Abstract #1236

Biomarkers of Response to Abiraterone and the Polo-Like Kinase 1 (PLK1) Inhibitor Onvansertib in Metastatic Castration Resistant Prostate Cancer (mCRPC) Patients

M Ridinger PhD[1], E Samuelsz[1], PJP Croucher PhD[1], M Erlander PhD[1], K Ruffner MD[1], T Smeal PhD[1], DJ Einstein MD[2]

[1]Cardiff Oncology, San Diego, CA; [2]Beth Israel Deaconess Medical Center, Boston, MA

Background

Metastatic CRPC

- Metastatic CRPC is a leading cause of cancer death worldwide.
- Abiraterone (abi) + prednisone is a standard-of care in either castration-sensitive or castration-resistant disease and increases survival.
- Unfortunately, over time (~9-15 months) resistance develops to anti-androgen therapy and new therapeutic approaches are necessary for these patients.

PLK1 – A Promising Target for Prostate Cancer

- PLK1 is a serine/threonine kinase, master regulator of the cell cycle progression:
 - controls mitotic entry and progression.[1]
 - is involved in the DNA damage response through the regulation of homologous recombination-mediated DNA repair and the promotion of the G2/M DNA damage checkpoint recovery.[2]
- PLK1 is overexpressed in prostate cancer and linked to higher tumor grades.[3]

1. Zitouni et al., *Nat Rev Mol Cell Biol.* 2014,15(7); **2.** Joukov V and De Nicolo A., *Sci. Signal.* 2018, 11(543); **3.** Weichert et al., *Prostate* 2004, 60(3).

Onvansertib

- Is a highly specific and orally available PLK1 inhibitor with a 24-hour half-life.
- Has demonstrated safety in advanced/metastatic solid tumors.[1]

1. Weiss et al., *Invest New Drugs* 2018, 36(1).

Onvansertib Synergizes with Abi in an AR-Independent Manner

- Onvansertib induced synergistic cell death and mitotic arrest in combination with abi in CRPC cells.
- The synergy between onvansertib and abi was AR-independent:
 - other antiandrogens, such as enzalutamide, did not synergize with onvansertib.
 - onvansertib synergized with abi in non-prostate cancer cell lines lacking AR.
- Onvansertib induced significant tumor growth inhibition in combination with abi in the AR-V7+ abi-resistant patient derived xenograft model LVCaP2CR.

Clinical Study – NCT03414034

Trial Design

Key Eligibility Criteria

- Initial signs of abiraterone resistance defined as 2 rising PSAs; one rise of ≥0.3 ng/mL separated by one week.

Key Exclusion Criteria

- Prior treatment with either enzalutamide or apalutamide.
- Rapidly progressing disease or significant symptoms related to disease progression.

Efficacy Endpoints

- **Primary:** Disease control evaluated as PSA decline or stabilization (PSA rise <25% over baseline) and no radiographic or clinical progression after 12 weeks of treatment.
- **Secondary:** Radiographic response per RECIST v1.1 criteria, time to PSA progression, and time to radiographic response.

FIGURE 1. TREATMENT SCHEDULE

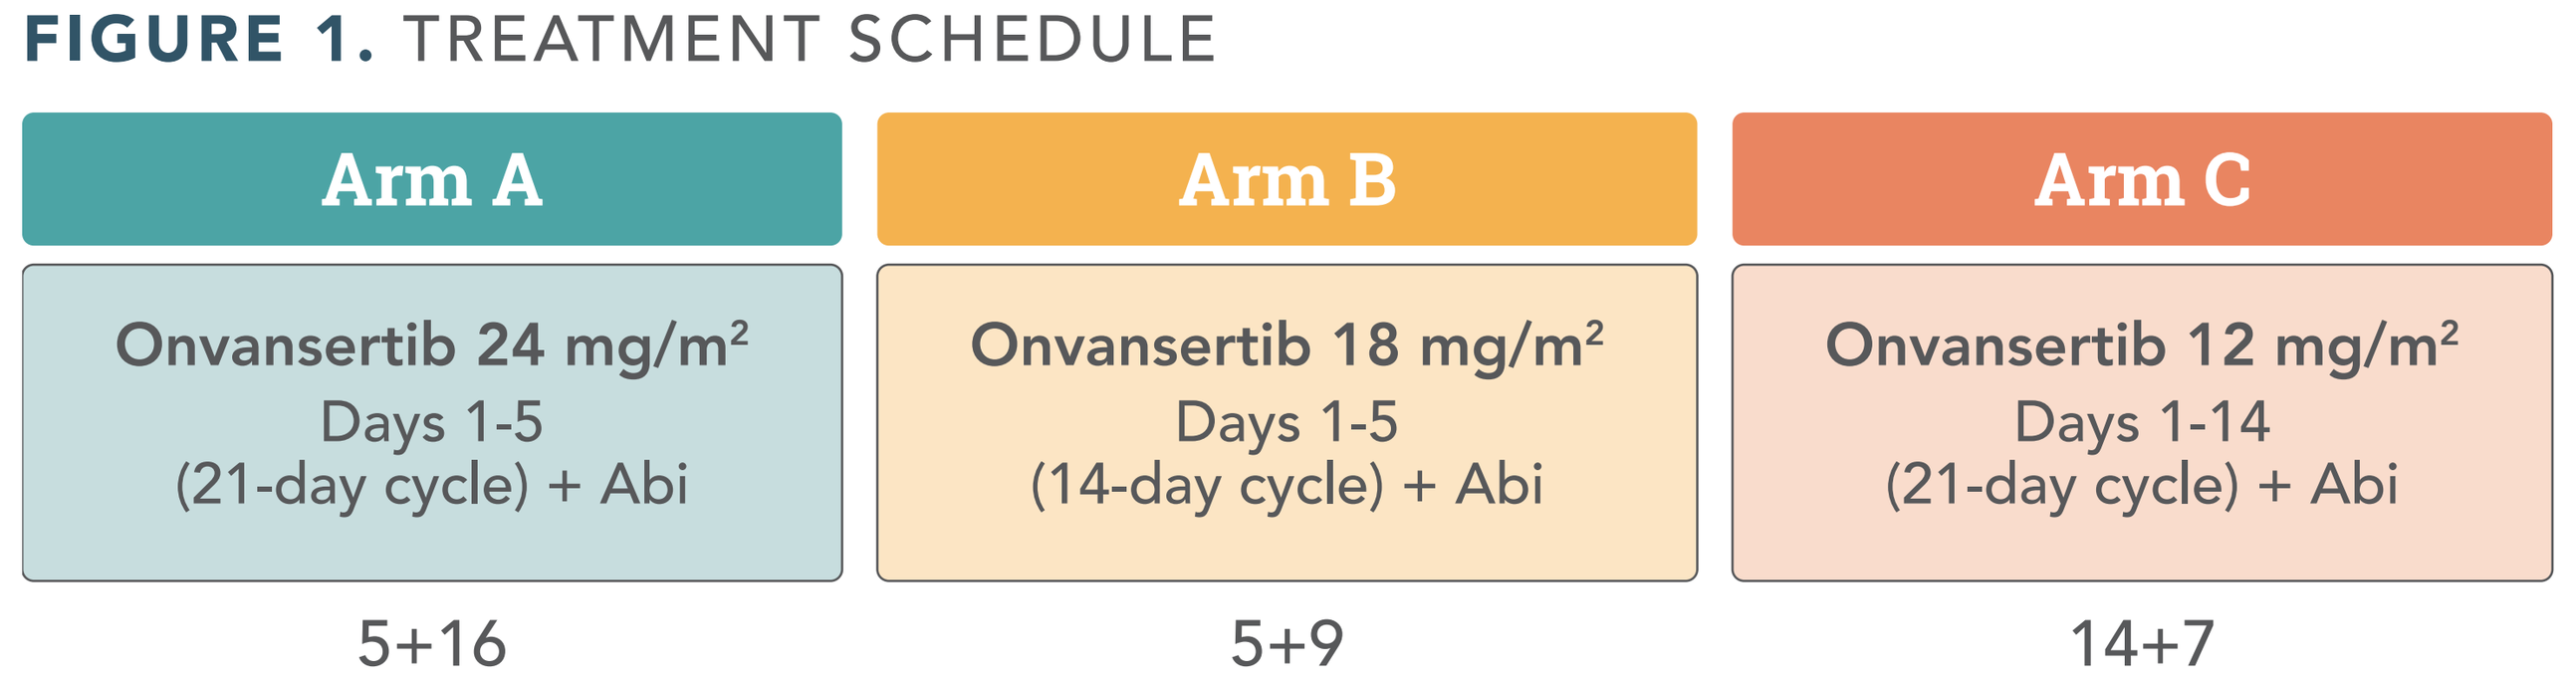
Arm A
Onvansertib 24 mg/m²
Days 1-5
(21-day cycle) + Abi
5+16
Arm B
Onvansertib 18 mg/m²
Days 1-5
(14-day cycle) + Abi
5+9
Arm C
Onvansertib 12 mg/m²
Days 1-14
(21-day cycle) + Abi
14+7

Correlative Analyses

FIGURE 2. ANALYSES OF CIRCULATING TUMOR DNA (ctDNA), CIRCULATING TUMOR CELLS (CTC) AND ARCHIVAL TISSUE TO IDENTIFY RESPONSE BIOMARKERS

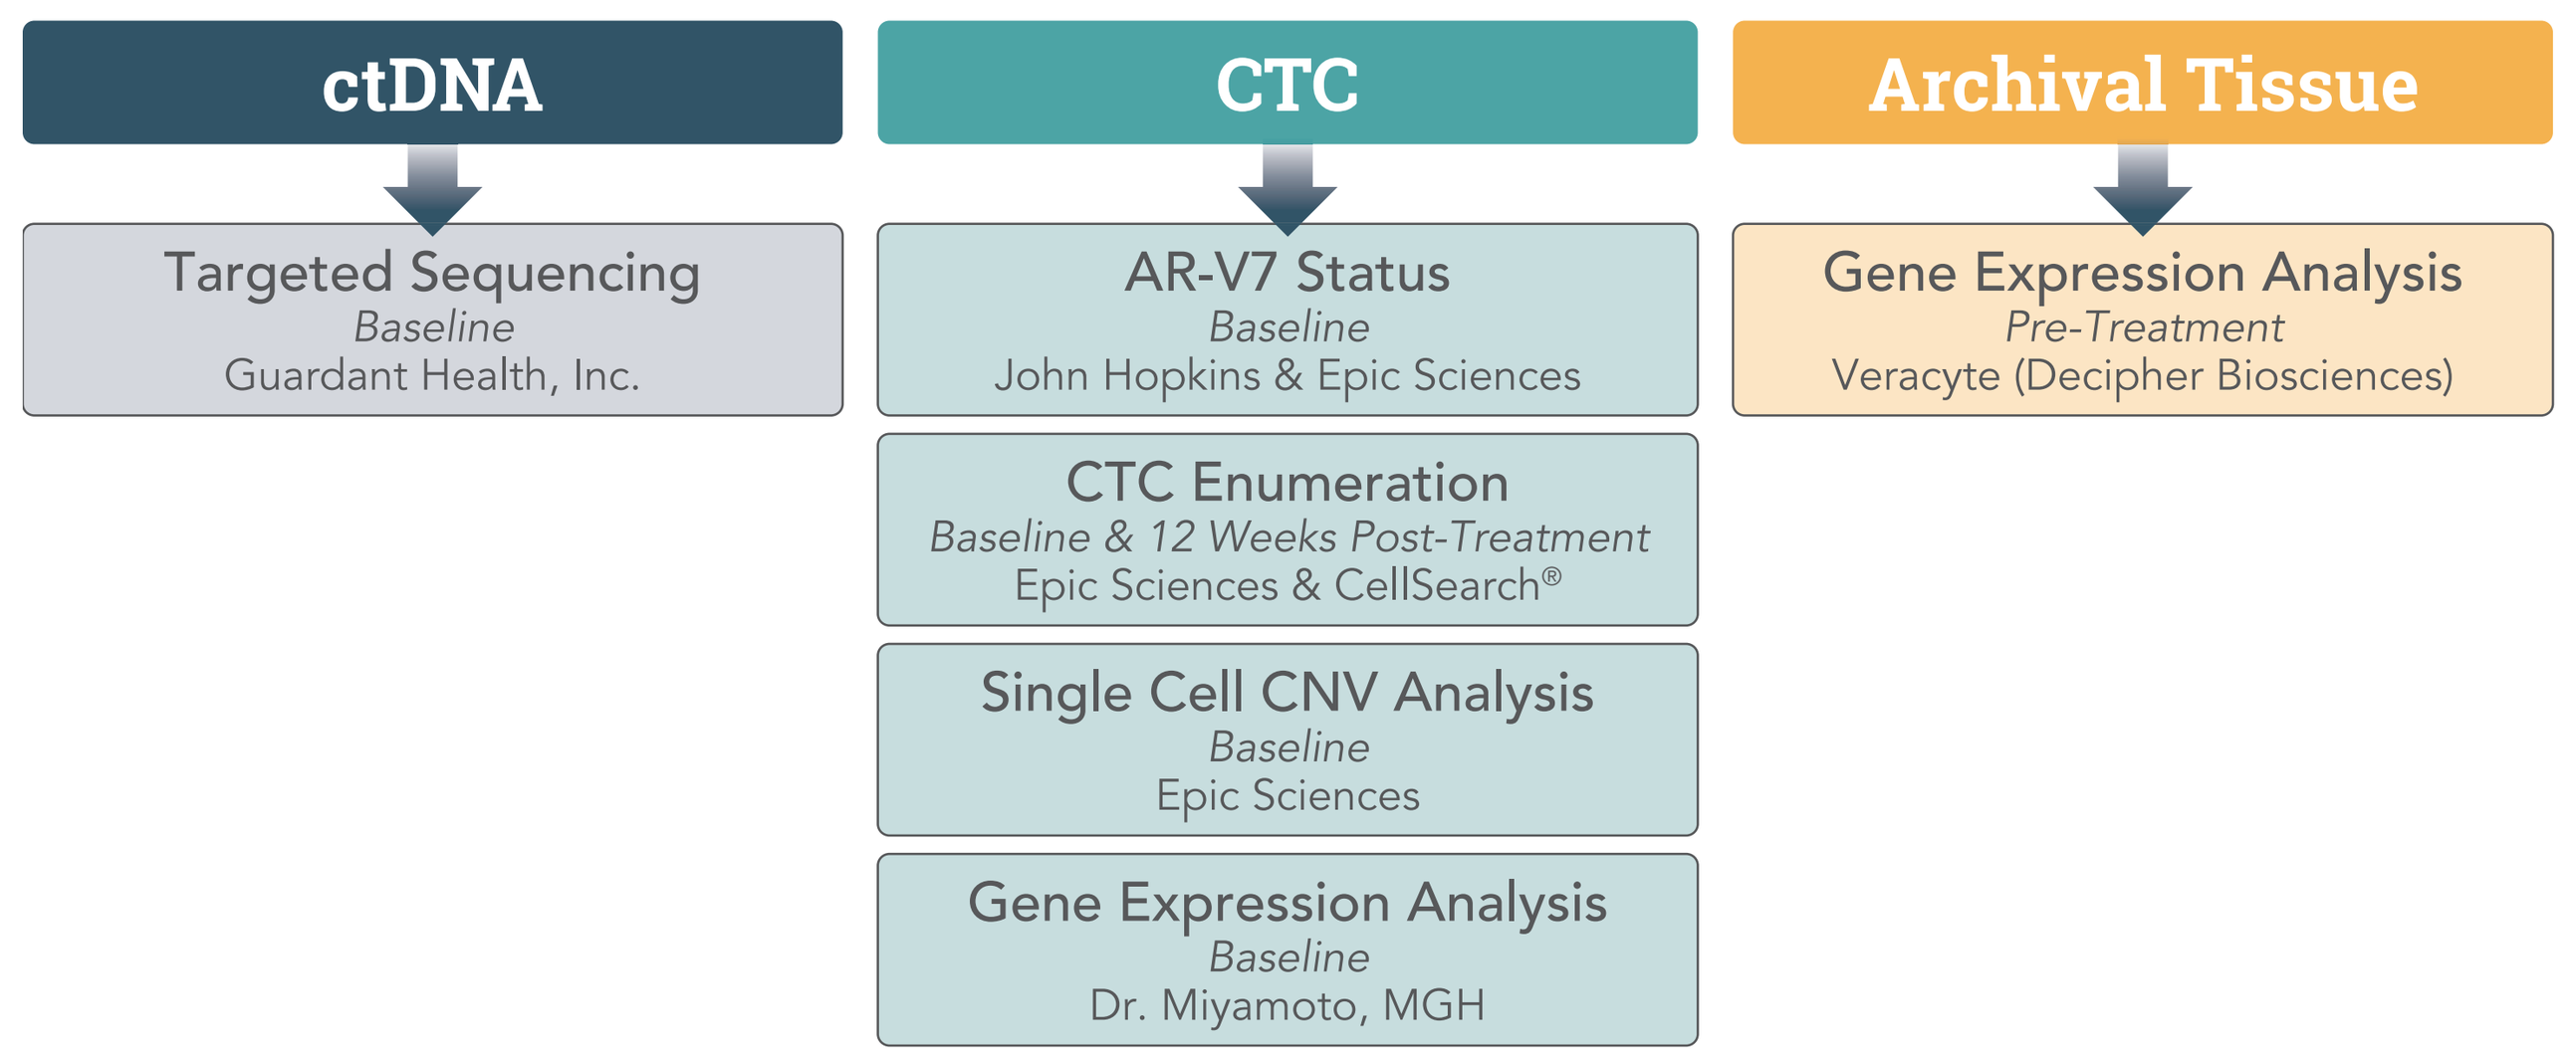
ctDNA
Targeted Sequencing
Baseline
Guardant Health, Inc.
CTC
AR-V7 Status
Baseline
John Hopkins & Epic Sciences
CTC Enumeration
Baseline & 12 Weeks Post-Treatment
Epic Sciences & CellSearch®
Single Cell CNV Analysis
Baseline
Epic Sciences
Gene Expression Analysis
Baseline
Dr. Miyamoto, MGH
Archival Tissue
Gene Expression Analysis
Pre-Treatment
Veracyte (Decipher Biosciences)

TABLE 1. ENROLLMENT AS OF 2-FEB-2022

	Arm A (5+16)	Arm B (5+9)	Arm C (14+7)	All Arms
Enrolled	24	20	24	68
On Treatment	0	3	11	14

Arms A, B and C have been added sequentially; Arms A and B are closed for enrollment, Arm C is enrolling.

Results

Efficacy

TABLE 2. EFFICACY ACROSS ARMS AS OF 2-FEB-2022

	Arm A (5+16)	Arm B (5+9)	Arm C (14+7)	All Arms
Evaluable for Efficacy*	17	19	20	56
PSA Stabilization at 12 Weeks**	5 (29%)	8 (42%)	9 (45%)	22 (39%)
Radiographic Stable Disease or Partial Response (SD/PR) at 12 Weeks	9 (53%)	11 (58%)	15 (75%)	35 (63%)

*Completed at least 12 weeks of treatment or had radiographic/clinical progression within 12 weeks.
**PSA rise <25% over baseline or less than 2 ng/mL.

Genomic Profiles

- Genomic analysis of ctDNA was performed at baseline[1] on 52 patients using Guardant OMNI™ assay (500-gene panel, 49 patients) or Guardant 360™ assay (74-gene panel, 3 patients).
- Somatic mutations or copy number variants (CNVs) were detected in all patients, with an average of 9.5 (range 1-52) alterations per patient.
- Most common alterations across patients were TP53 mutations (30/52, 57.7%), AR mutations/amplifications (20/52, 38.5%) and TMPRSS2 fusions (13/52, 25%, **Fig. 3**).

1. Blood samples collected pre-treatment, except for 2 patients who were analyzed using samples collected after the 1st cycle of treatment.

FIGURE 3. HEATMAP OF TOP INDIVIDUAL SOMATIC ALTERATIONS OBSERVED ACROSS PATIENTS

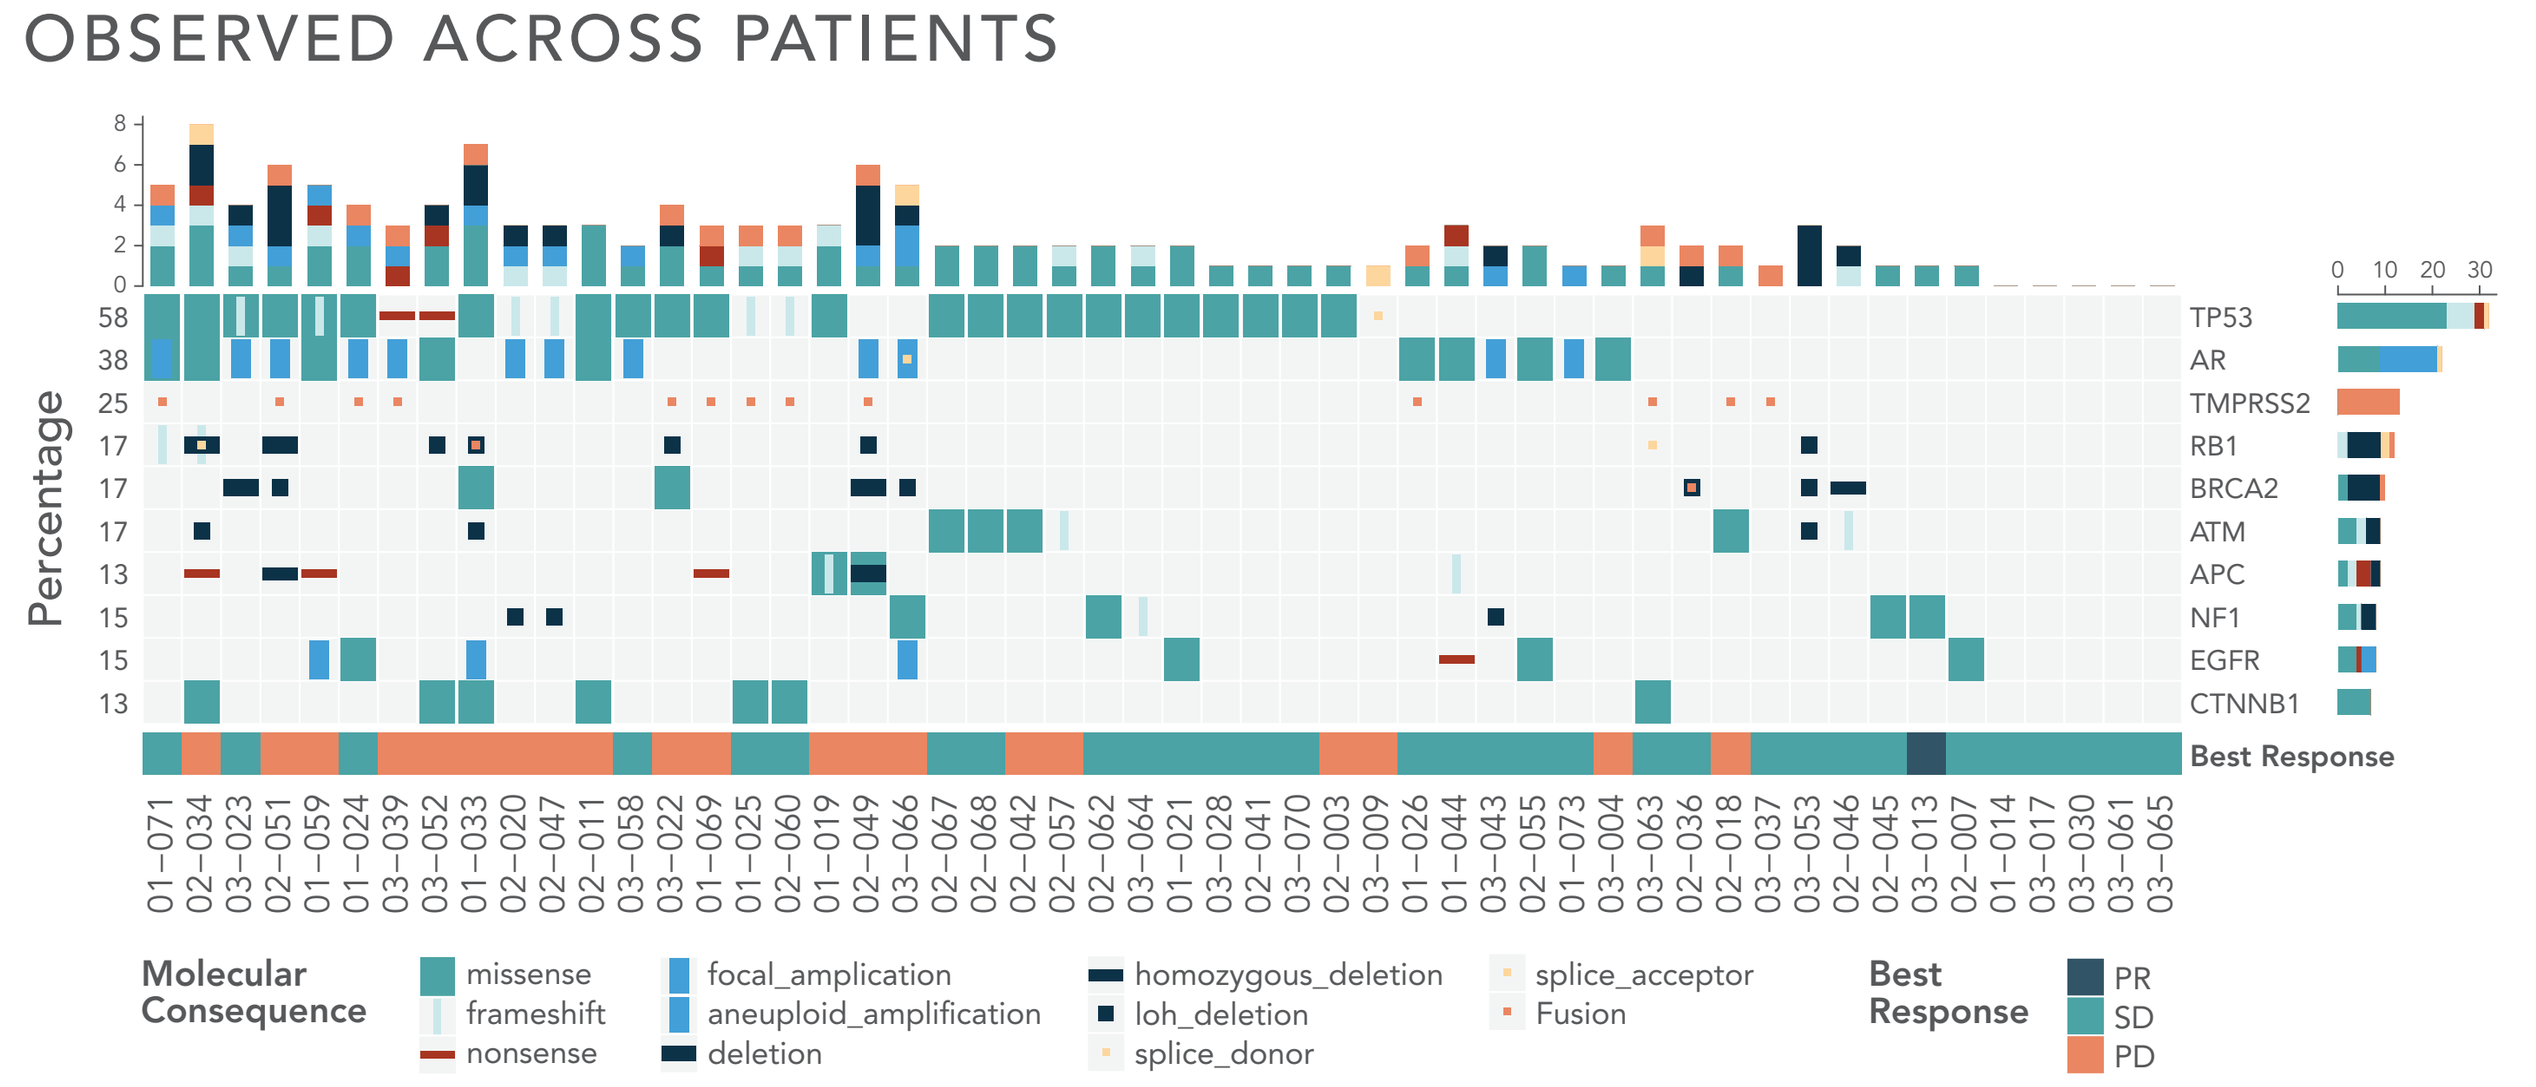
Percentage
TP53
AR
TMPRSS2
RB1
BRCA2
ATM
APC
NF1
EGFR
CTNNB1
Best Response
Molecular Consequence
missense
frameshift
nonsense
focal_amplication
aneuploid_amplification
deletion
homozygous_deletion
loh_deletion
splice_donor
splice_acceptor
Fusion
Best Response
PR
SD
PD

Genomic Alterations and Clinical Response

- Genomic profiles of patients who progressed within 12 weeks of treatment ("PD" patients, n=20) were compared to patients who had radiographic SD or PR at 12 weeks ("SD/PR" patients, n=32).
- On average, PD patients had more alterations than SD/PR patients (14.2 vs 6.6, p=0.015, **Fig. 4A**). Further, of the SNVs/InDels detected the mean variant allele frequency (VAF) in PD patients (n=133), was significantly higher (5.63% vs 1.16%, p=4.2e-05, **Fig. 4B**) when compared to SD/PR (n=171).
- SD/PR was positively associated with mutations in MTOR, FAT1, PTEN and FOXA1 and negatively associated with APC and PREX2 alterations (Chi-square test of independence, X-squared=213.09, df=46, p-value <2.2e-16, **Fig. 5**).
- 6/32 (19%) of SD/PR patients had an mTOR mutation vs none of the PD patients. Five of these 6 mutations were predicted to have a deleterious functional impact on the MTOR protein.

FIGURE 4. NUMBER OF SOMATIC ALTERATIONS AND VARIANT ALLELE FREQUENCY (VAF) OF SOMATIC SNV/InDEL'S DETECTED IN ctDNA

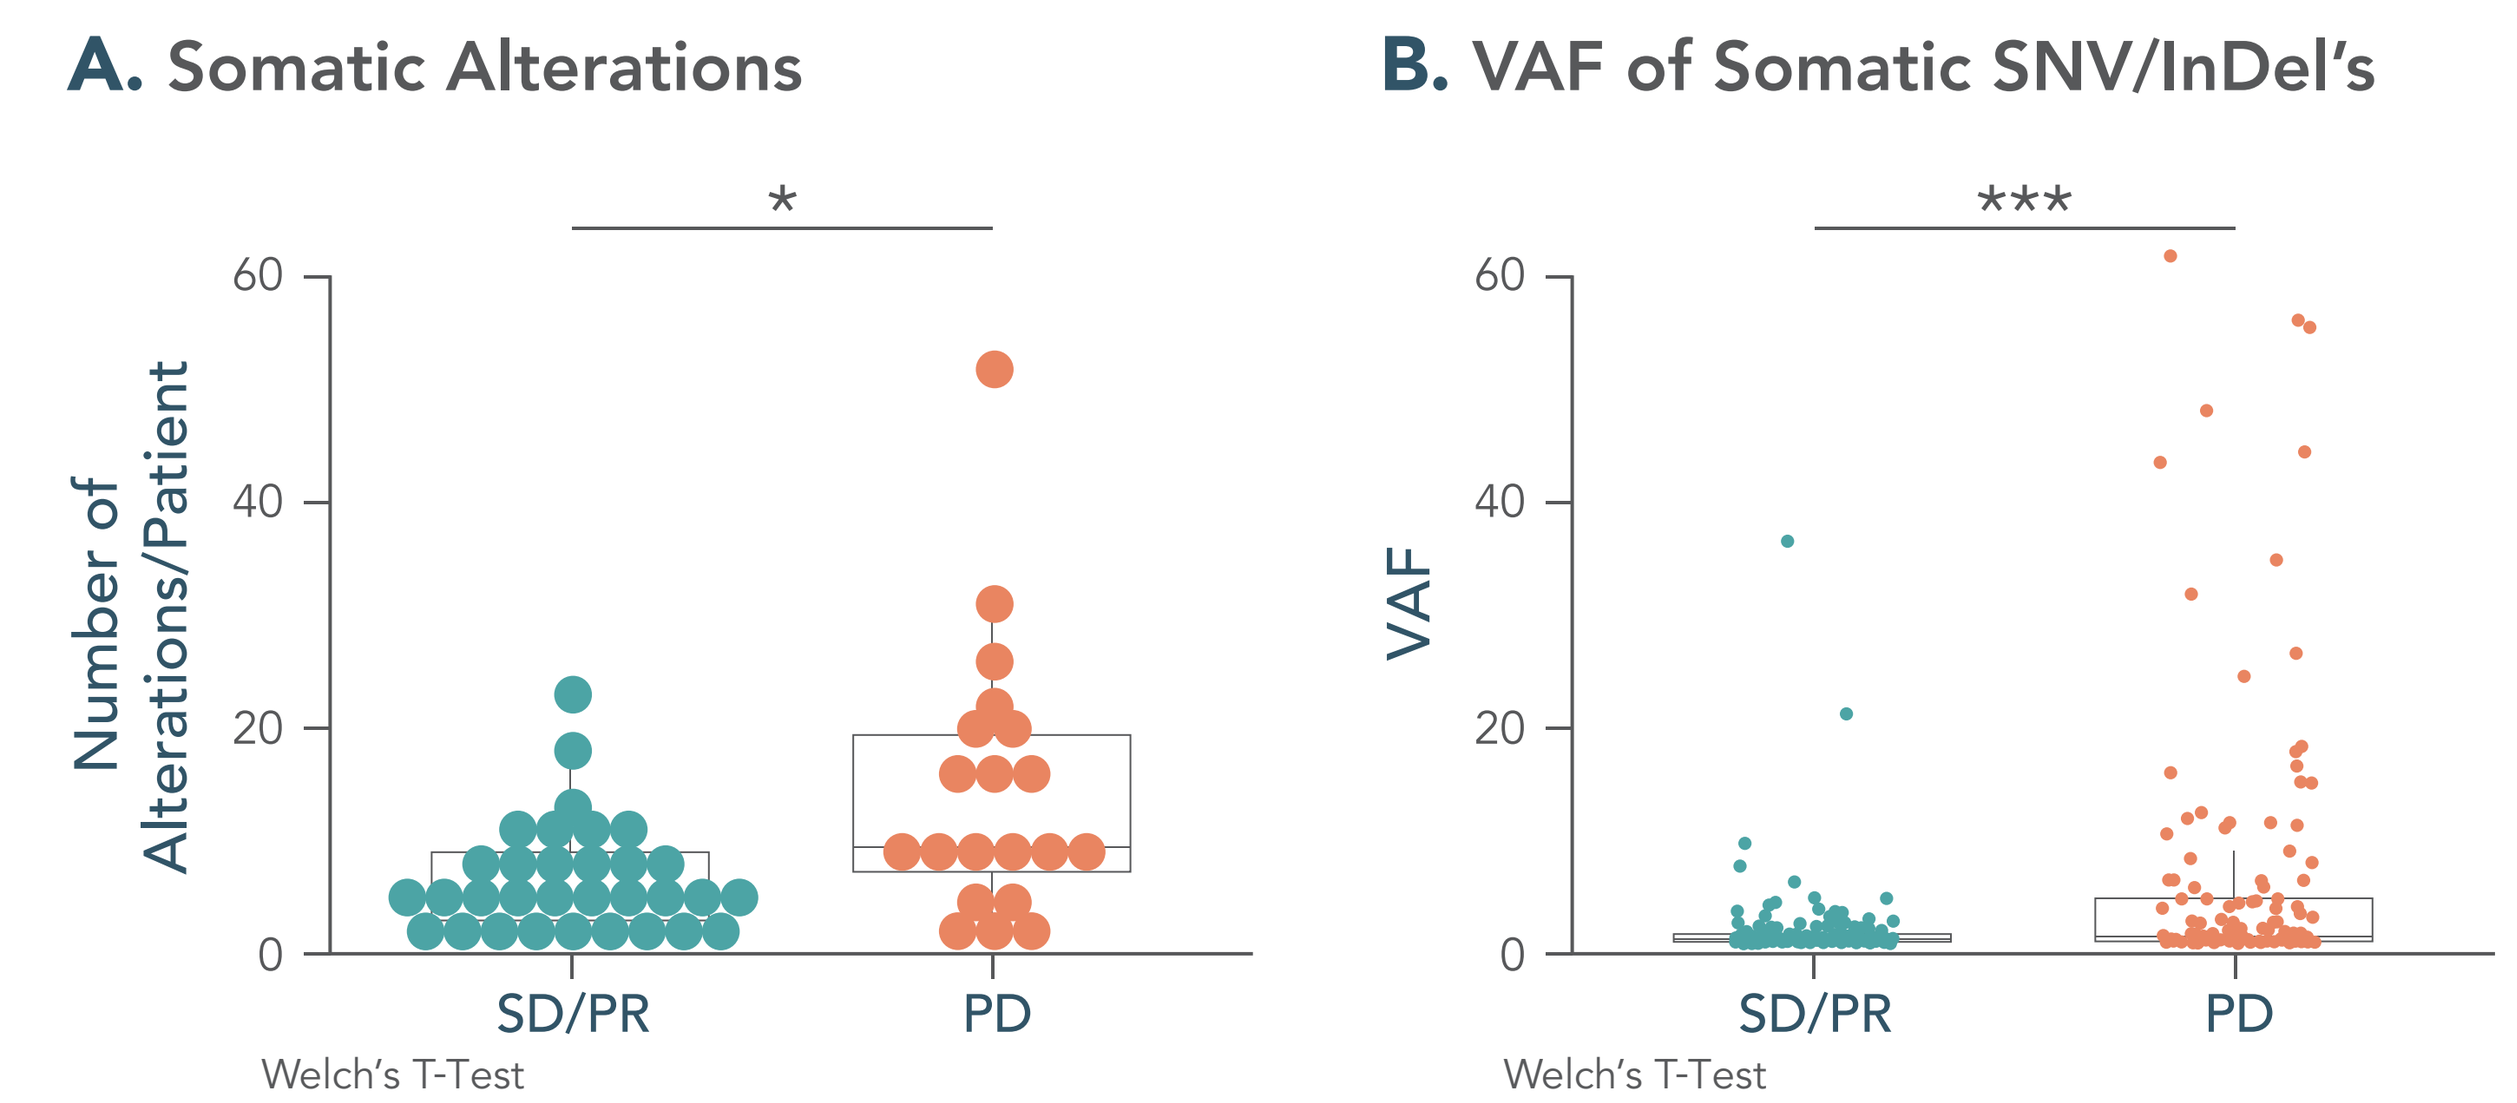
A. Somatic Alterations
*
Number of Alterations/Patient
SD/PR
PD
Welch's T-Test
B. VAF of Somatic SNV/InDel's

VAF
SD/PR
PD
Welch's T-Test

FIGURE 5. HEATMAP OF TOP INDIVIDUAL SOMATIC ALTERATIONS OBSERVED IN PATIENTS WITH SD/PR AND IN PATIENTS WITH PD

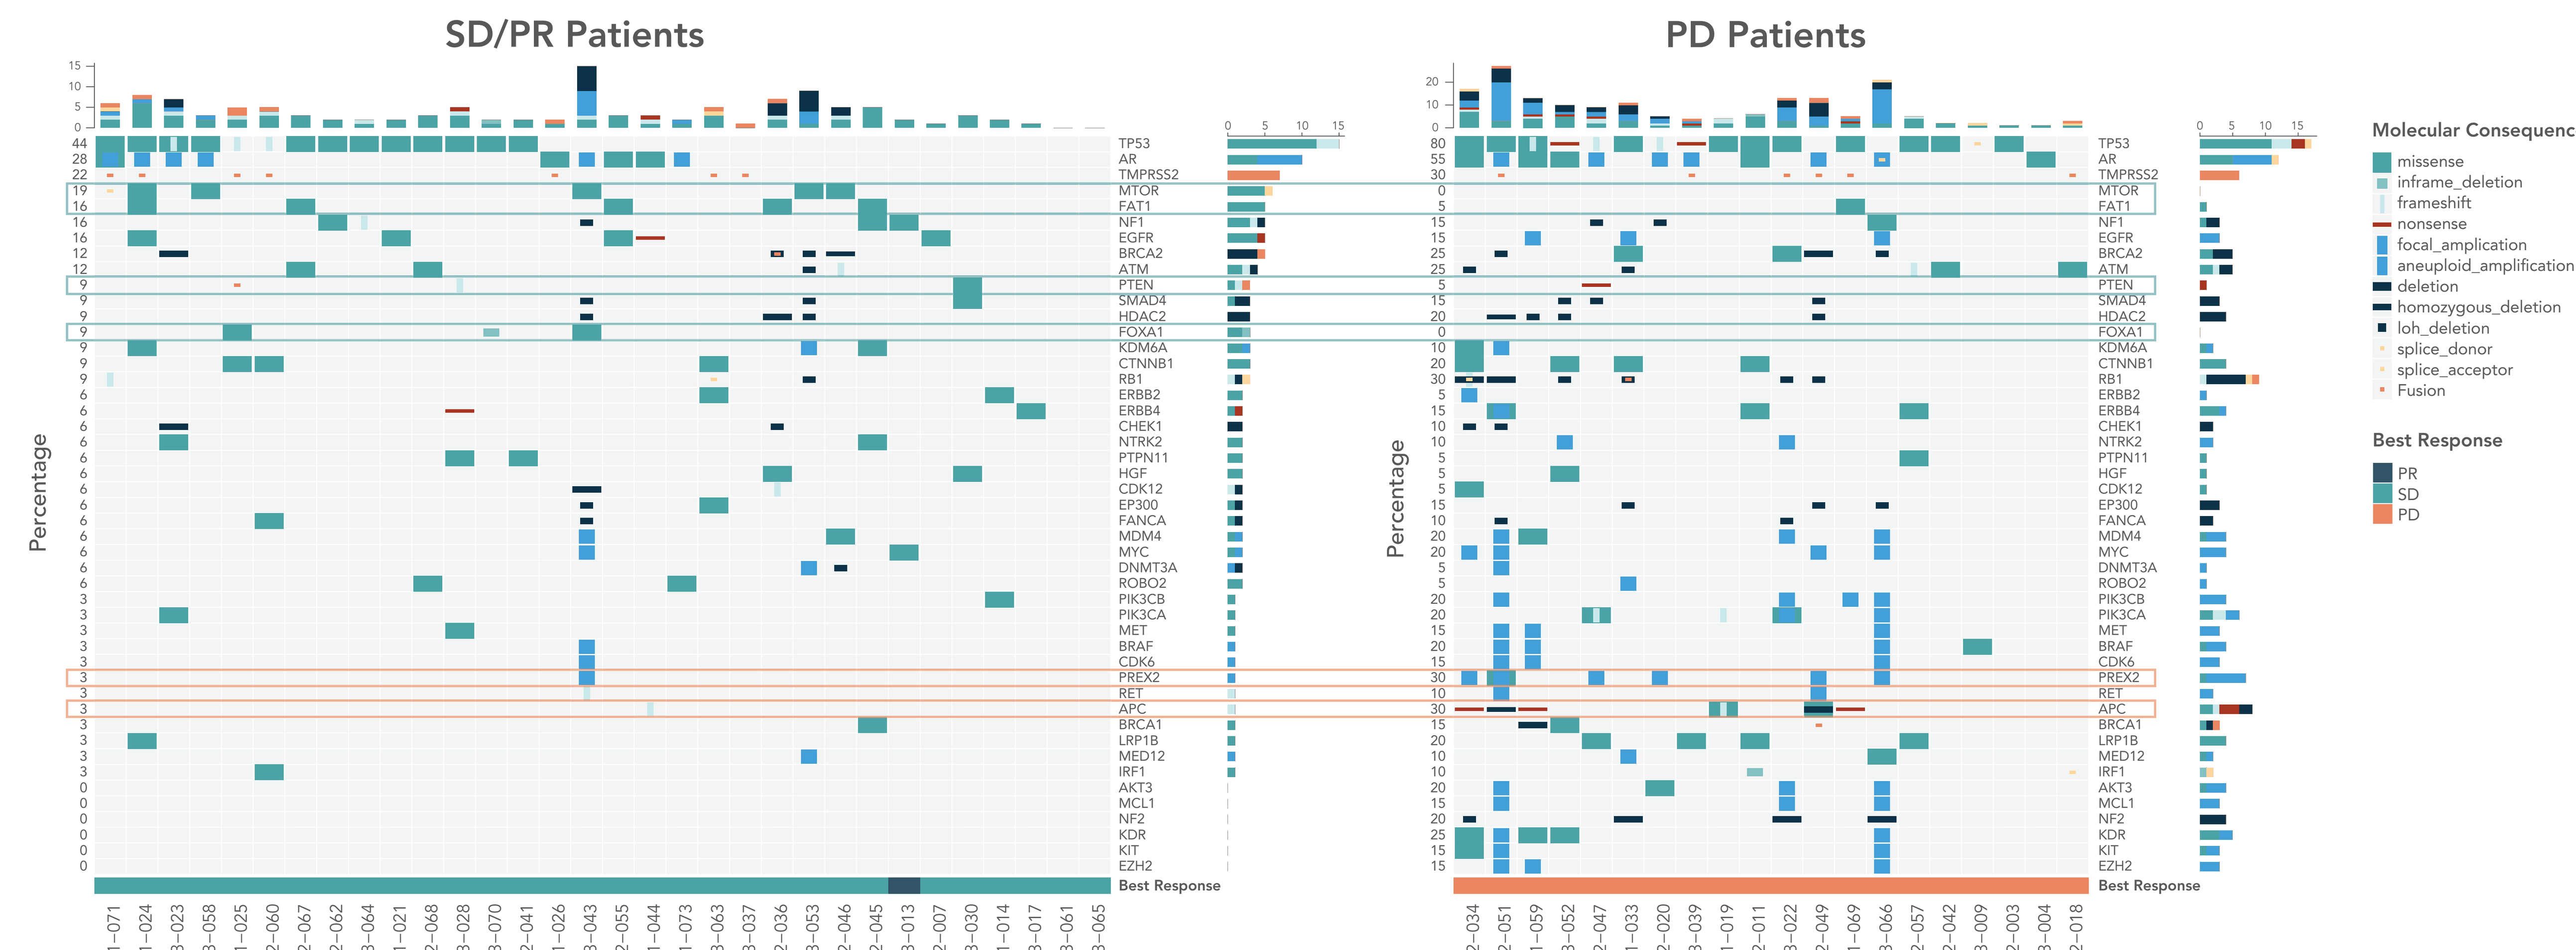
SD/PR Patients
PD Patients
Percentage
Best Response
Molecular Consequence
missense
inframe_deletion
frameshift
nonsense
focal_amplication
aneuploid_amplification
deletion
homozygous_deletion
loh_deletion
splice_donor
splice_acceptor
Fusion
Best Response
PR
SD
PD

Gene Expression and Clinical Response

- Gene expression data from archival tissues were obtained and analyzed from 14 patients; 8 patients with SD/PR and 6 patients with PD (**Fig. 6**).
- The gene signatures most strongly correlated with SD/PR were ERG+ signature and Notch signaling signature (**Table 3**); two pathways involved in cell invasion, epithelial-mesechymal transition (EMT) and metastasis.[1,2]
- Additionally, differential gene expression analysis and gene set enrichment analysis (GSEA) revealed that genes involved in mitochondrial functions and immune functions were downregulated in SD/PR patients in comparison with PD patients.

1. Adamo P. and Ladomery MR., *Oncogene* 2016 35(4); **2.** Zhang et al., *Cell Cycle* 2017 16(10).

FIGURE 6. TRANSCRIPTOMIC ANALYSIS

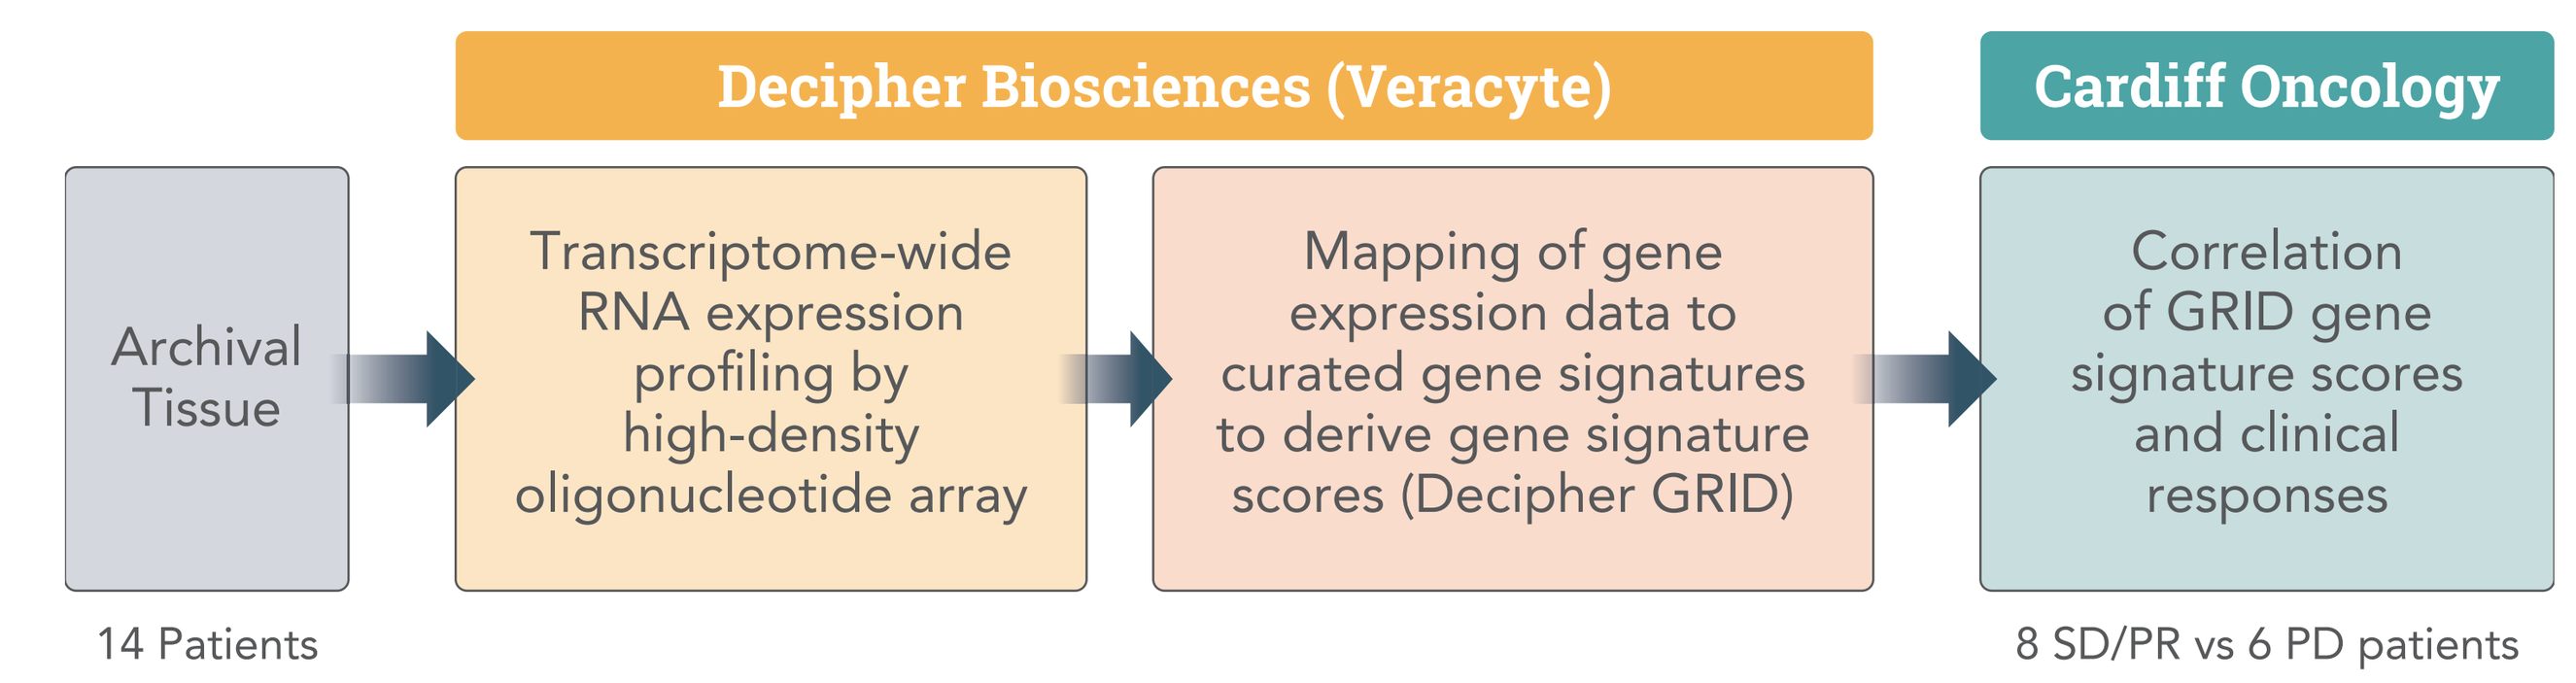
Decipher Biosciences (Veracyte)
Cardiff Oncology
Archival Tissue
Transcriptome-wide RNA expression profiling by high-density oligonucleotide array
Mapping of gene expression data to curated gene signatures to derive gene signature scores (Decipher GRID)
Correlation of GRID gene signature scores and clinical responses
14 Patients
8 SD/PR vs 6 PD patients

TABLE 3. GRID GENE SIGNATURES POSITIVELY CORRELATED WITH SD/PR CLINICAL OUTCOME

Signature Description	GRID Gene Signature	Bi-Serial Correlation	P-Value (permuted)	Significance Score
ERG+	ergmodel_1	0.71	0.012	1.35
Notch Signaling	hallmark_notch_signaling	0.56	0.023	0.92
AR Activity	aros_1	0.5	0.044	0.68
Presence of Activating FGFR3 Mutation	sjodahl2012_fgfr3	0.48	0.04	0.67
Prostate Cancer vs Bladder Cancer	pca_vs_mibc_1	0.41	0.027	0.63
Predicted Response to CTLA-4	immunophenoscore_1_HLA_DPA1	0.45	0.047	0.6

Conclusions

- Genomic analysis of ctDNA showed a correlation between alterations in two key genes of the PI3K signaling pathway—MTOR and PTEN, and sensitivity to onvansertib/abi combination in mCRPC patients with early abi-resistance.
- Preliminary gene expression analysis using archival tissues suggested an association between response to onvansertib/abi and expression of genes/pathways related to EMT, mitochondrial and immune functions. Analysis of additional patient samples is warranted to confirm these findings.
- Additional correlative analyses such as single-cell CNV and gene expression analyses of CTCs are underway to further identify or confirm response biomarkers for the onvansertib/abi combination.

Contact: Maya Ridinger PhD, mridinger@cardiffoncology.com; **Presented at:** AACR 2022 – Biomarkers Predictive of Therapeutic Benefit 1