FPN: 366P

The PLK1 Inhibitor Onvansertib Overcomes Irinotecan Resistance in RAS-Mutated Metastatic Colorectal Cancer (mCRC) *In Vivo* and in Patients

S Kopetz MD PhD[1], M Ridinger PhD[2], AV Sorokin PhD[1], P Kanikarla PhD[1], F Gao[1], Z Liu MD[1], E Samuelsz[2], T Smeal PhD[2], JS Starr DO[3], MR Sharma MD[4]

[1]Department of Gastrointestinal Medical Oncology, The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Cardiff Oncology, San Diego, CA; [3]Department of Oncology, Mayo Clinic, Jacksonville, FL; [4]Cancer and Hematology Centers of Western Michigan, Grand Rapids, MI

Background

- Chemotherapy based on fluoropyrimidines and oxaliplatin (FOLFOX) or irinotecan (FOLFIRI) remains 1st- and 2nd-line standard-of-care (SoC) for KRAS-mutated mCRC patients.
- Chemotherapy resistance is present or develops in most patients and overcoming resistance constitutes a highly unmet medical need.[1]
- Approved 3rd-line treatments (regorafenib, TAS-102) provide limited benefit with a PFS of 2-3 months and OS of 6-8 months.[2]
- Onvansertib, is a highly specific PLK1 inhibitor, that demonstrated preliminary safety and efficacy in combination with FOLFIRI/bevacizumab (bev) in 2nd-line (2L) treatment of KRAS-mutated mCRC patients who failed or were intolerant to fluoropyrimidine and oxaliplatin (NCT03829410).[3]
- An Expanded Access Program (EAP) was opened to provide access to onvansertib + FOLFIRI/bev to KRAS-mutated mCRC patients, who had failed or progressed on SoC, including irinotecan (NCT04446793).[4]

We aimed at evaluating the potential of onvansertib to overcome resistance to irinotecan-based therapies in RAS-mutated CRC by:

1. **Assessing the anti-tumor activity of onvansertib + irinotecan in irinotecan-resistant patient derived xenograft (PDX) models.**
2. **Assessing the clinical benefit of onvansertib + FOLFIRI/bev in EAP patients with prior irinotecan treatment and the utility of circulating tumor DNA (ctDNA) as a response biomarker.**

1. Longley and Johnston *J Pathol.* 2005, 205(2); **2.** Bekaii-Saab et al., *Clin Colorectal Cancer* 2019, 18(1); **3.** Lenz et al. *JCO* 2022, 40(4_suppl); **4.** Sharma et al., *Cancer Res* 2021, 81 (13_suppl).

Methods

Preclinical Studies

- CRC PDX models (**Table 1**):
 - RAS-mutated (5 KRAS, 1 NRAS).
 - Intrinsic resistance to irinotecan (n=4) or acquired-resistance (n=2).
- Treatment: after tumors reached a mean volume of 200-250 mm^3, mice (5-7/group) were treated for 21 days with vehicle, onvansertib (oral, 60 mg/kg, daily), irinotecan (intraperitoneal, 40 mg/kg, weekly) or the combination of onvansertib + irinotecan.
- Tumor volumes were measured twice a week and tumor volume changes from baseline were calculated.
- An unpaired t-test was used to test statistical differences between onvansertib and onvansertib + irinotecan treatment groups at Day 21.

TABLE 1. CHARACTERISTIC OF PDX MODELS

PDX Model	RAS Mutation	Irinotecan Resistance
B8141R	NRAS Q61R	Induced
C1177R	KRAS G12C	Induced
C1143	KRAS G12D	Intrinsic
B8086	KRAS G12V	Intrinsic
B8182	KRAS G12C	Intrinsic
C1144	KRAS G12C	Intrinsic

Clinical Study

Expanded Access Program

- **Eligibility:** mCRC patients with confirmed KRAS mutation, who failed or progressed on multiple lines of SoC systemic therapy including irinotecan.
- Treatment (28-day cycle): onvansertib 15 mg/m^2 on Days 1-5 and 15-19 in combination with FOLFIRI/bev (Days 1 and 15).
- **Objectives:** safety (primary), progression-free survival (PFS), changes in ctDNA after 1 treatment cycle using liquid biopsies.

ctDNA Analysis

- Blood samples were collected at baseline (C1D1) and after 1 cycle of treatment (C2D1, ≈4 weeks) to measure changes in ctDNA (**Figure 1**):
 - KRAS mutant allele frequency (MAF) was assessed by digital droplet PCR (ddPCR).
 - Guardant360® assay, a NGS-based liquid biopsy panel covering 74 cancer genes, was used to measure the mean MAF of somatic SNVs, insertions/deletions, and gene fusions.[1]

1. Mak et al., *Cancer Res* 2021, 81 (13_suppl).

FIGURE 1. WORKFLOW OF ctDNA ANALYSIS

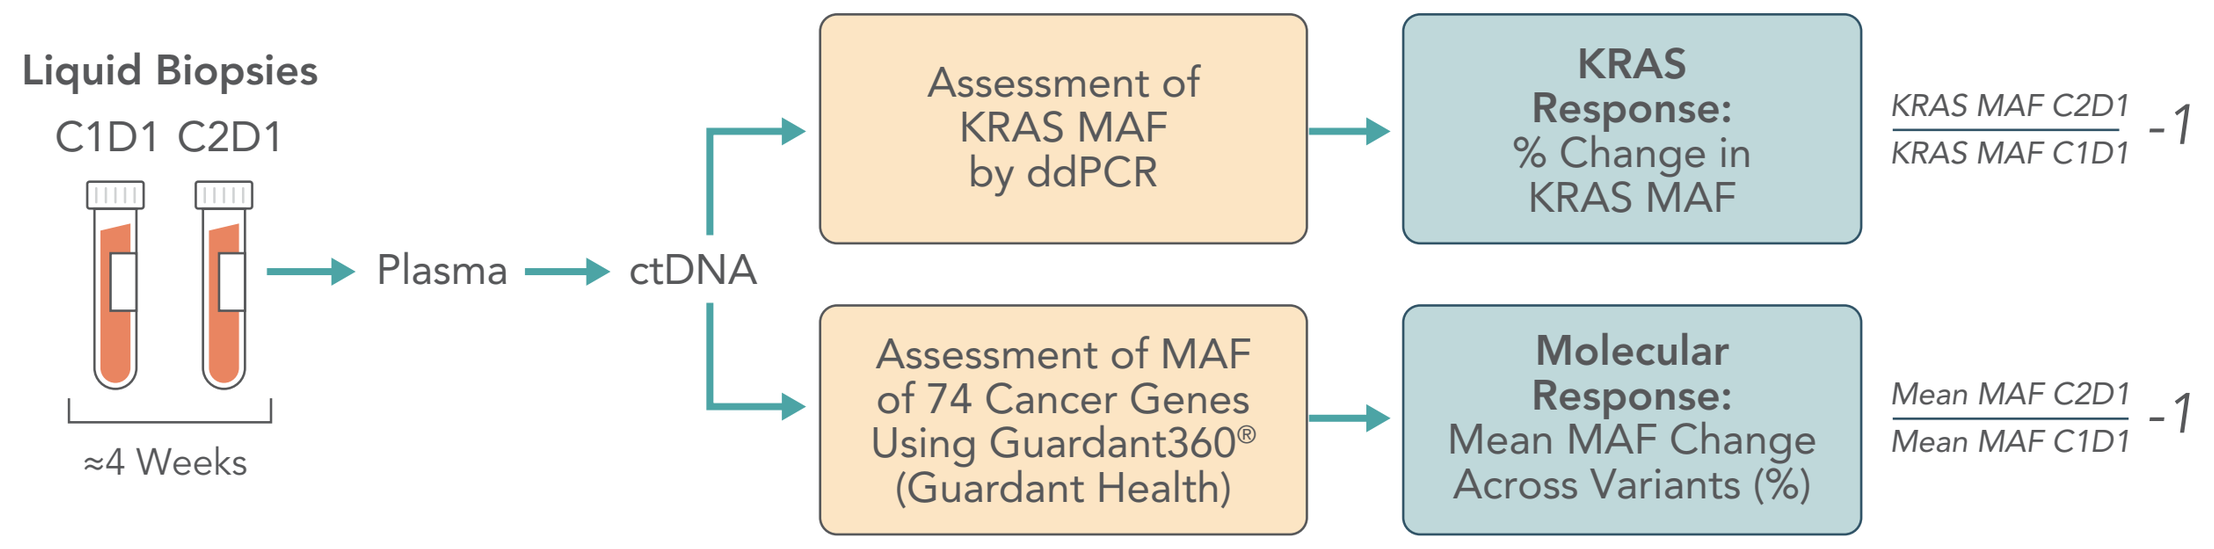

Liquid Biopsies
C1D1 C2D1
≈4 Weeks
Plasma
ctDNA
Assessment of KRAS MAF by ddPCR
Assessment of MAF of 74 Cancer Genes Using Guardant360® (Guardant Health)
KRAS Response: % Change in KRAS MAF
Molecular Response: Mean MAF Change Across Variants (%)
KRAS MAF C2D1 / KRAS MAF C1D1 - 1
Mean MAF C2D1 / Mean MAF C1D1 - 1

Results – Preclinical Studies

- As expected, all models were resistant to irinotecan, with a tumor size increase at D21 of at least 100% from baseline (**Table 2**).
- The combination of onvansertib + irinotecan showed anti-tumor activity in RAS-mutated PDX models with acquired and intrinsic resistance to irinotecan (**Table 2** and **Figure 2**):
 - 2 models showed tumor regression.
 - 3 models showed tumor stabilization.
 - 1 model showed a tumor volume increase >100% from baseline, which represented a 72% tumor growth inhibition from vehicle (partial inhibition).
- Importantly, the combination of onvansertib + irinotecan showed significant increased anti-tumor activity compared to onvansertib single agent in 5 of the 6 models (**Figure 2**).
- All together, this data supports that onvansertib + irinotecan is an active combination in RAS-mutated PDX models with acquired or intrinsic resistance to irinotecan and that onvansertib can re-sensitize tumors to irinotecan treatment.

TABLE 2. MEAN TUMOR VOL. CHANGE AT D21 FROM BASELINE

Model	Vehicle	Irinotecan	Onvansertib	Combination
B8086	518	324	-64	-68
C1143	383	112	28	-8
C1177R	315	216	79	15
C1144	200	183	125	19
B8141R	344	121	127	28
B8182	760	466	398	143

FIGURE 2. EFFECT OF TREATMENTS ON TUMOR VOLUME CHANGE IN 6 RAS-MUTATED CRC PDX MODELS

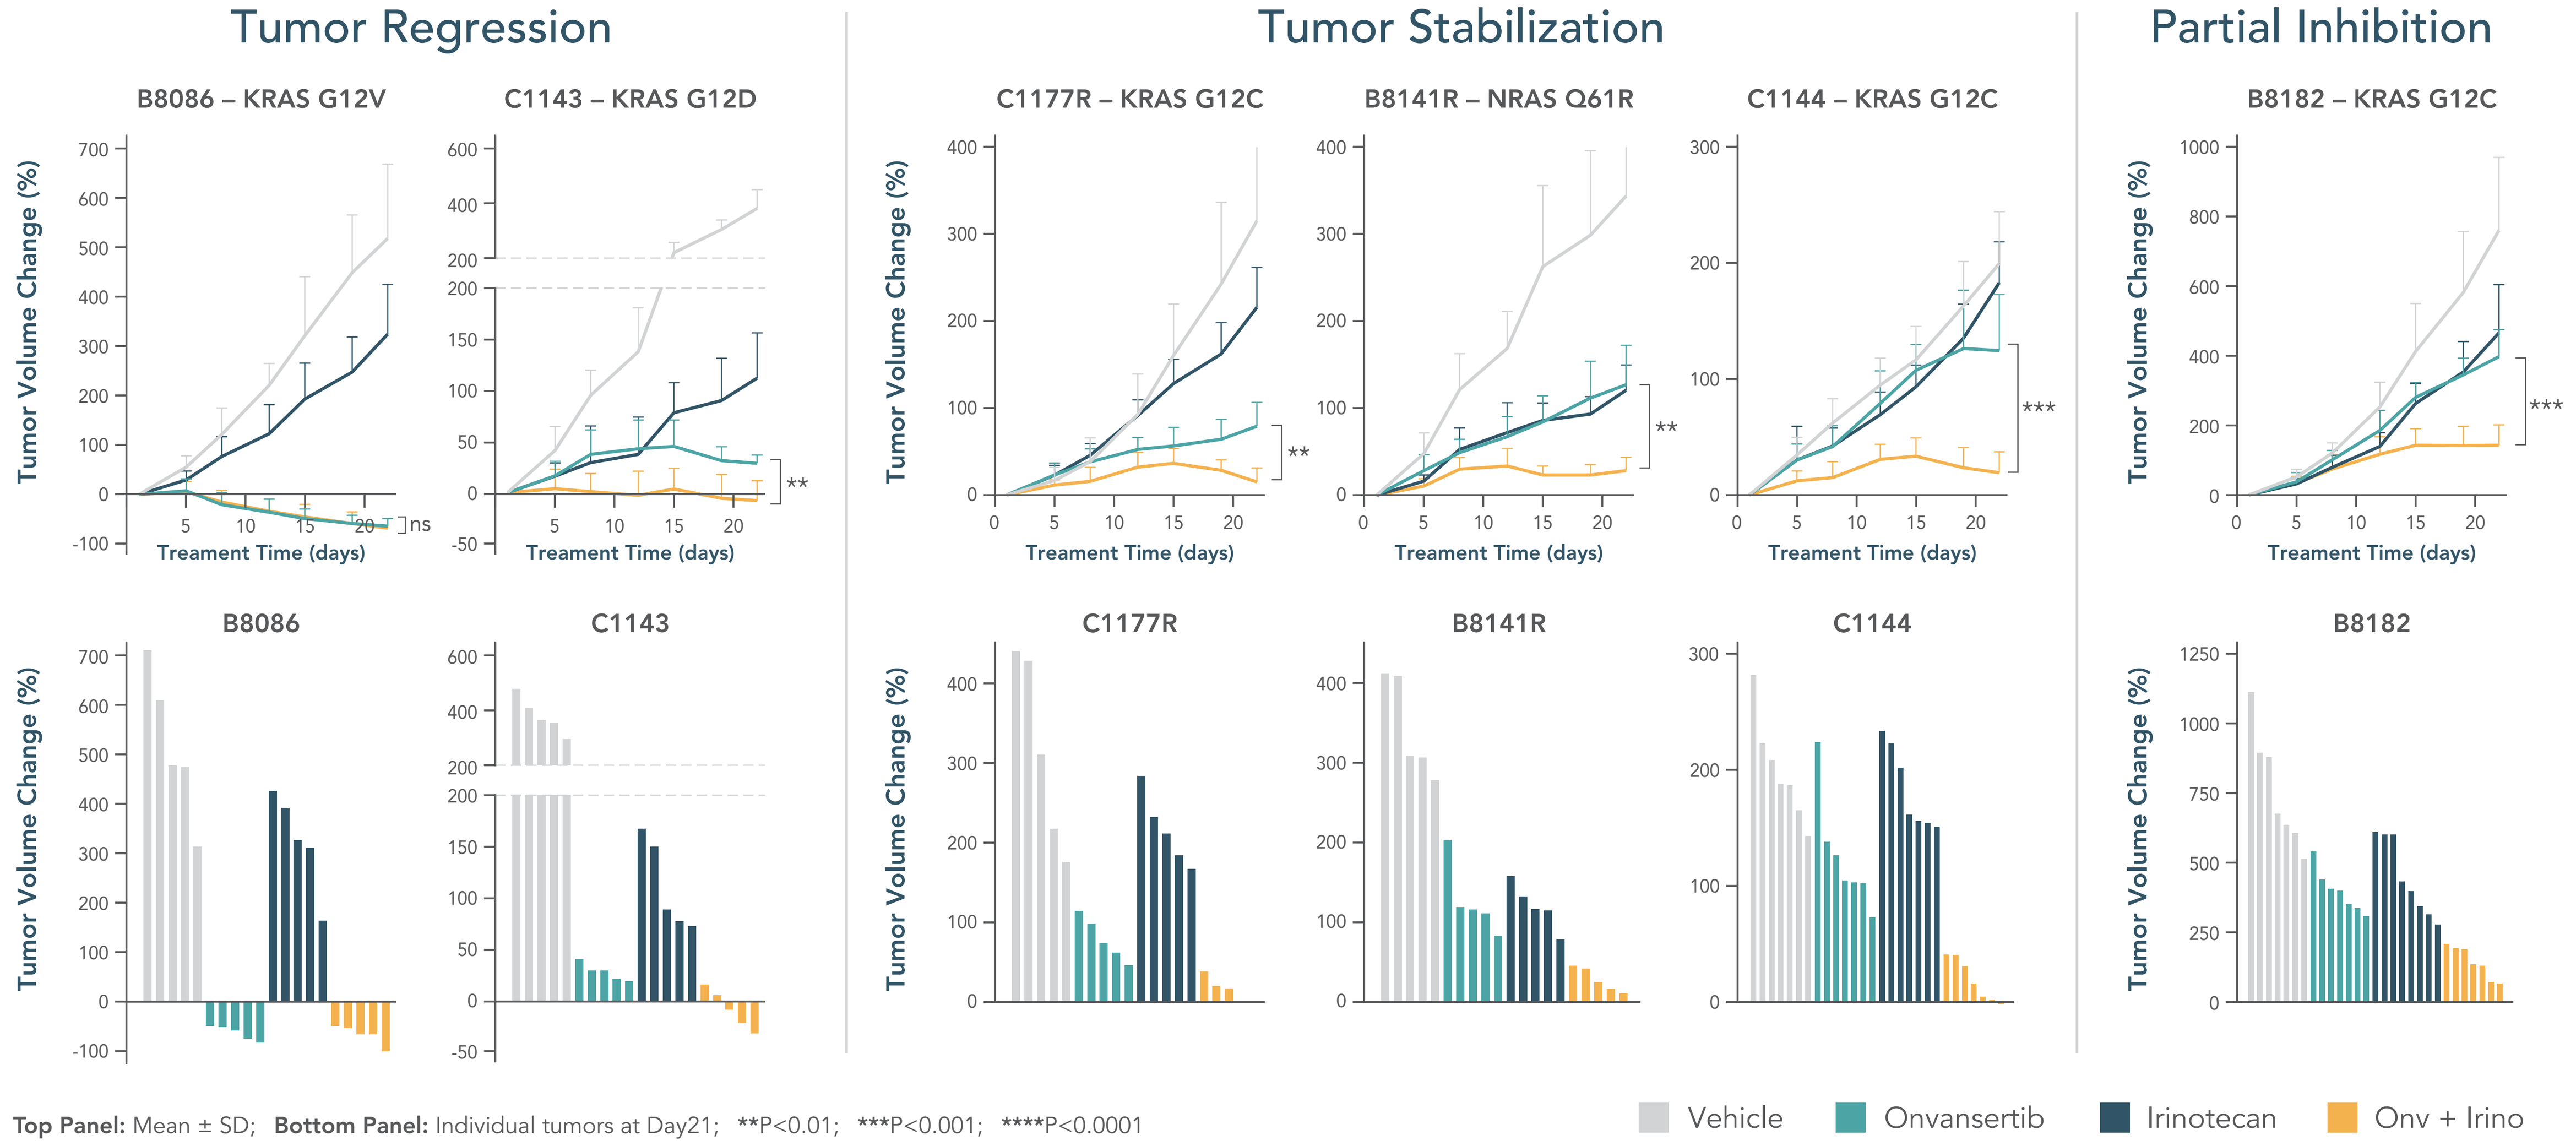

Tumor Regression
Tumor Stabilization
Partial Inhibition
B8086 – KRAS G12V
C1143 – KRAS G12D
C1177R – KRAS G12C
B8141R – NRAS Q61R
C1144 – KRAS G12C
B8182 – KRAS G12C
Tumor Volume Change (%)
Treament Time (days)
ns
**

B8086
C1143
C1177R
B8141R
C1144
B8182
Vehicle
Onvansertib
Irinotecan
Onv + Irino

Top Panel: Mean ± SD; **Bottom Panel:** Individual tumors at Day21; **P<0.01; ***P<0.001; ****P<0.0001

Conclusions

- Onvansertib + irinotecan is an active combination in RAS-mutated PDX models with acquired or intrinsic resistance to irinotecan. The combination had significantly increased anti-tumor activity compared to onvansertib single agent, supporting that onvansertib can overcome irinotecan-resistance.
- KRAS-mutated mCRC patients with prior irinotecan treatment showed a clinical benefit to onvansertib + FOLFIRI/bev treatment, in particular patients with early ctDNA decreases.
- All together this data supports the use of onvansertib + FOLFIRI/bev in RAS-mutated mCRC patients who progressed on SoC, and the utility of assessing early changes in ctDNA to identify patients who are more likely to benefit from therapy.

Results – Clinical Study

EAP Patients

- 51 patients were enrolled in the EAP between JUL-2020 and JUN-2021.
- 43 (84%) of patients had received irinotecan in a prior line of treatment.
- As of 05-AUG-2022, EAP patients with prior irinotecan treatment had a PFS of 4.04 months [CI: 2.96-8.38] and a 6-month PFS of 37.3% [CI: 24.9-55.8].

Changes in ctDNA are Associated with Increase Clinical Benefit

- In the Phase1b/2 of onvansertib + FOLFIRI/bev in 2L treatment of KRAS-mutated mCRC, patients with a ≥90% decrease in KRAS MAF after 1 treatment cycle ("KRAS Responders") had significantly higher ORR and longer PFS than KRAS Non-responders.[1]
- We tested whether early changes in KRAS MAF could be used to identify EAP patients with increased clinical benefit.
- 32 EAP patients with prior irinotecan treatment, were evaluable for KRAS Response[2]; 11 (34%) patients were determined to be KRAS Responders (i.e., ≥90% decrease in KRAS MAF after 1 treatment cycle).
- **KRAS Responders had significantly longer PFS and higher 6-month PFS than KRAS Non-responders (p=0.0014) (Table 3** and **Figure 3**).

1. Lenz et al, ESMO 2022, poster 397P; **2.** Defined as patients with C1D1 and C2D1 plasma samples and detectable baseline KRAS MAF.

TABLE 3. EFFICACY OF ONVANSERTIB + FOLFIRI/BEV IN KRAS RESPONDERS AND NON-RESPONDERS

	KRAS Responder	KRAS Non-Responder	All Patients Evaluable for KRAS Response	All Patients with Prior Irinotecan
Patients (n)	11 (34%)	21 (66%)	32	43
Median PFS [CI] (months)	11.18 [5.19-NR]	3.25 [2.20-6.64]	3.98 [3.25-7.43]	4.04 [2.96-8.38]
6-Month PFS [CI] (%)	54.5 [31.8-93.6]	23.8 [11.1-51.2]	33.9 [20.8-55.2]	37.3 [24.9-55.8]

NR: Not reached; **CI:** 95% confidence intervals.

FIGURE 3. PFS OF KRAS RESPONDERS AND NON-RESPONDERS

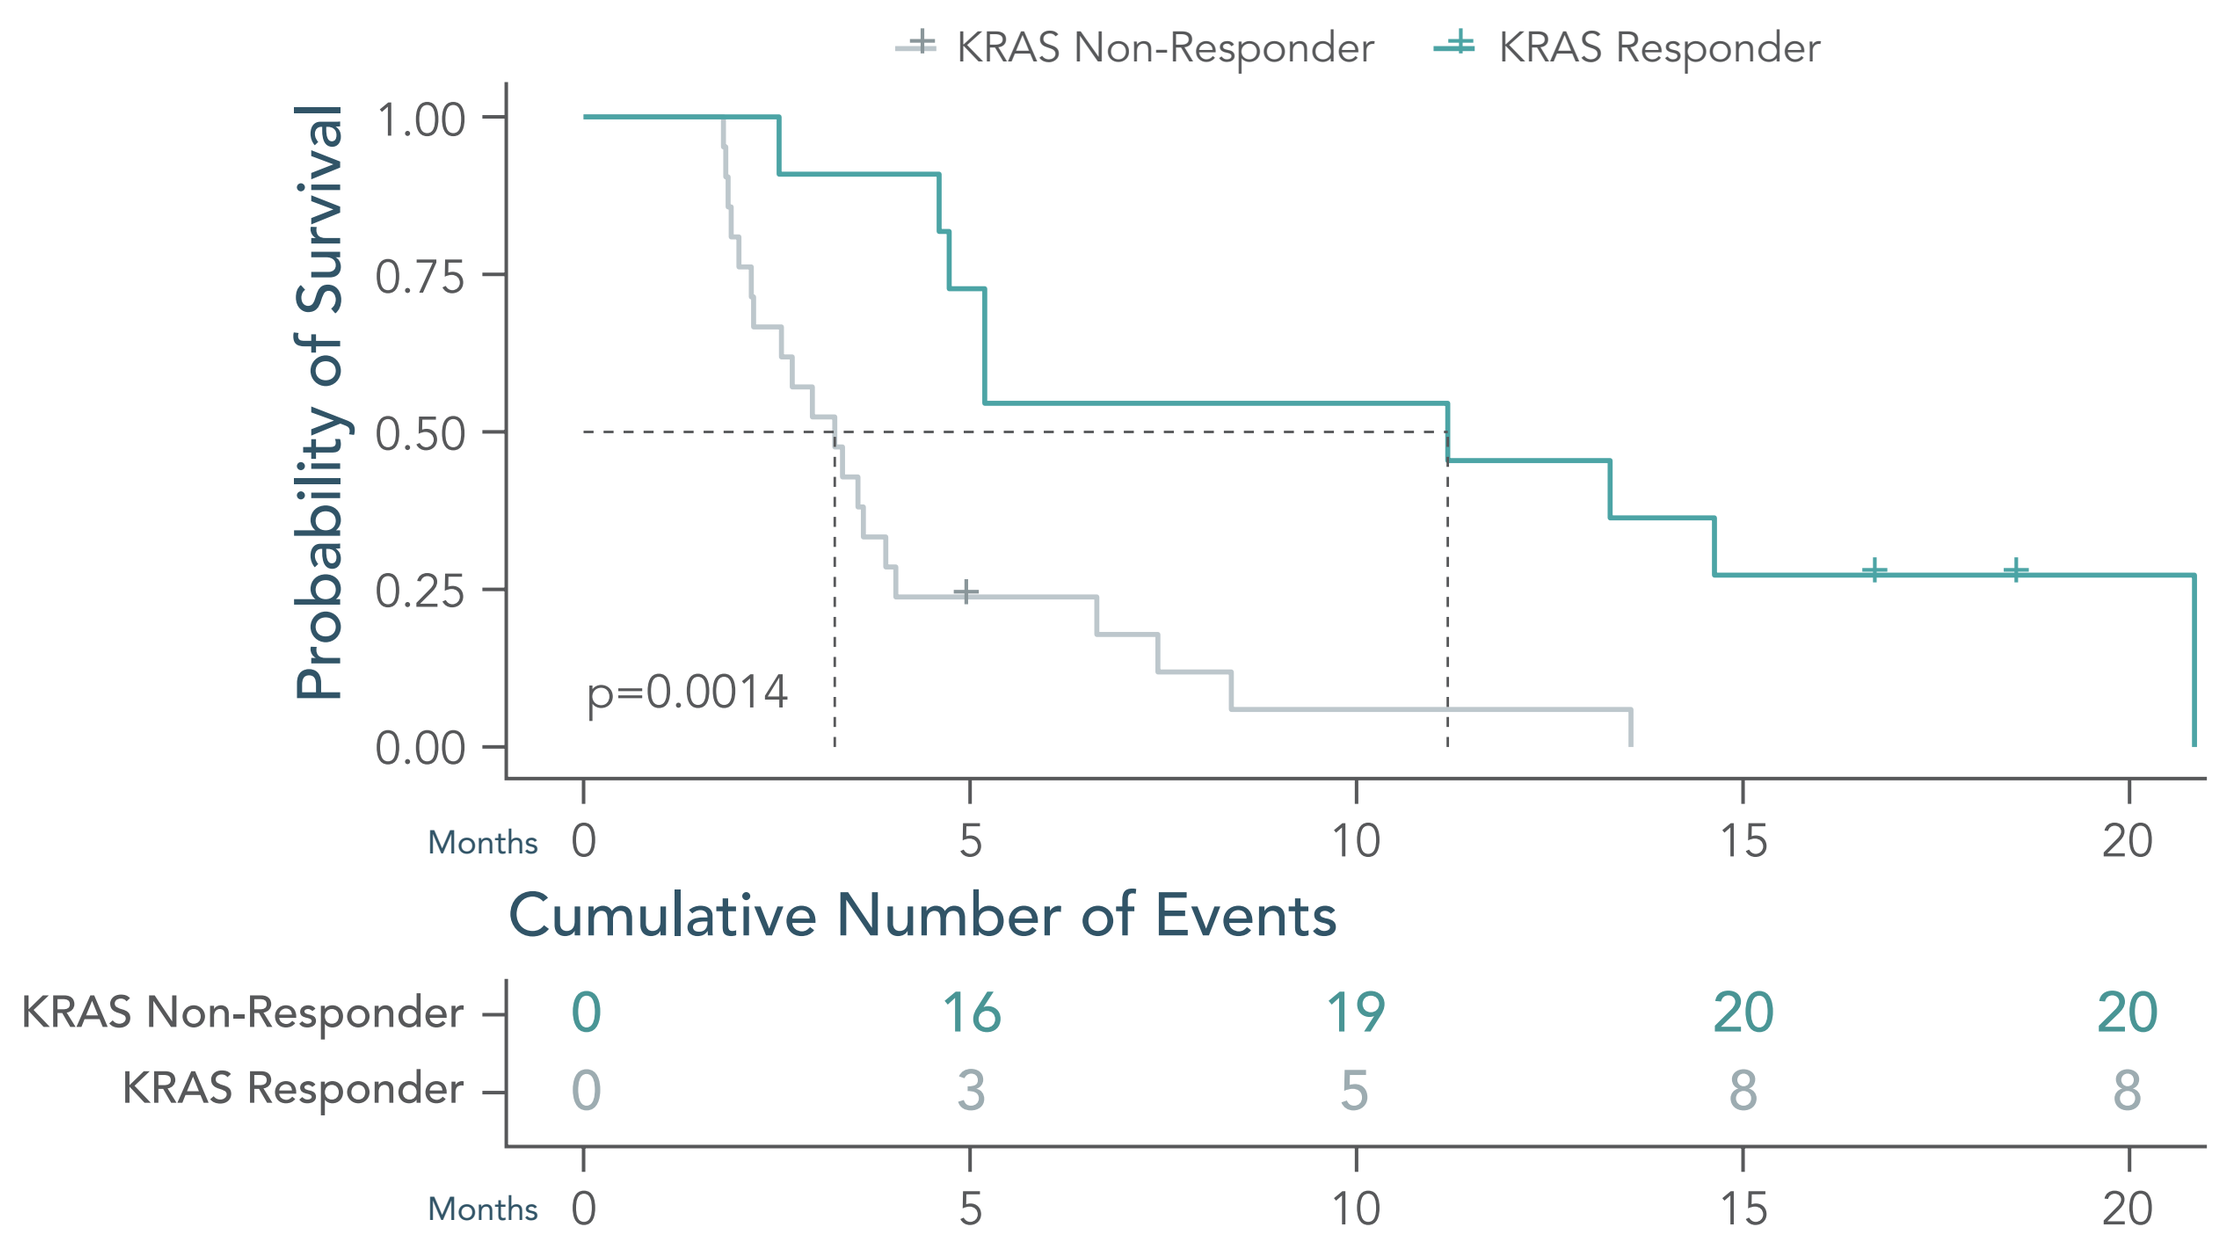

KRAS Non-Responder
KRAS Responder
Probability of Survival
1.00
0.75
0.50
0.25
0.00
p=0.0014
Months 0 5 10 15 20
Cumulative Number of Events
KRAS Non-Responder 0 16 19 20 20
KRAS Responder 0 3 5 8 8
Months 0 5 10 15 20

- Similarly, we tested whether changes in mean MAF using the NGS-based Guardant360® assay could predict clinical benefit of EAP patients with prior irinotecan.
- Molecular Responders were defined as patients with a ≥90% decrease in mean MAF after 1 treatment cycle.[1]
- 21 EAP patients with prior-irinotecan were evaluated for Molecular Response; 7 (33%) patients were identified as Molecular Responders.
- **Molecular Responders had significantly longer PFS and higher 6-month PFS than Molecular Non-responders (p=0.013)** (**Table 4**).

1. Lenz et al, ESMO 2022, poster 397P.

TABLE 4. EFFICACY OF ONVANSERTIB + FOLFIRI/BEV IN MOLECULAR RESPONDERS AND NON-RESPONDERS

	Molecular Responder	Molecular Non-Responder	All Patients Evaluated for Molecular Response	All Patients with Prior Irinotecan
Patients (n)	7 (33%)	14 (67%)	21	43
Median PFS [CI] (months)	13.28 [4.73-NR]	3.77 [3.35-7.43]	4.60 [3.55-13.3]	4.04 [2.96-8.38]
6-Month PFS [CI] (%)	57.1 [30.1-100.0]	21.4 [7.9-58.4]	33.3 [18.2-61.0]	37.3 [24.9-55.8]

Study Contact: Scott Kopetz MD PhD, SKopetz@mdanderson.org

Presented at: ESMO 2022, Poster Session 7