls (difference=-0.406 and -0.337 with p=0.262 and 0.340, respectively, at 21 days).
- The combination Onvansertib plus Paclitaxel was significantly superior to single agent treatment with difference of -1.346 compared to Onvansertib alone and -1.414 compared to Paclitaxel alone (p<0.0001 and p<0.0001, respectively).

Conclusions and Perspective

- Onvansertib, an oral highly-selective PLK1 inhibitor, and paclitaxel showed synergy in cell models of breast cancer with p53 mutations
- PLK1 inhibition induced mitotic arrest in SUM149 and SUM159
- Aberrant mitotic exit and apoptosis was observed when SUM149 and SUM159 cells were treated with the combination of PLK1 inhibitor plus docetaxel
- Onvansertib demonstrated in vivo activity in a xenograft model of TNBC, especially when in combination with paclitaxel
- Our results showed a substantial synergy of PLK1 inhibition and chemotherapy in mutp53 breast cancer and warrant development of a clinical trial with onvansertib in combination with paclitaxel within the subset of HER2-negative patients with mutated p53, and for which no targeted therapies exist

Correspondence: email: giordana@musc.edu @antgiorda