Abstract #100

A Phase 1b/2 Trial of the PLK1 Inhibitor Onvansertib in Combination with FOLFIRI-bev in 2L Treatment of KRAS-Mutated Metastatic Colorectal Carcinoma (mCRC)

Heinz-Josef Lenz[1], Anup Kasi[2], Lawrence Mendelsohn[3], Timothy Cannon[4], Jason Starr[5], Joleen Hubbard[6], Tanios Bekaii-Saab[7], Maya Ridinger[8], Errin Samuelsz[8], Katherine Ruffner[8], Mark Erlander[8], Daniel Ahn[7]

1. USC Norris Comprehensive Cancer Center, Los Angeles, CA; 2. Kansas University Medical Center, Kansas City, KS; 3. CARTI Cancer Center, Little Rock, AR; 4. Inova Schar Cancer Institute, Fairfax, VA; 5. Mayo Clinic, Jacksonville, FL; 6. Mayo Clinic, Rochester, MN; 7. Mayo Clinic, Phoenix, AZ; 8. Cardiff Oncology, San Diego, CA

Background

Effective second-line (2L) treatment is needed in KRAS-mutated mCRC

- Second-line treatments (chemotherapy ± targeted agents) have a poor prognosis:
 - ORR: 5%-13%, PFS: ~5.7 months, OS: ~11.5 months.[1-4]
- KRAS is mutated in ~50% of CRC patients and, to-date, RAS-directed therapies have been unsuccessful with the majority of KRAS mutations considered to be undruggable:
 - Covalent inhibitors of KRAS G12C (representing 8% of KRAS mutations in CRC) have shown limited activity in CRC.
- Alternative strategies to inhibit KRAS include targeting synthetic lethal partners of mutant KRAS (i.e., proteins that are essential in KRAS-mutant but not wild-type cells).

Onvansertib, an oral and highly-selective PLK1 inhibitor, is a promising therapeutic option for KRAS-mutated CRC:

- PLK1, a key regulator of mitosis, is overexpressed in CRC and associated with adverse clinical features.[5]
- A genome-wide RNAi screen identified PLK1 inhibition to be synthetic lethal with mutant KRAS in CRC cells.[6]
- Onvansertib demonstrated potent anti-tumor activity as single agent and showed synergy in combination with irinotecan and with 5-FU in the HCT-116 KRAS mutant xenograft model.

1. Giessen et al., *Acta Oncologica*, 2015, 54: 187-193; **2.** Cremolini et al., *Lancet Oncol* 2020, 21: 497–507; **3.** Antoniotti et al., Correspondence *Lancet Oncol* June 2020; **4.** Bennouna et al., *Lancet Oncol* 2013; 14: 29–37; **5.** Weichert et al., *World J Gastroenterol.* 2005; 11(36):5644-50; **6.** Luo et al, *Cell.* 2009; 137 835-48.

Methods

Phase 1b/2 Trial Design

FIGURE 1. TREATMENT SCHEDULE: FOLFIRI/BEVACIZUMAB + ONVANSERTIB

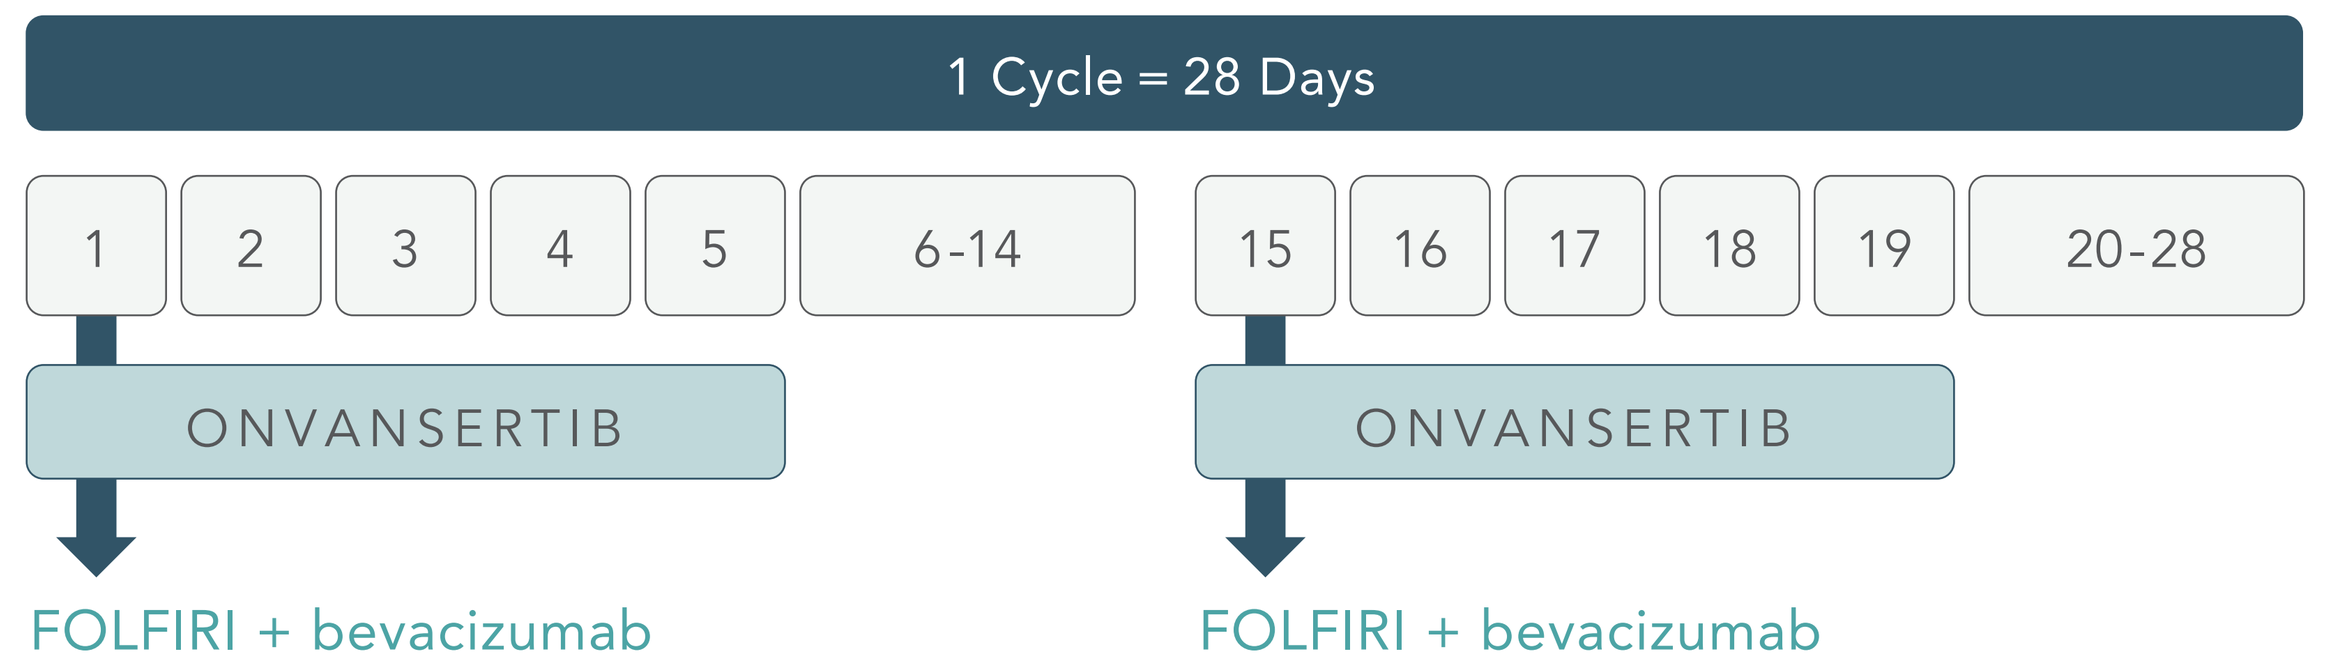
Treatment Schedule
1 Cycle = 28 Days
1
2
3
4
5
6-14
15
16
17
18
19
20-28
ONVANSERTIB
ONVANSERTIB
FOLFIRI + bevacizumab
FOLFIRI + bevacizumab

Primary Endpoints

- **Phase 1b:** Characterization of dose-limiting toxicities (DLTs), adverse events (AEs) and toleratbility; determination of maximum tolerated dose (MTD) and recommended Phase 2 dose (RP2D).
- **Phase 2:** Objective response rate (ORR) per RECIST v1.1 in patients who receive at least 1 cycle of treatment (28 days).

Secondary Endpoints

- Progression-free survival (PFS).
- Reduction in KRAS mutant allelic frequency (MAF) assessed in circulating tumor DNA (ctDNA).

Key Eligibility Criteria

- Metastatic and unresectable CRC with KRAS mutation determined in a CLIA-certified lab.
- Failure of or intolerance to first line (1L) treatment of fluoropyrimidine and oxaliplatin with or without bevacizumab.

TABLE 1. ENROLLMENT AS OF 3-DEC-2021

Number of patients (N)	Phase 1b Dose Level 0 Onvansertib 12 mg/m²	Phase 1b Dose Level +1 Onvansertib 15 mg/m²	Phase 1b Dose Level +2 Onvansertib 18 mg/m²	Phase 2 RP2D Onvansertib 15 mg/m²	Total Patients All Doses
Treated	6	6	6	32	50
Currently on Treatment	0	1	0	15	16

TABLE 2. BASELINE CHARACTERISTICS

Total Number of Patients = 50	Median [range] or n (%)
Age, Years	61 (35-83)
Sex	
Male	28 (56%)
Female	22 (44%)
ECOG[1]	
0	33 (69%)
1	15 (31%)
Primary Tumor Site[2]	
Colon	27 (55%)
Rectum	17 (35%)
Other	5 (10%)
Liver Metastasis[3]	
None	11 (22%)
Liver and Other	29 (59%)
Liver Only	9 (18%)
Number of Metastatic Organs[4]	
1	17 (35%)
≥2	32 (65%)
Prior Bevacizumab Treatment[5]	
Yes	33 (67%)
No	16 (33%)

1. ECOG not reported for two patients; **2.** Primary tumor site not reported for one patient; **3.** Liver metastasis presence not reported for one patient; **4.** Number of metastatic organs not reported for one patient; **5.** Prior bevacizumab treatment not reported for one patient.

Study Contact: Heinz-Josef Lenz, MD, lenz@usc.edu; **Presented at:** ASCO GI 2022, Poster Session C: Cancers of the Colon, Rectum, and Anus

Results

Safety

TABLE 3. EVENTS OCCURRING IN ≥10% OF PATIENTS IN DESCENDING ORDER OF FREQUENCY (N=50)

Treatment Emergent Adverse Events*	Grade 1	Grade 2	Grade 3	Grade 4	All Grades
Neutropenia	1	13	15	6	35
Fatigue	15	15	3	0	33
Nausea	24	7	2	0	33
Diarrhea	15	7	2	0	24
Abdominal Pain	13	7	1	0	21
Mucositis	11	6	2	0	19
Alopecia	17	2	0	0	19
WBC Decrease	6	9	2	1	18
Platelet Count Decrease	10	4	1	0	15
Hypertension	2	8	5	0	15
Anemia	9	4	1	0	14
Vomiting	9	3	1	0	13
Musculoskeletal Pain[1]	11	1	0	0	12
Infection[2]	3	4	4	0	11
Hemorrhage[3]	8	0	1	0	9
Headache	7	0	0	0	7
Neuropathy	5	2	0	0	7
GERD	7	0	0	0	7
ALT Increase	4	0	1	0	5

*Numbers indicate number of patients experiencing the event, (regardless of causality). Each patient is only counted once and only for the highest grade of a given event; **1.** Includes the terms back pain, arthralgia, myalgia, musculoskeletal pain, neck pain, bone pain and temporomandibular joint syndrome; **2.** Includes the terms localized infection, urinary tract infection, sepsis, liver abscess, abdominal abscess, hepatitis B, oral candida, sinusitis, H. pylori infection, and cellulitis; **3.** Includes the terms epistaxis, hematochezia, rectal hemorrhage, gingival bleeding, hematuria, bleeding J tube site.

- 7 patients had a total of 11 G4 adverse events:
 - Neutropenia (n=7); Decreased WBC (n=2); Neutropenic fever (n=1); Hyperphosphatemia (n=1).
- Combination regimen was well tolerated:
 - Of all AEs only 11% (84/788) were G3/G4.
- Discontinuation of the 5-FU bolus from the FOLFIRI regimen and use of growth factors ameliorated the severity of neutropenia observed.
- No major or unexpected toxicities were attributed to onvansertib.

Efficacy

TABLE 4. EFFICACY

Summary	RP2D	All Doses
# of Patients	15 mg/m²	12 mg/m², 15 mg/m², 18 mg/m²
Treated Patients	35*	50
Patients Discontinued Prior to Completing Cycle 1 (28-days)	0	2
Patients on Treatment at 3-Dec-21	14	16
Patients Having First Post-Baseline Scan	35	48
Best Objective Response		
Partial Response (PR) + Complete Response (CR)**	11/35 (31%)	16/48 (33%)
Stable Disease (SD)	22/35 (63%)	28/48 (58%)
Progressive Disease (PD)	2/35 (6%)	4/48 (8%)

*Three patients were considered unevaluable at the RP2D because they were assigned to 15 mg/m² but dosed at 12 mg/m²;
**Includes both confirmed and unconfirmed PR's.

- Of the 48 patients evaluable for efficacy across the study, 16 patients (33%) have had a PR (15) or CR (1), 13 (27%) of which (12 PR, 1 CR) were confirmed.
- Of the 35 patients treated at the RP2D of 15 mg/m² evaluable for efficacy, 11 (31%) achieved either a CR (1 patient) or a PR (10 patients), 10 (29%) of which were confirmed (1CR, 9 PR).
- 3 initial PRs were not confirmed for the following reasons:
 - Discontinuation from trial for an unrelated AE (hep B) prior to confirmatory scan.
 - Discontinuation from trial to pursue metastasis-directed therapy (microwave ablation).
 - Regressed to SD; still on treatment.
- Of the 48 patients evaluable for response, a total of 5 patients discontinued from therapy to pursue potentially-curative metastasis-directed therapy (surgery or microwave ablation), including 2 patients with SD.

FIGURE 2. DURATION OF THERAPY AND DISPOSITION

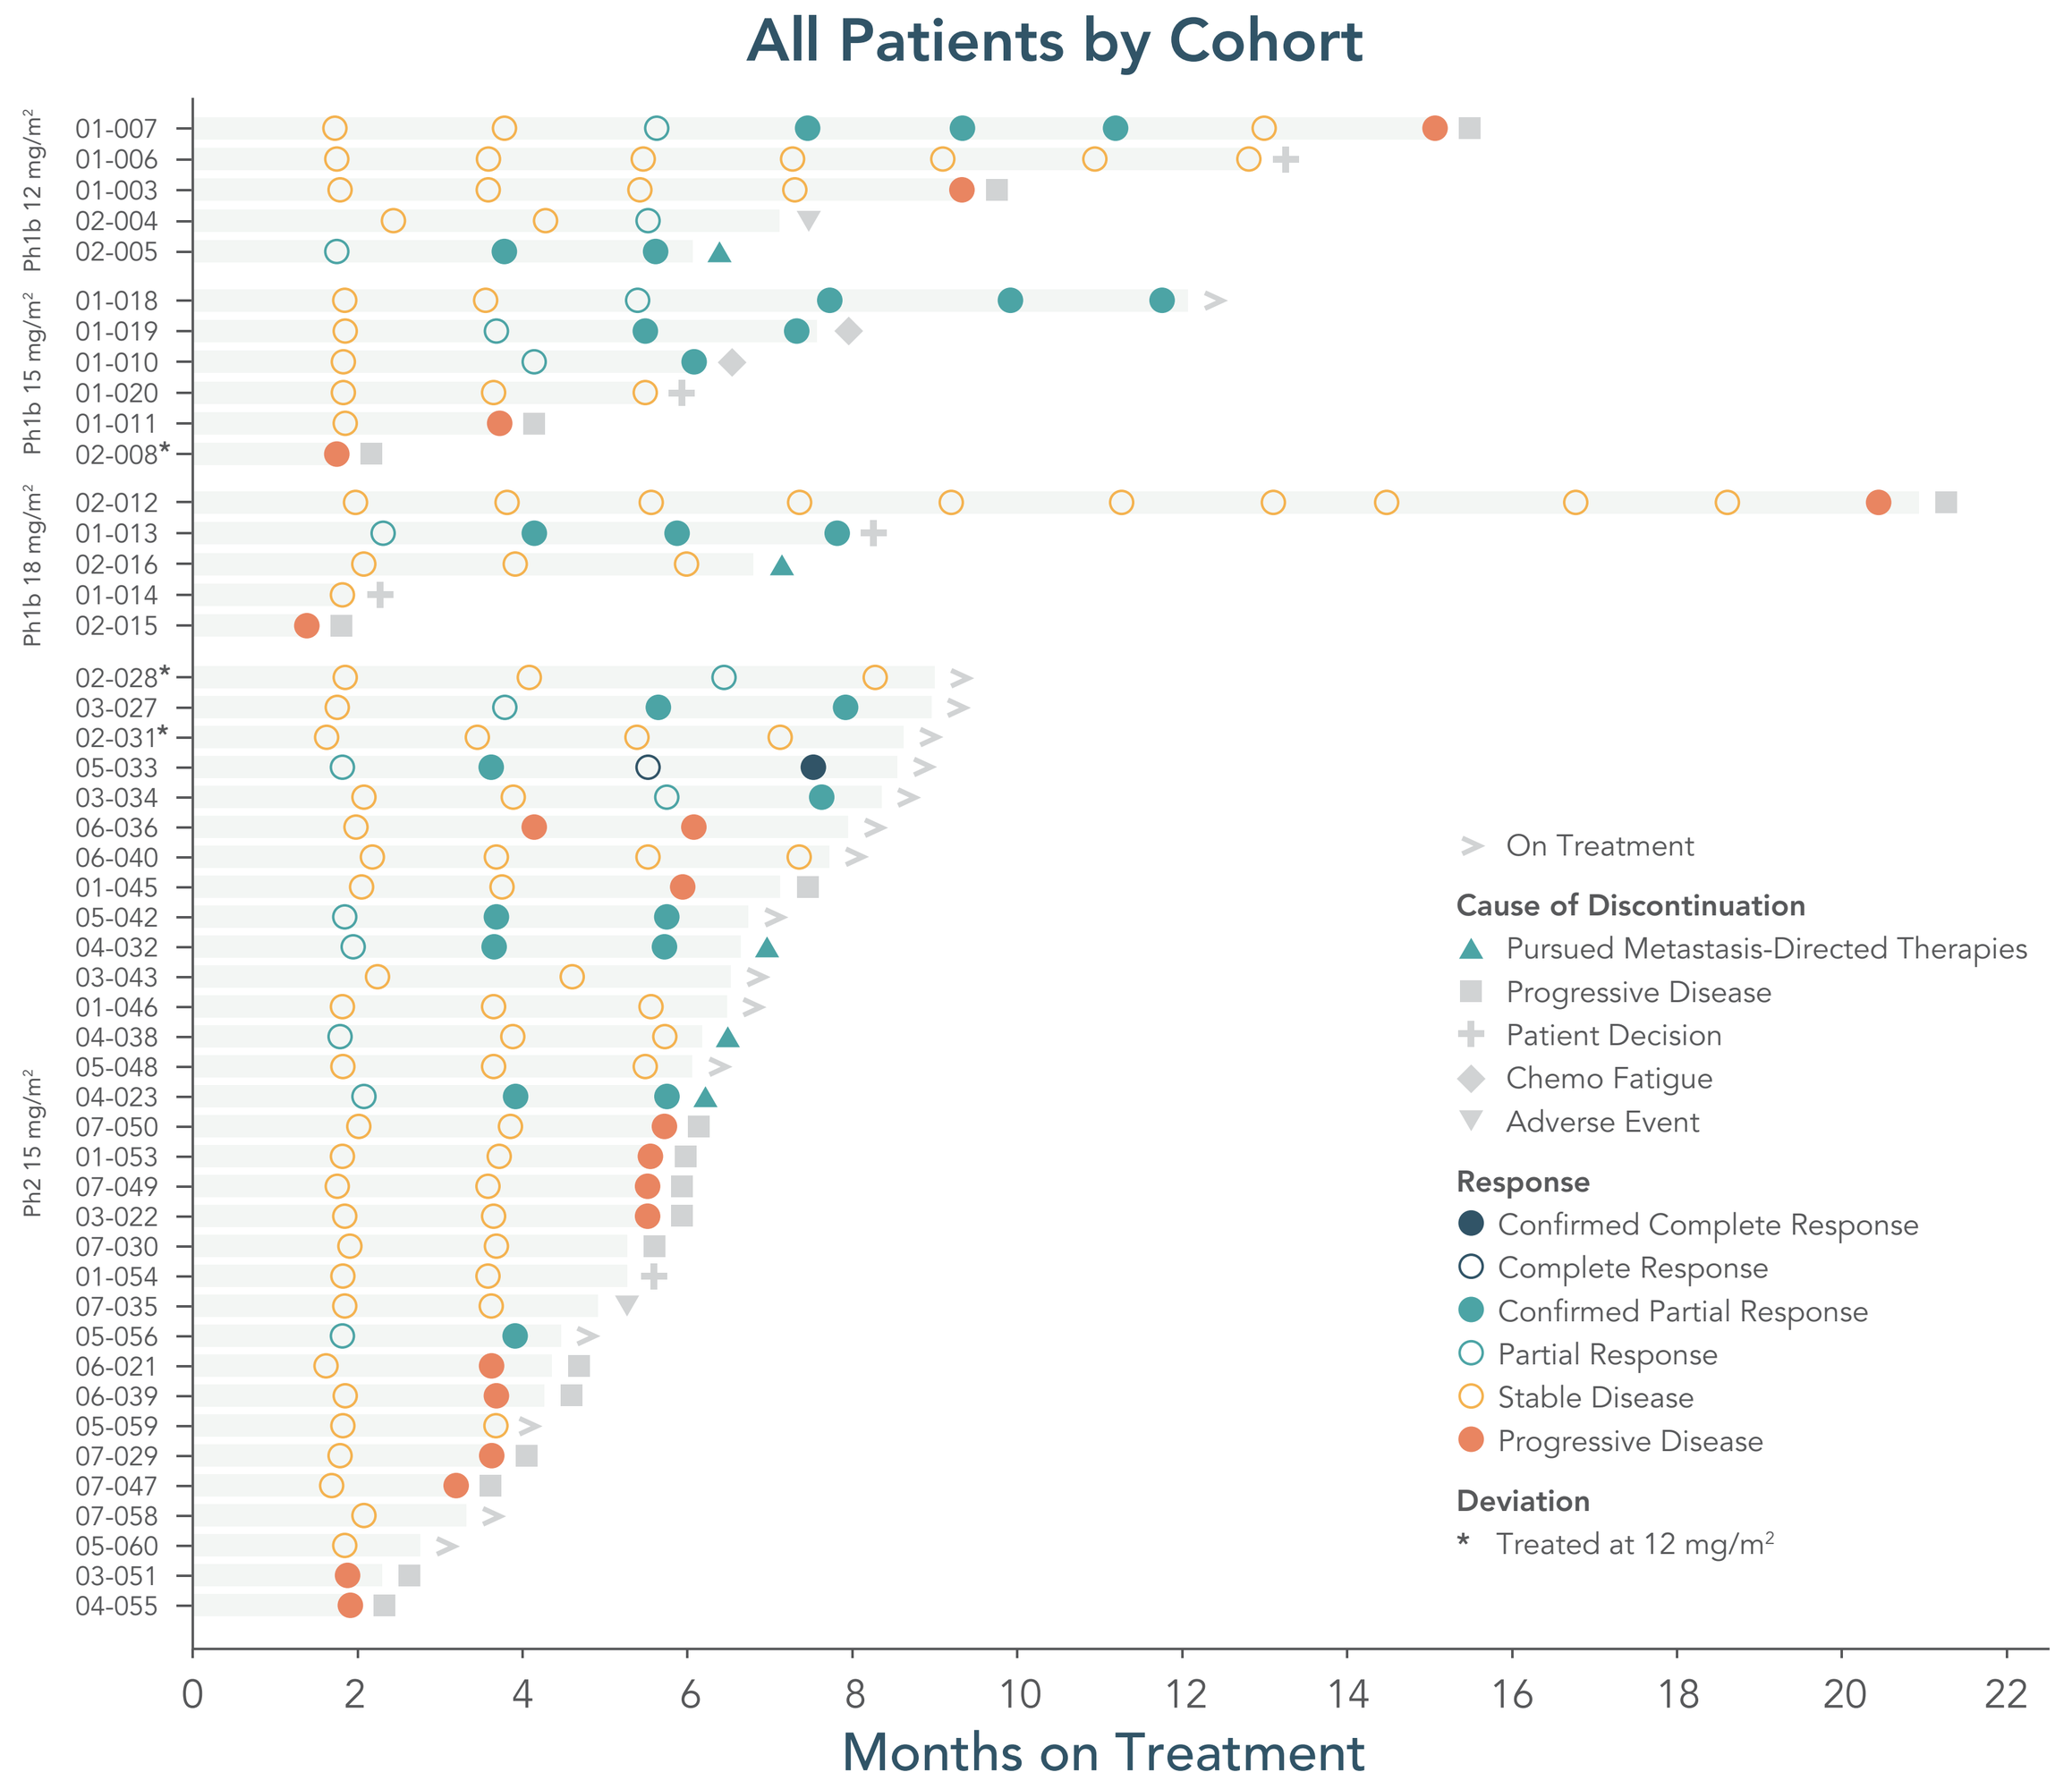
All Patients by Cohort
Ph1b 12 mg/m²
Ph1b 15 mg/m²
Ph1b 18 mg/m²
Ph2 15 mg/m²
On Treatment
Cause of Discontinuation
Pursued Metastasis-Directed Therapies
Progressive Disease
Patient Decision
Chemo Fatigue
Adverse Event
Response
Confirmed Complete Response
Complete Response
Confirmed Partial Response
Partial Response
Stable Disease
Progressive Disease
Deviation
* Treated at 12 mg/m²
Months on Treatment

FIGURE 3. BEST RADIOGRAPHIC RESPONSE, ALL DOSES (N=48)

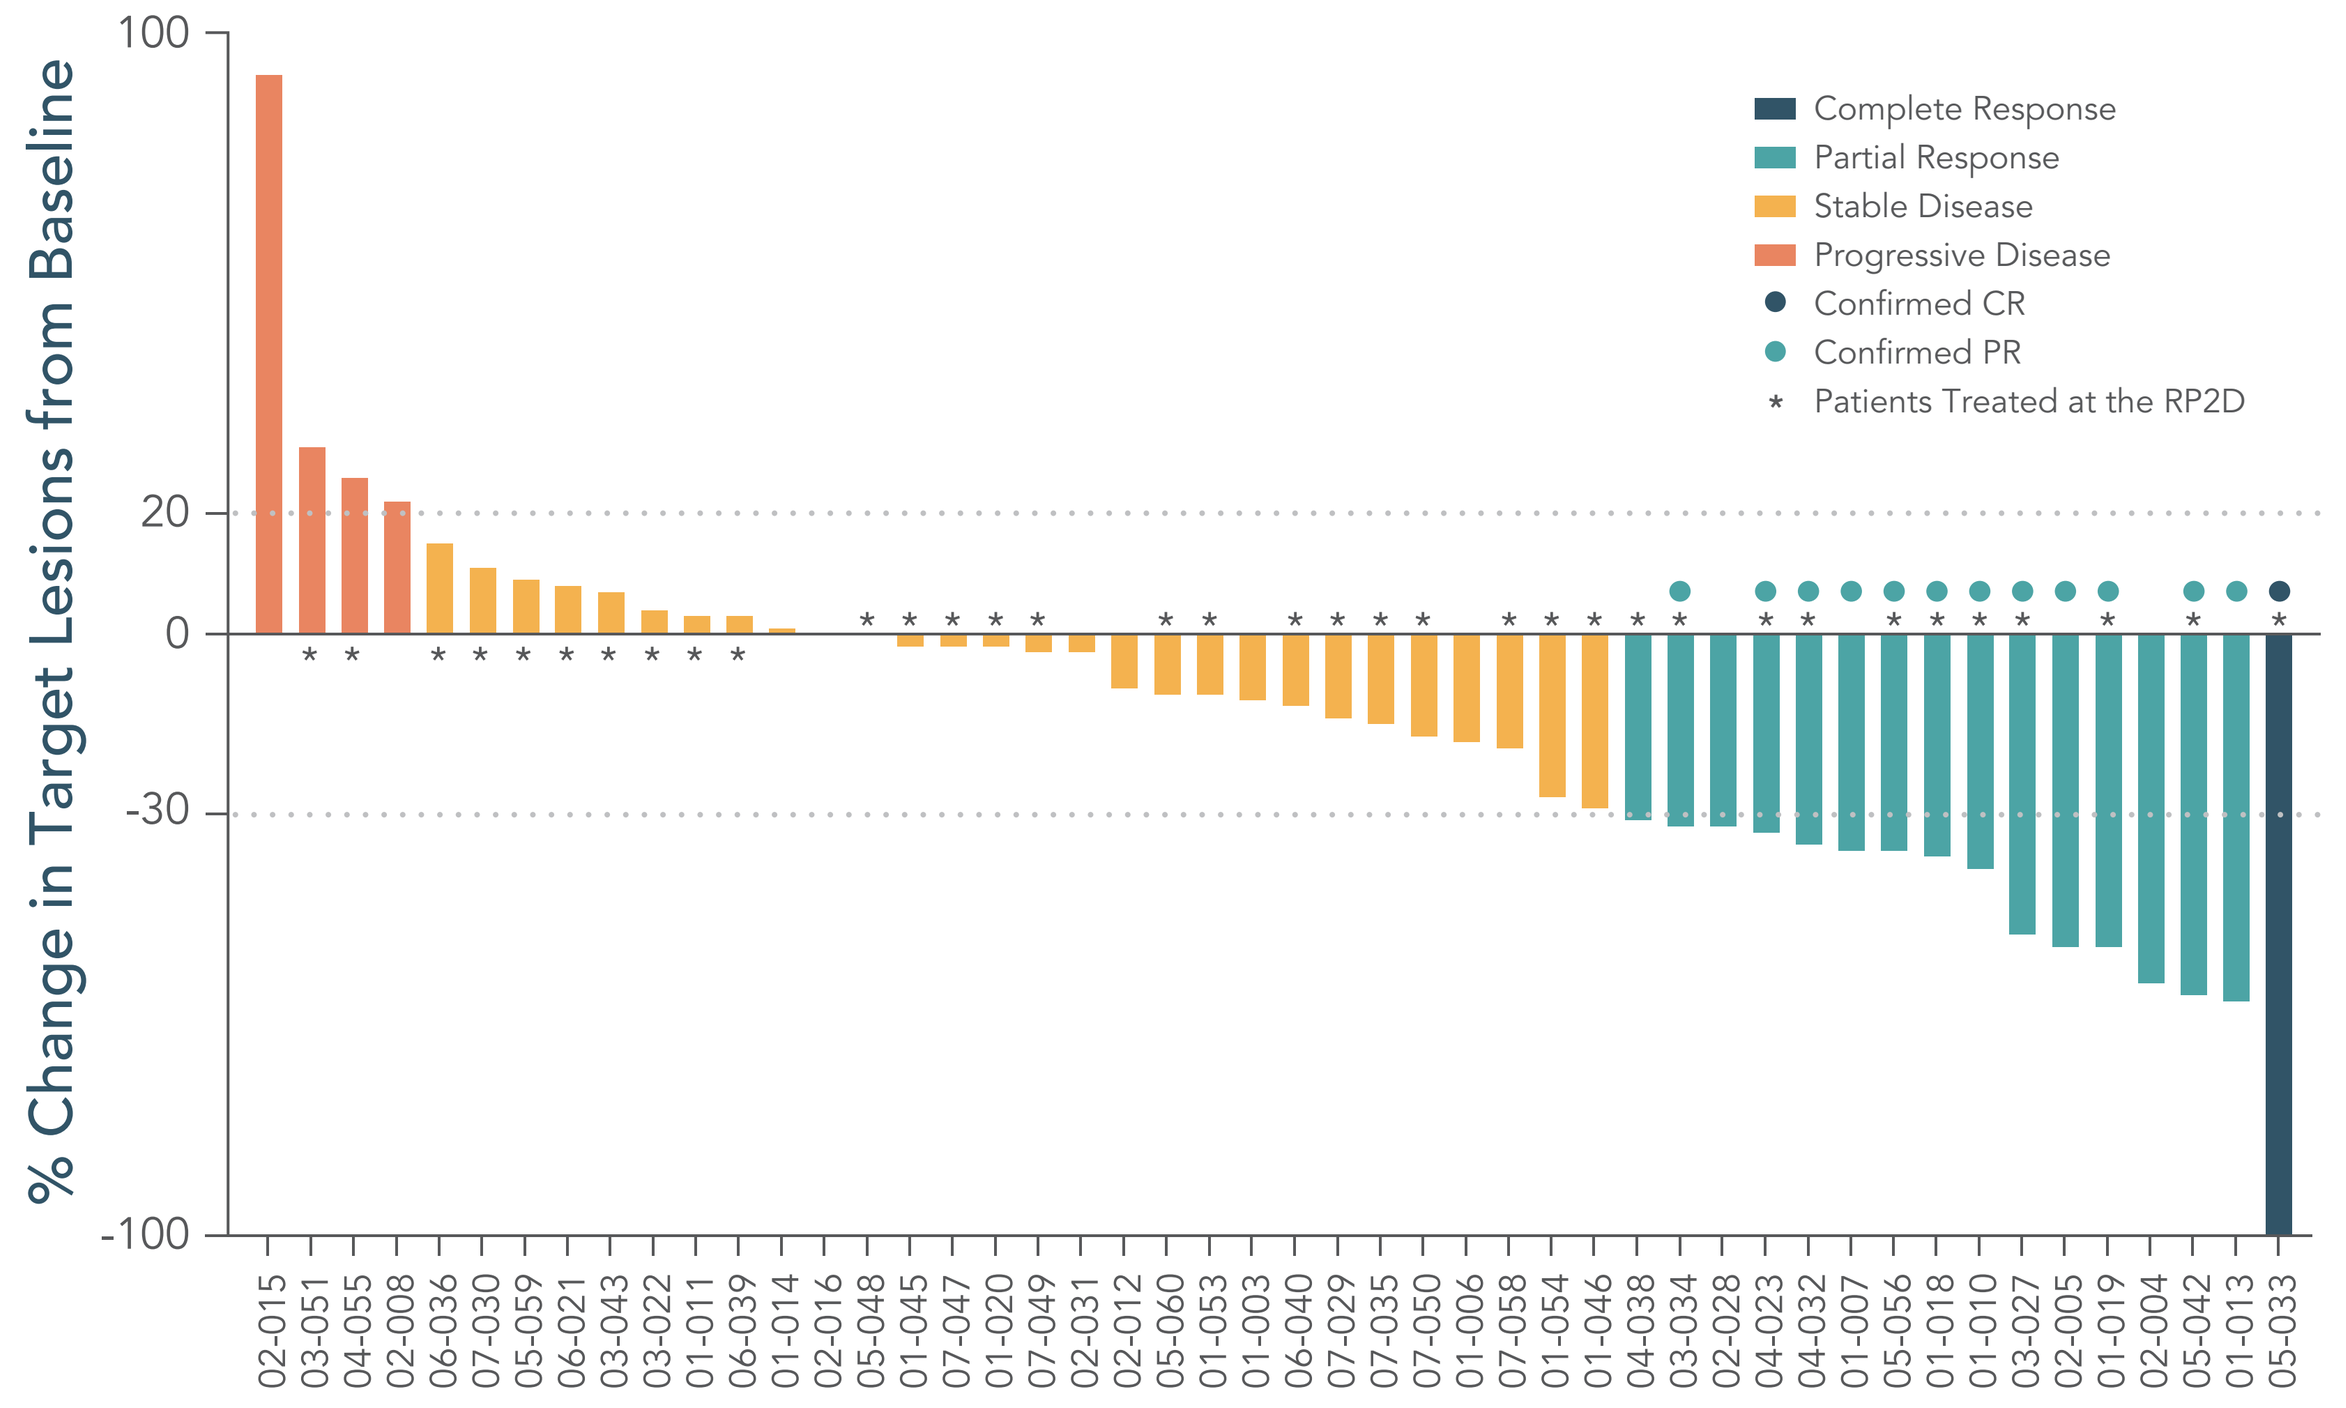
% Change in Target Lesions from Baseline
Complete Response
Partial Response
Stable Disease
Progressive Disease
Confirmed CR
Confirmed PR
* Patients Treated at the RP2D

TABLE 5. PROGRESSION-FREE SURVIVAL: ALL DOSES – 48 EVALUABLE PATIENTS

N	Events	Median	0.95 LCL	0.95 UCL
48	19	9.37	7.13	NA

FIGURE 4. KRAS MUTATIONS AND DISEASE RESPONSE

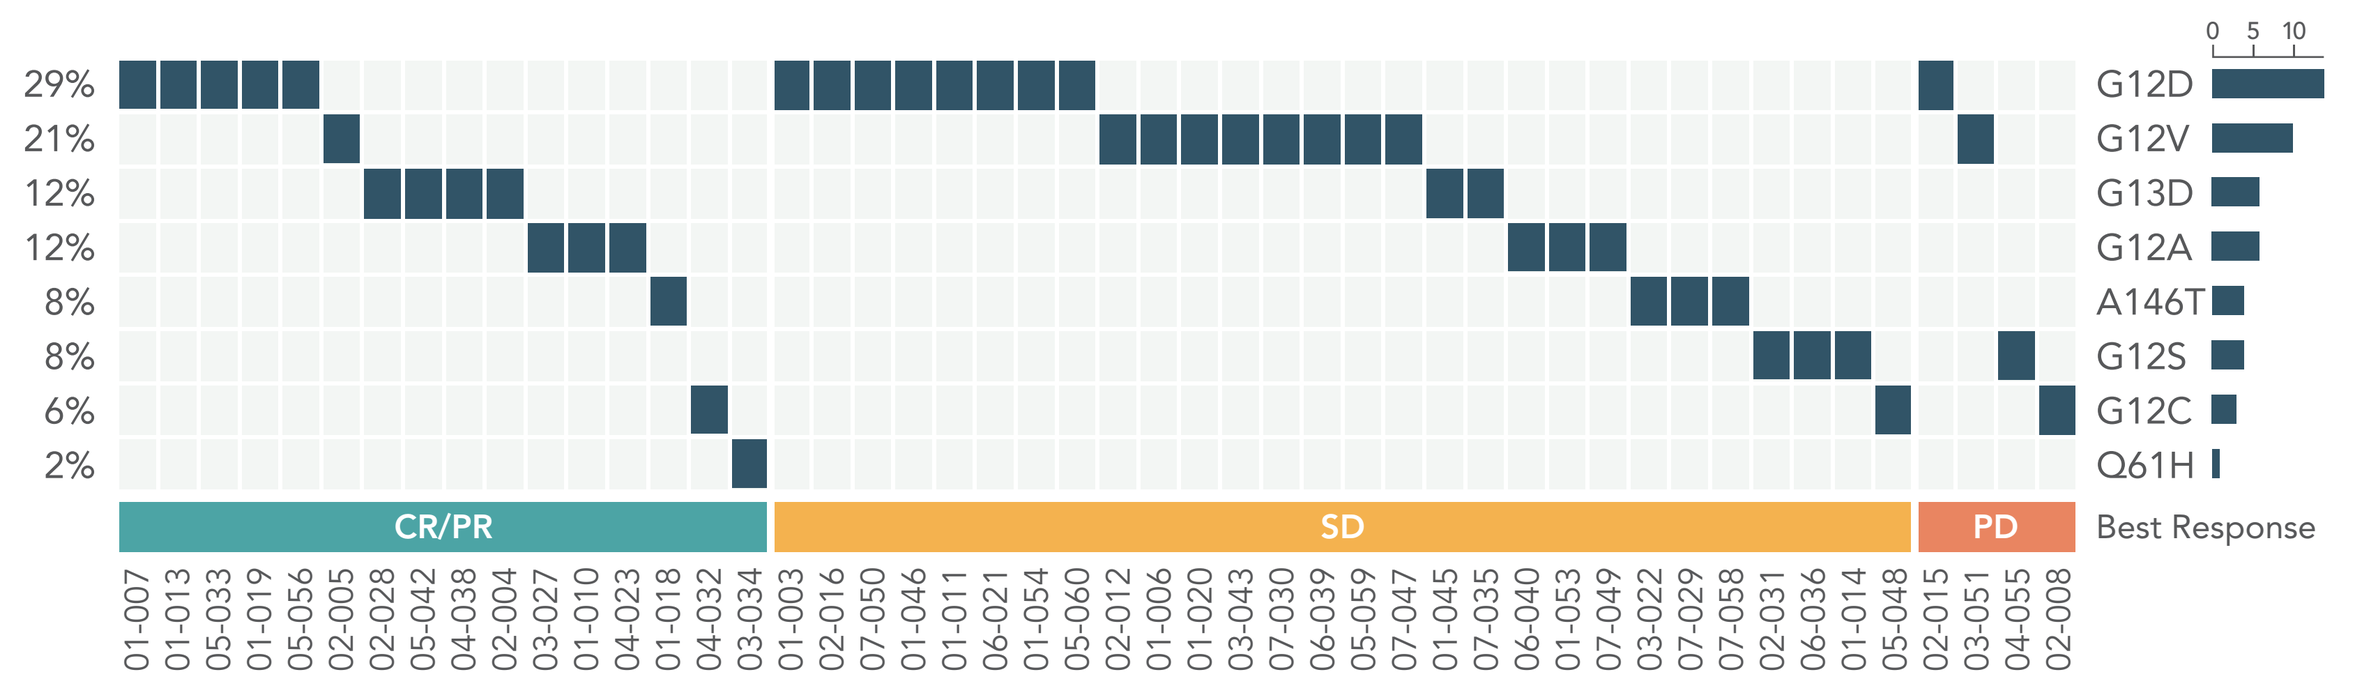
29% G12D
21% G12V
12% G13D
12% G12A
8% A146T
8% G12S
6% G12C
2% Q61H
CR/PR
SD
PD
Best Response

- Clinical responses were observed across different KRAS variants.

FIGURE 5. BEST RESPONSE AND KRAS MUTATIONAL BURDEN CHANGE

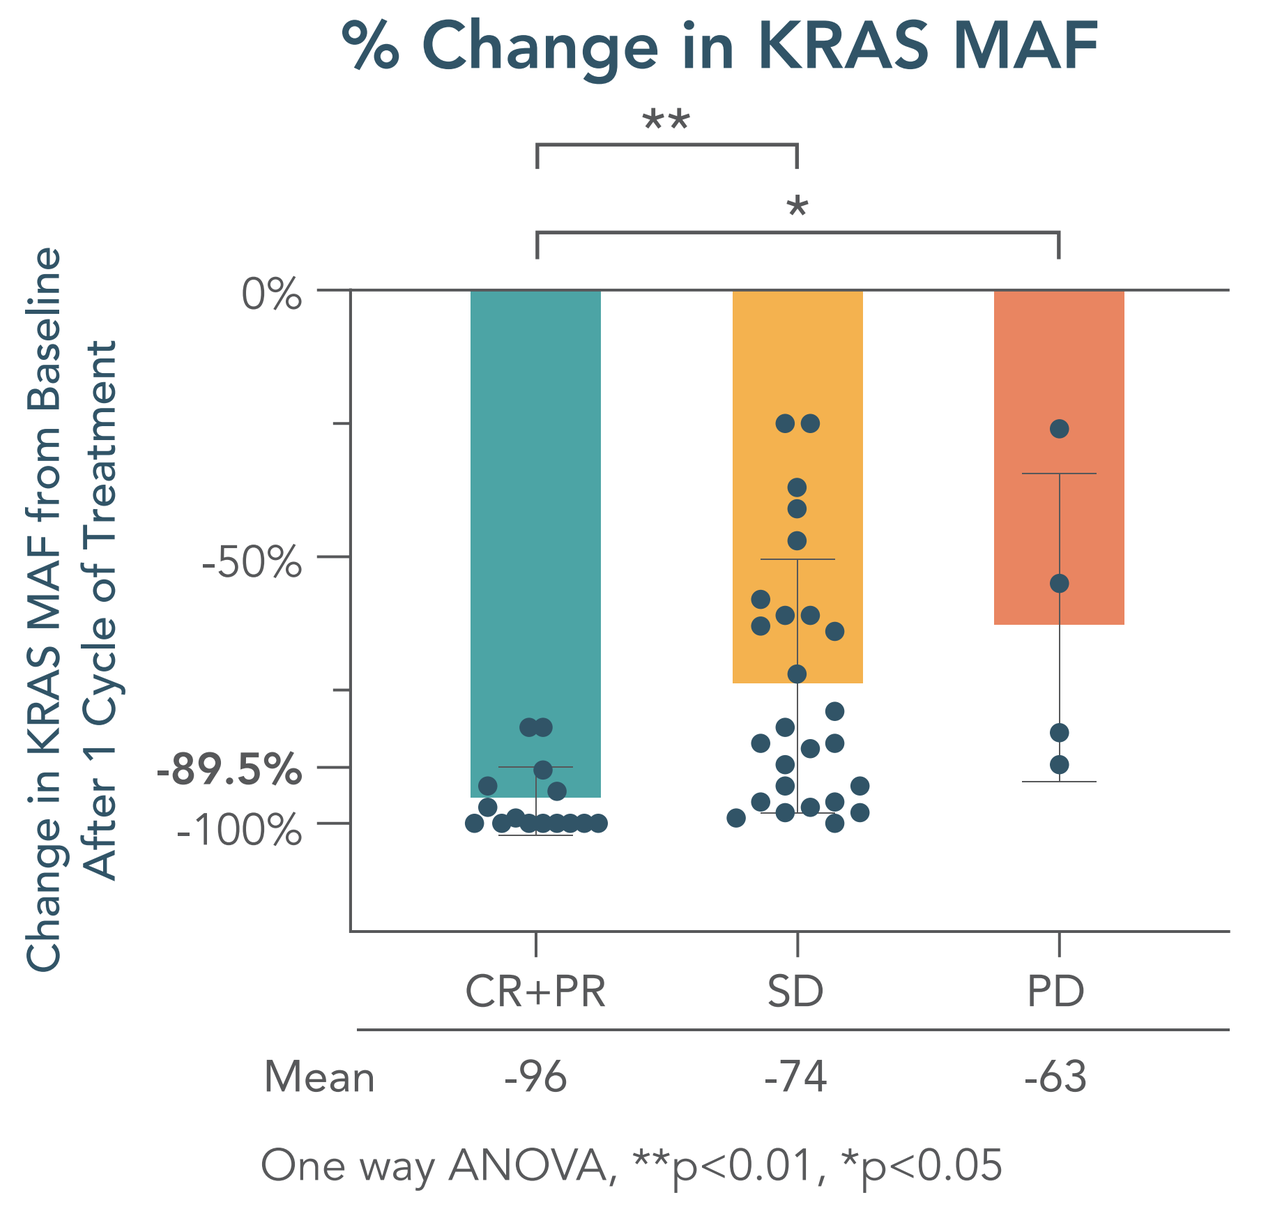
% Change in KRAS MAF
Change in KRAS MAF from Baseline After 1 Cycle of Treatment
0%
-50%
-89.5%
-100%
CR+PR
SD
PD
Mean -96 -74 -63
One way ANOVA, **p<0.01, *p<0.05

- % Change in KRAS MAF by digital droplet polymerase chain reaction (ddPCR) after one cycle of treatment relative to baseline.
- Decrease in KRAS MAF was significantly greater in patients achieving PR vs patients with SD or PD.

Conclusions

- The combination of onvansertib with FOLFIRI-bev is well-tolerated at the onvansertib RP2D of 15 mg/m² given on days 1-5 and 15-19 of a 28 day cycle.
 - Neutropenia is the most common treatment emergent AE and has been easily managed by elimination of the 5FU bolus from the FOLFIRI and use of supportive care with myeloid growth factors.
- Onvansertib in combination with standard of care FOLFIRI-bev has demonstrated a promising objective response rate (ORR) of 33% in all doses and 31% at the RP2D compared to historical controls of 5-13% for FOLFIRI-bev alone in the 2L treatment of KRAS-mutated mCRC following failure of oxaliplatin + fluoropyrimidine in 1L.
 - Onvansertib is a novel agent targeting a broad range of KRAS mutations with promising efficacy in 2L treatment of mCRC.
- Decreases in plasma mutant KRAS after the 1st cycle of treatment were associated with radiographic disease response and may be a promising biomarker.