FPN: 397P

Early Decreases in KRAS Mutant Allele Frequency (MAF) Predict Clinical Benefit to the PLK1 Inhibitor Onvansertib in Combination with FOLFIRI/bev in 2L Treatment of Metastatic Colorectal Carcinoma (mCRC)

HJ Lenz MD[1], M Ridinger PhD[2], E Samuelsz[2], T Smeal PhD[2], DH Ahn DO[3]

[1]USC Norris Comprehensive Cancer Center, Los Angeles, CA; [2]Cardiff Oncology, San Diego, CA; [3]Mayo Clinic, Phoenix, AZ

Background

Effective Second-Line Treatment is Needed in KRAS-Mutated mCRC

- Second-line (2L) treatments (chemotherapy ± targeted agents) have a poor prognosis:
 - ORR: 5%-13%, PFS: ~5.7 months, OS: ~11.5 months.[1-4]
- KRAS is mutated in ~50% of CRC patients and, to-date, RAS-directed therapies have been unsuccessful. The majority of KRAS mutations are considered to be undruggable and the covalent inhibitors of KRAS G12C (representing 8% of KRAS mutations in CRC) have shown limited activity in CRC.
- Alternative strategies to inhibit KRAS include targeting synthetic lethal partners of mutant KRAS (i.e., proteins that are essential in KRAS-mutant but not wild-type cells).

Onvansertib: A Promising Therapeutic Option for KRAS-Mutated mCRC

- Onvansertib is an oral and highly-selective PLK1 inhibitor.
- PLK1, a key regulator of the cell cycle, is overexpressed in CRC and associated with adverse clinical features.[5]
- A genome-wide RNAi screen identified PLK1 inhibition to be synthetic lethal with mutant KRAS in CRC cells.[6]
- Onvansertib demonstrated potent anti-tumor activity as single agent and showed synergy in combination with irinotecan and with 5-FU in the HCT-116 KRAS-mutant CRC xenograft model.

1. Giessen et al., *Acta Oncologica* 2015, 54: 187-193; **2.** Cremolini et al., *Lancet Oncol* 2020, 21: 497–507; **3.** Antoniotti et al., Correspondence *Lancet Oncol* June 2020; **4.** Bennouna et al., *Lancet Oncol* 2013; 14: 29–37; **5.** Weichert et al., *World J Gastroenterol.* 2005; 11(36):5644-50; **6.** Luo et al, *Cell.* 2009; 137 835-48.

Phase 1b/2 Trial

NCT03829410

Key Eligibility Criteria

- mCRC with KRAS mutation determined in a CLIA-certified lab.
- Failure of or intolerance to first line treatment of fluoropyrimidine and oxaliplatin with or without bevacizumab (bev).

Treatment (28-day cycle)

- FOLFIRI/bev, Days 1 and 15.
- Onvansertib, Days 1-5 and 15-19.

Efficacy Endpoints

- **Primary:**
 - Overall response rate (ORR) per RECIST v1.1.
- **Secondary:**
 - Progression-free survival (PFS).
 - Changes in circulating tumor DNA (ctDNA).

Correlative Studies

- Monitoring ctDNA changes by serial liquid biopsies has been shown to be a promising response biomarker to therapy in CRC and a potential tool to guide patient treatment.[1]
- **We aimed at assessing the association between early changes in ctDNA and clinical response (ORR/PFS) to onvansertib + FOLFIRI/bev.**
- Blood samples were collected at baseline (C1D1) and after 1 cycle of treatment (C2D1, ≈4 weeks) to measure changes in ctDNA (**Figure 1**):
 - KRAS mutant allele frequency (MAF) was assessed by digital droplet PCR (ddPCR).
 - Guardant360® assay, a NGS-based liquid biopsy panel covering 74 cancer genes, was used to measure the mean MAF of somatic SNVs, insertions/deletions, and gene fusions.[2]

1. Reece et al., *Front Genet.* 2019, 10:1118; **2.** Mak et al., *Cancer Res*, 2021, 81 (13_suppl).

FIGURE 1. WORKFLOW OF ctDNA ANALYSIS

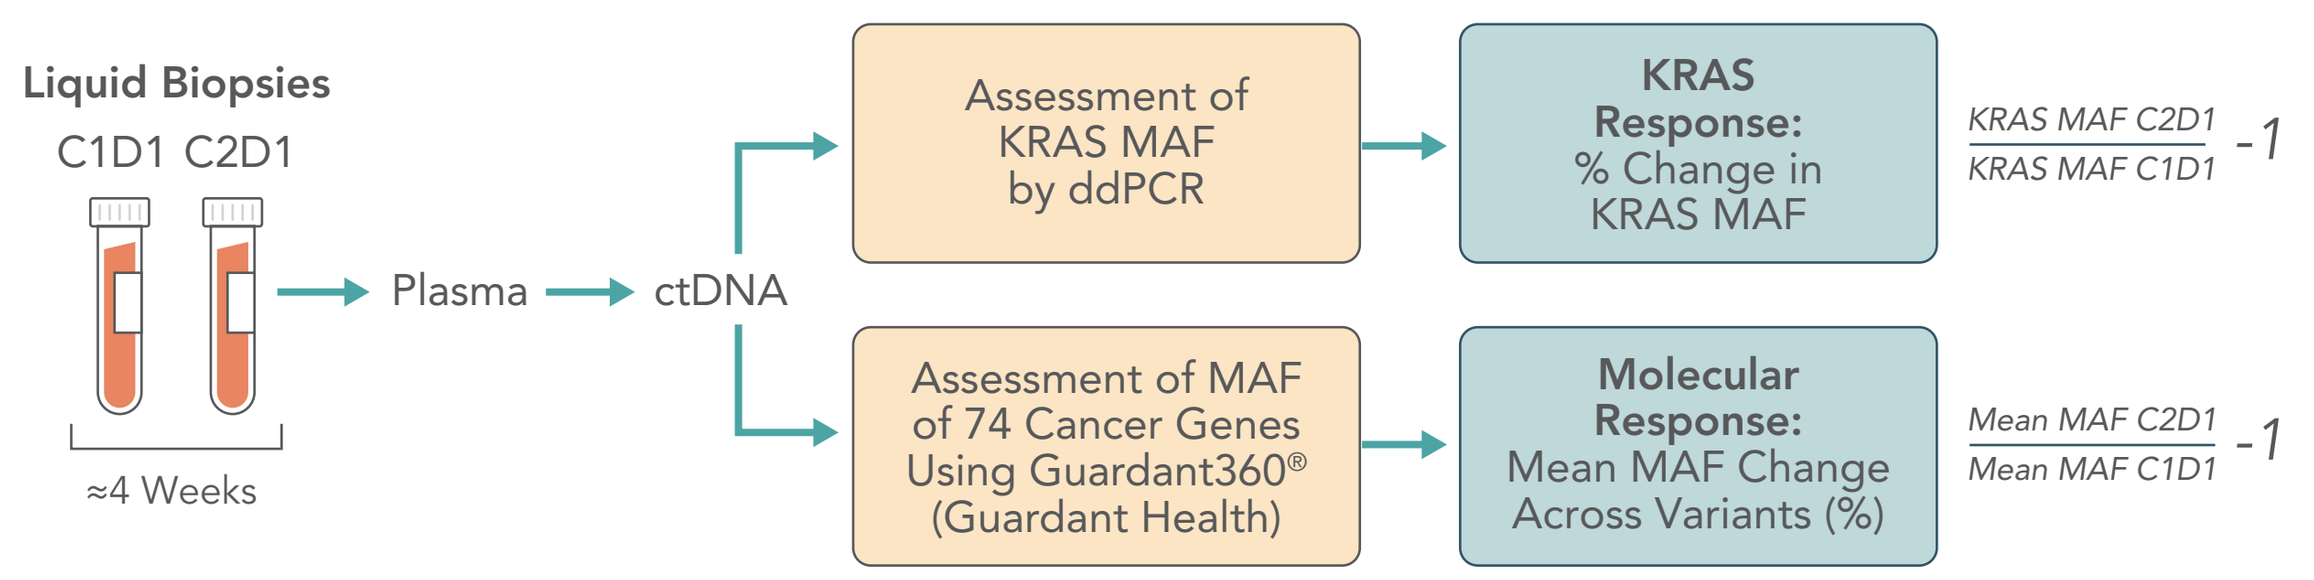

Liquid Biopsies
C1D1 C2D1
≈4 Weeks
Plasma
ctDNA
Assessment of KRAS MAF by ddPCR
KRAS Response: % Change in KRAS MAF
KRAS MAF C2D1 / KRAS MAF C1D1 - 1
Assessment of MAF of 74 Cancer Genes Using Guardant360® (Guardant Health)
Molecular Response: Mean MAF Change Across Variants (%)
Mean MAF C2D1 / Mean MAF C1D1 - 1

Changes in KRAS MAF: Best Threshold for Clinical Response Prediction

- At the cutoff date of 02-JUL-2021, changes in KRAS MAF were assessed in 29 patients.
- A Receiver-operating Characteristic (ROC) analysis was performed to determine the best threshold in % KRAS MAF change to predict clinical response (CR/PR) (**Figure 2**).
 - Of the 29 patients 12 had a PR, 15 SD and 2 PD as best response (ROC using 12 Responders vs 17 Non-responders).
- The best threshold in % KRAS MAF change to predict clinical response was determined to be **-90%**.
- Decrease in KRAS MAF of ≥90% after the 1st cycle of treatment predicted clinical response with 91.7% sensitivity and 82.4% specificity.

FIGURE 2. ROC CURVE FOR CLINICAL RESPONSE PREDICTION

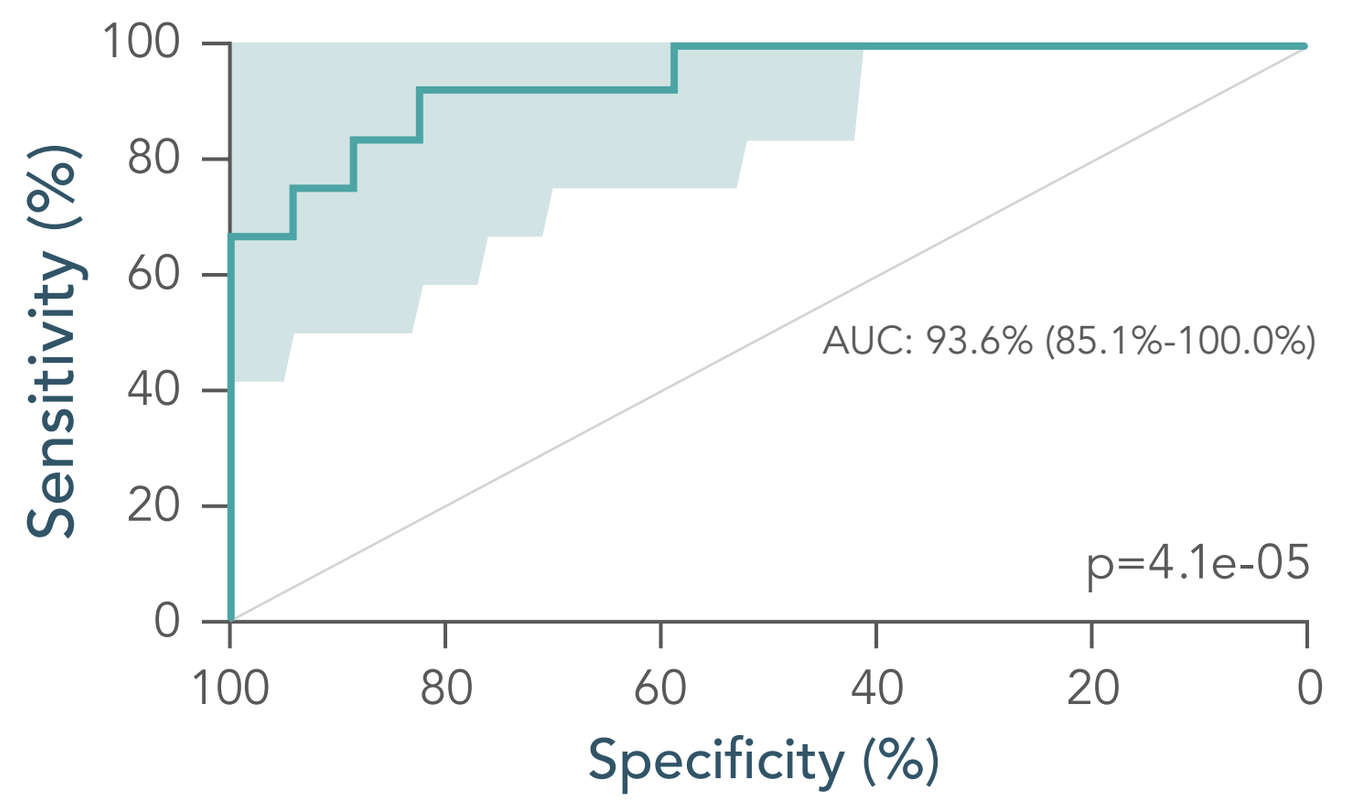

Sensitivity (%)
Specificity (%)
AUC: 93.6% (85.1%-100.0%)
p=4.1e-05

Results

Efficacy and KRAS Mutations

TABLE 1. EFFICACY OF ONVANSERTIB + FOLFIRI/BEV IN ALL PHASE 1B/2 PATIENTS (AS OF 25-JUL-2022)

Patients Treated (n)	Patients Evaluable for Efficacy[1] (n)	ORR (%)	Disease Control Rate[2] (%)	Median PFS [CI] (months)	Median Duration of Response[3] [CI] (months)
50	48	35.4	91.7	9.3 [7.6-13.5]	11.7 [8.9-NR]

1. Patients who received at least 1 cycle of treatment; **2.** CR+PR+SD; **3.** Defined as time between first response and progression; **NR:** Not reached; **CI:** 95% confidence intervals.

- Clinical responses (CR/PR) were observed across different KRAS variants (**Figure 3**).

FIGURE 3. KRAS MUTATIONS AND CLINICAL RESPONSE

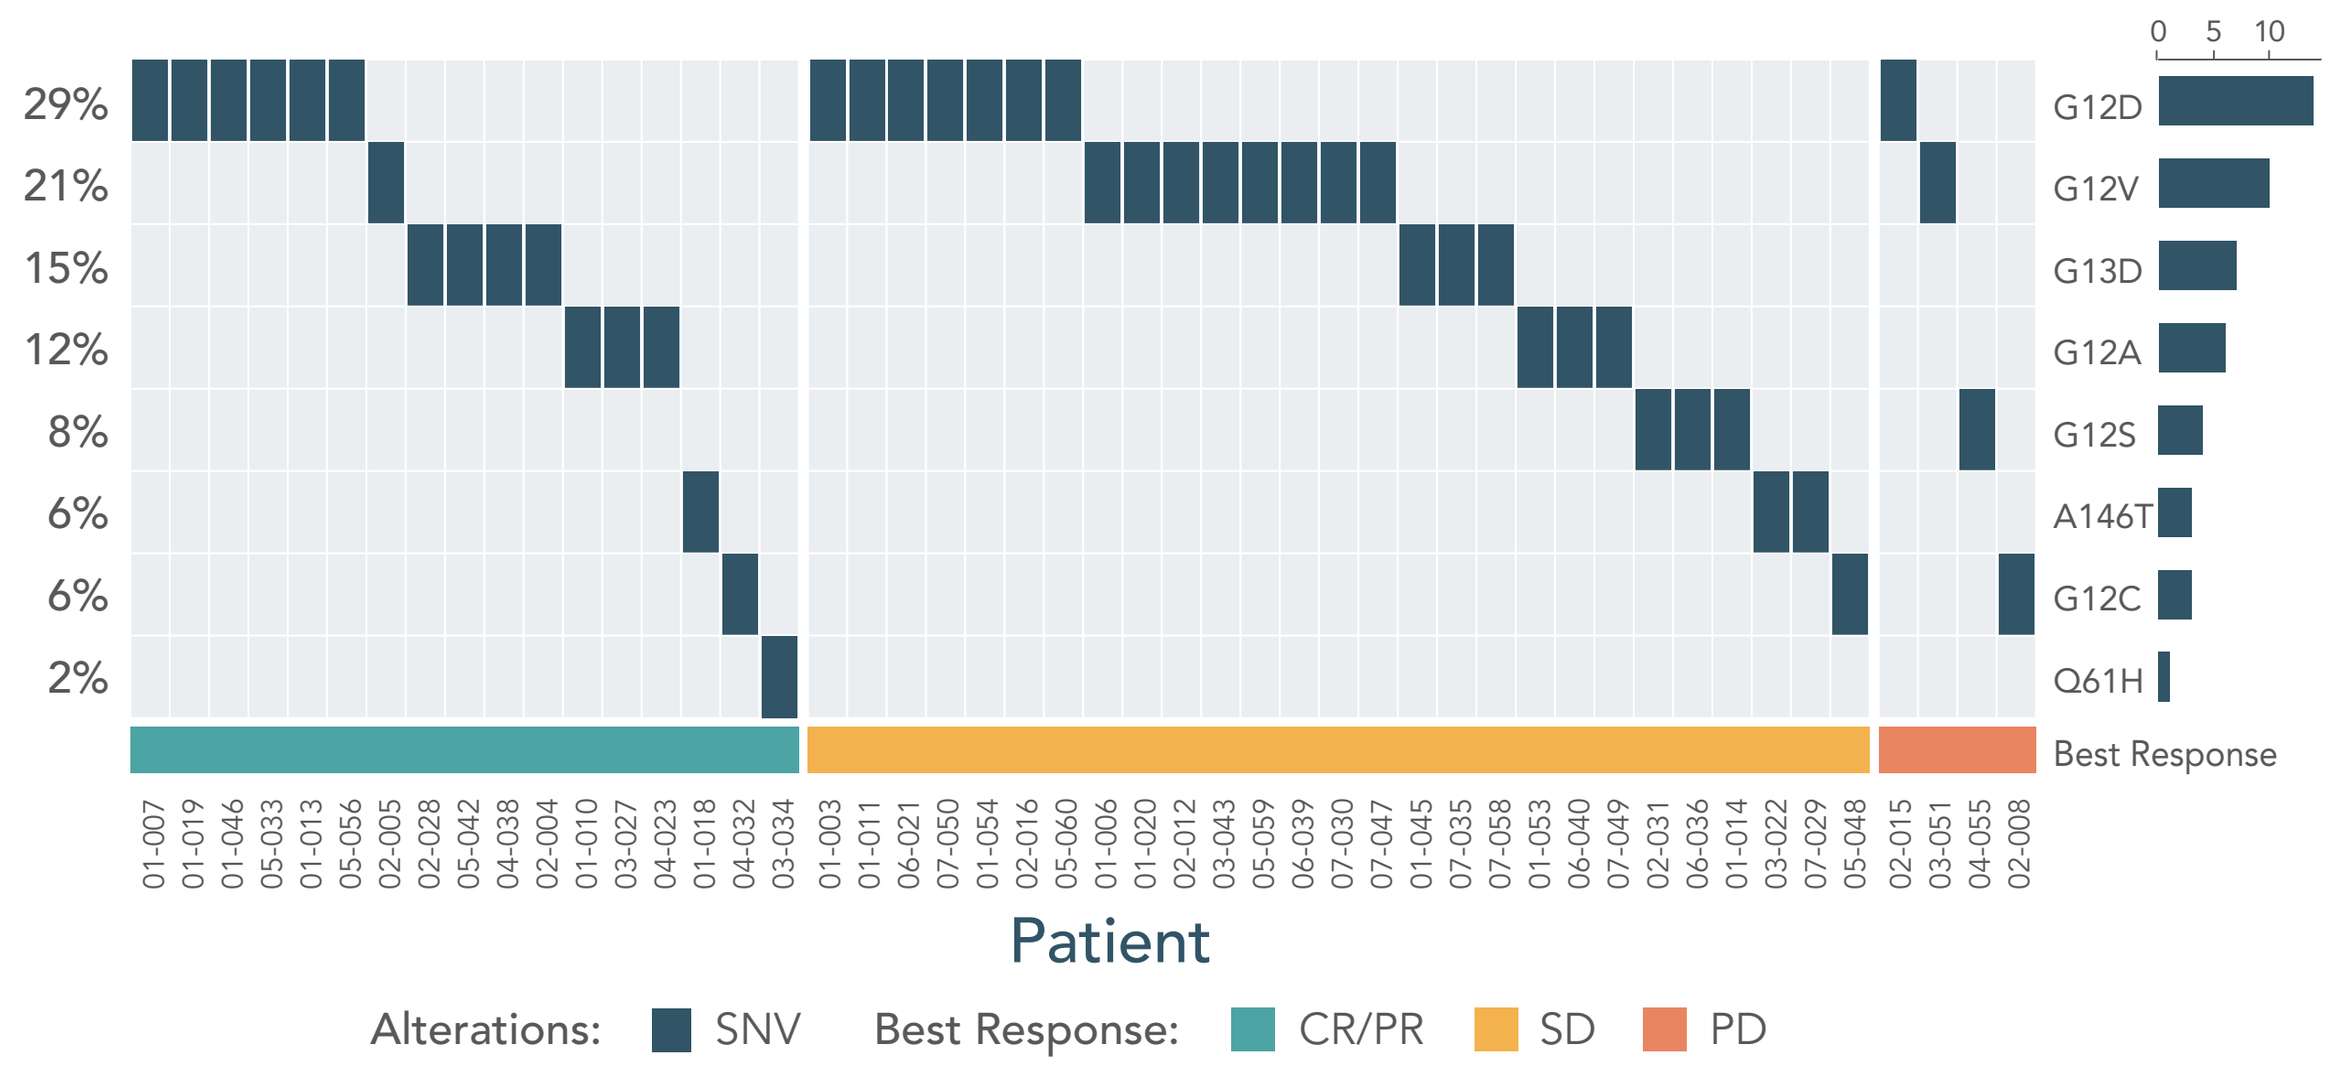

29% G12D
21% G12V
15% G13D
12% G12A
8% G12S
6% A146T
6% G12C
2% Q61H
Best Response
Patient
Alterations: SNV
Best Response: CR/PR SD PD

Early Changes in KRAS MAF Predicts Clinical Response

- Based on the ROC analysis, **KRAS Responders were defined as patients with a ≥90% decrease in KRAS MAF** after 1 cycle of treatment (assessed by ddPCR).
- As of 25-JUL-2022, 45 patients were evaluable for KRAS Response[1] (including the 29 patients used for the ROC analysis).
- 22 (49%) patients were determined to be KRAS Responders.
- **KRAS Responders showed significantly higher ORR and longer PFS than KRAS Non-responders (Table 2; Figures 4-5):**
 - ORR of 63.6% versus 8.7% (OR=16.2 [CI 3.5-131.4], p=0.00014).
 - PFS of 12.6 months versus 6.0 months (p=0.019).

1. Defined as patients with C1D1 and C2D1 plasma samples and detectable baseline KRAS MAF.

TABLE 2. EFFICACY OF ONVANSERTIB + FOLFIRI/BEV IN KRAS RESPONDERS AND NON-RESPONDERS

	KRAS Responders	KRAS Non-Responders
Patients (n)	22 (49%)	23 (51%)
CR+PR (n)	14	2
ORR (%)	63.6	8.7
Median PFS [CI] (months)	12.6 [8.2-NR]	6.0 [3.7-10.9]

FIGURE 4. CHANGES IN KRAS MAF AND BEST RESPONSE

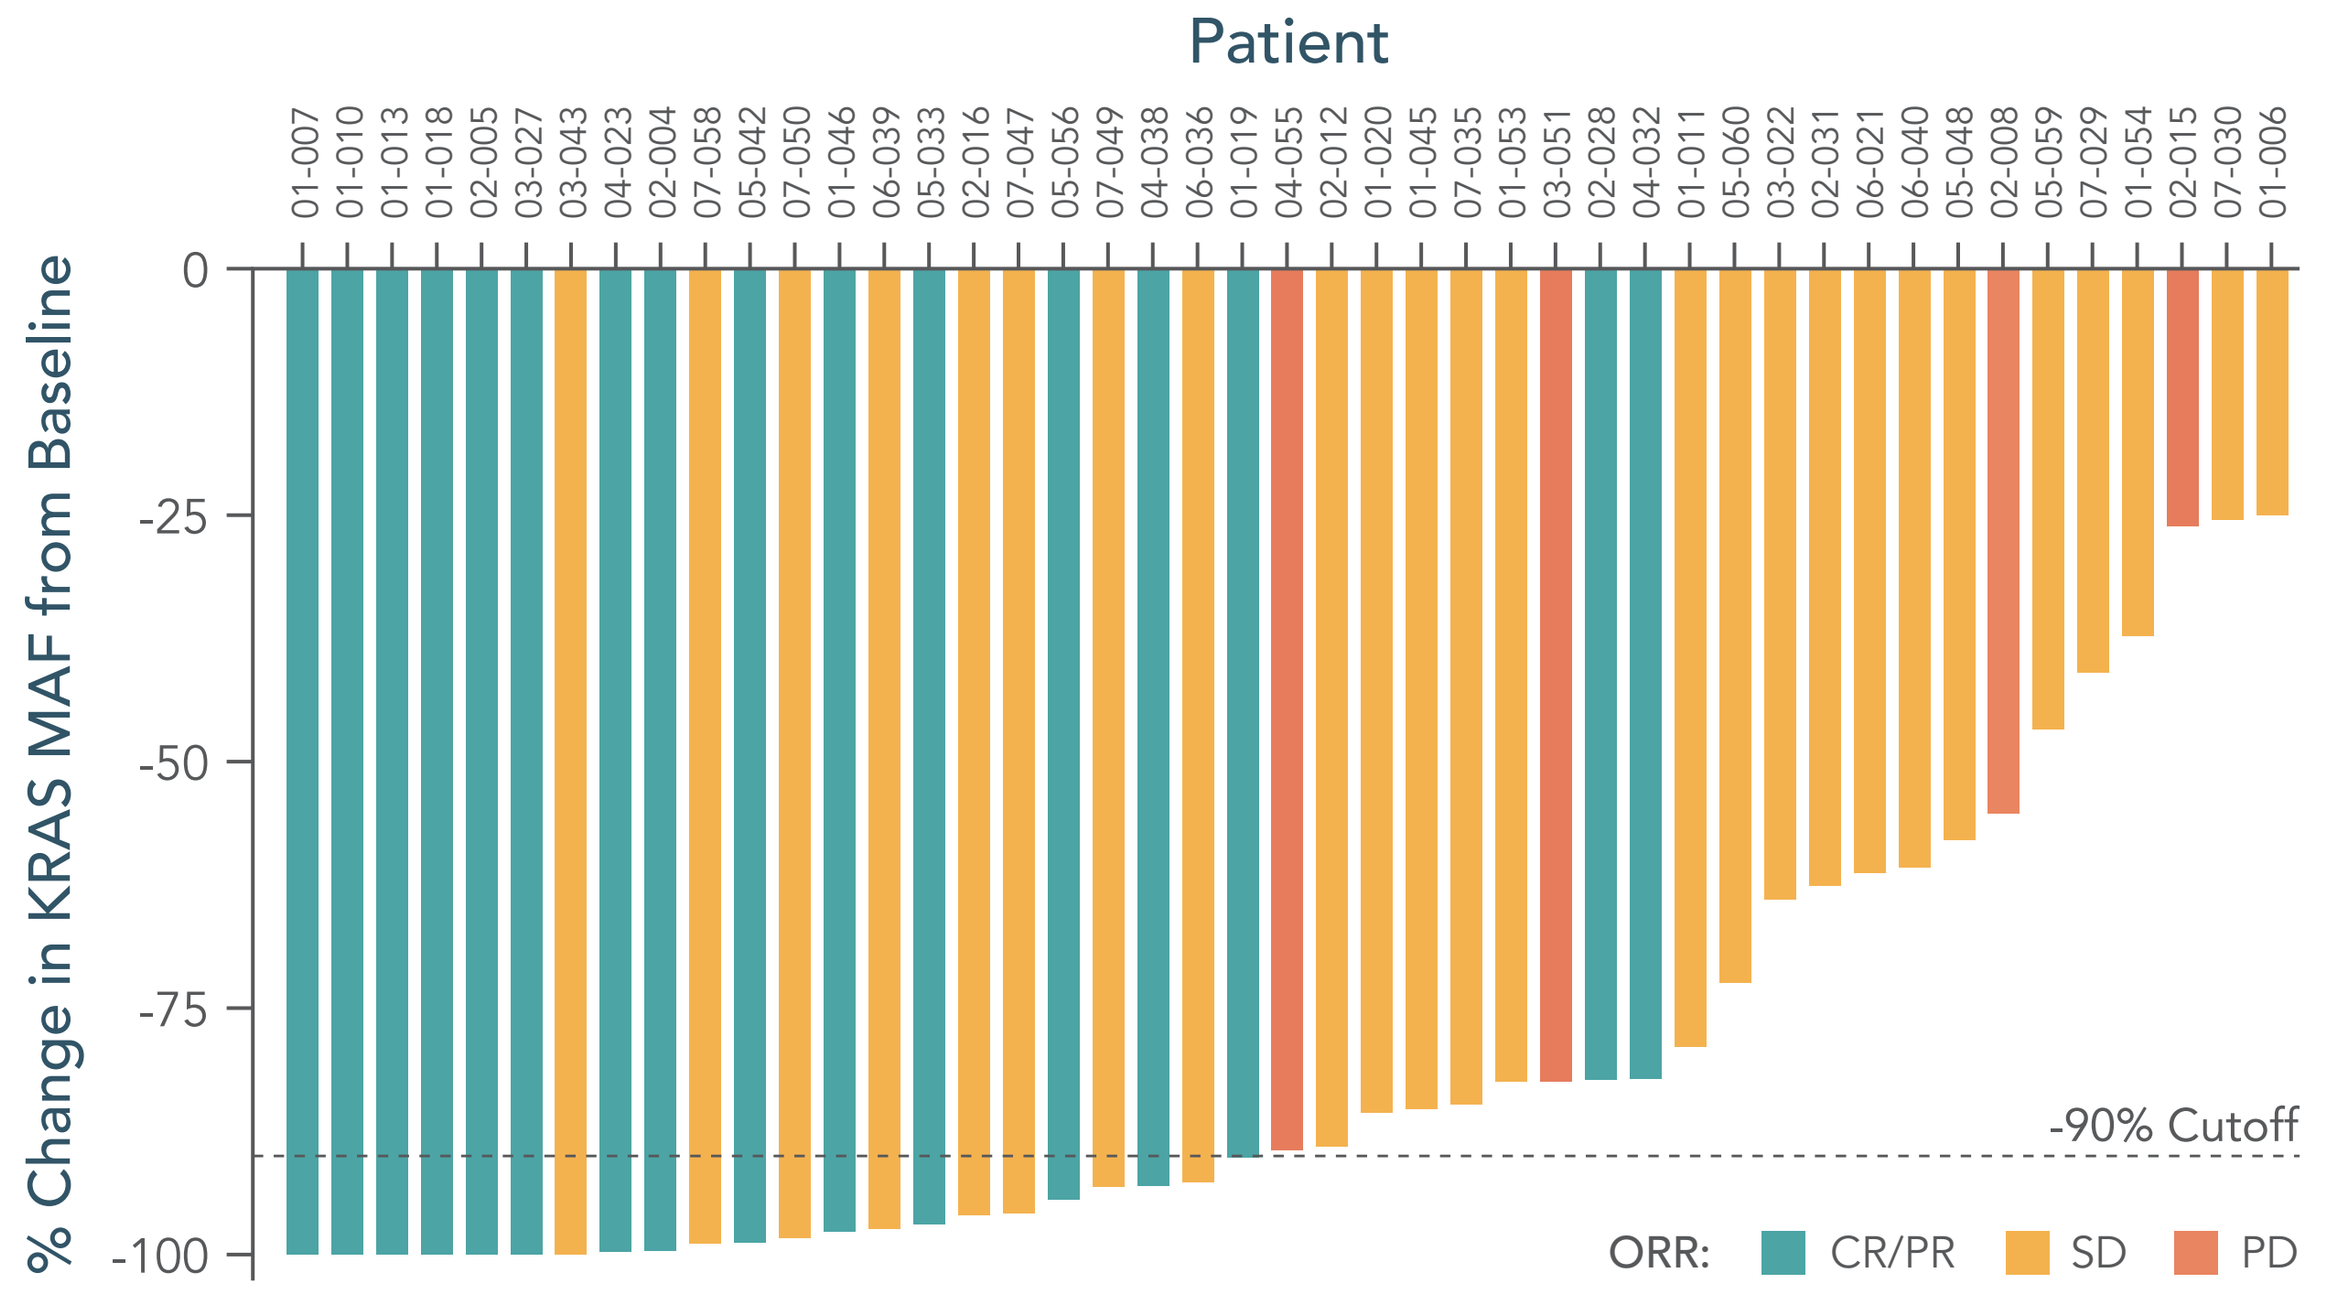

Patient
% Change in KRAS MAF from Baseline
0
-25
-50
-75
-100
-90% Cutoff
ORR: CR/PR SD PD

FIGURE 5. PFS OF KRAS RESPONDERS AND NON-RESPONDERS

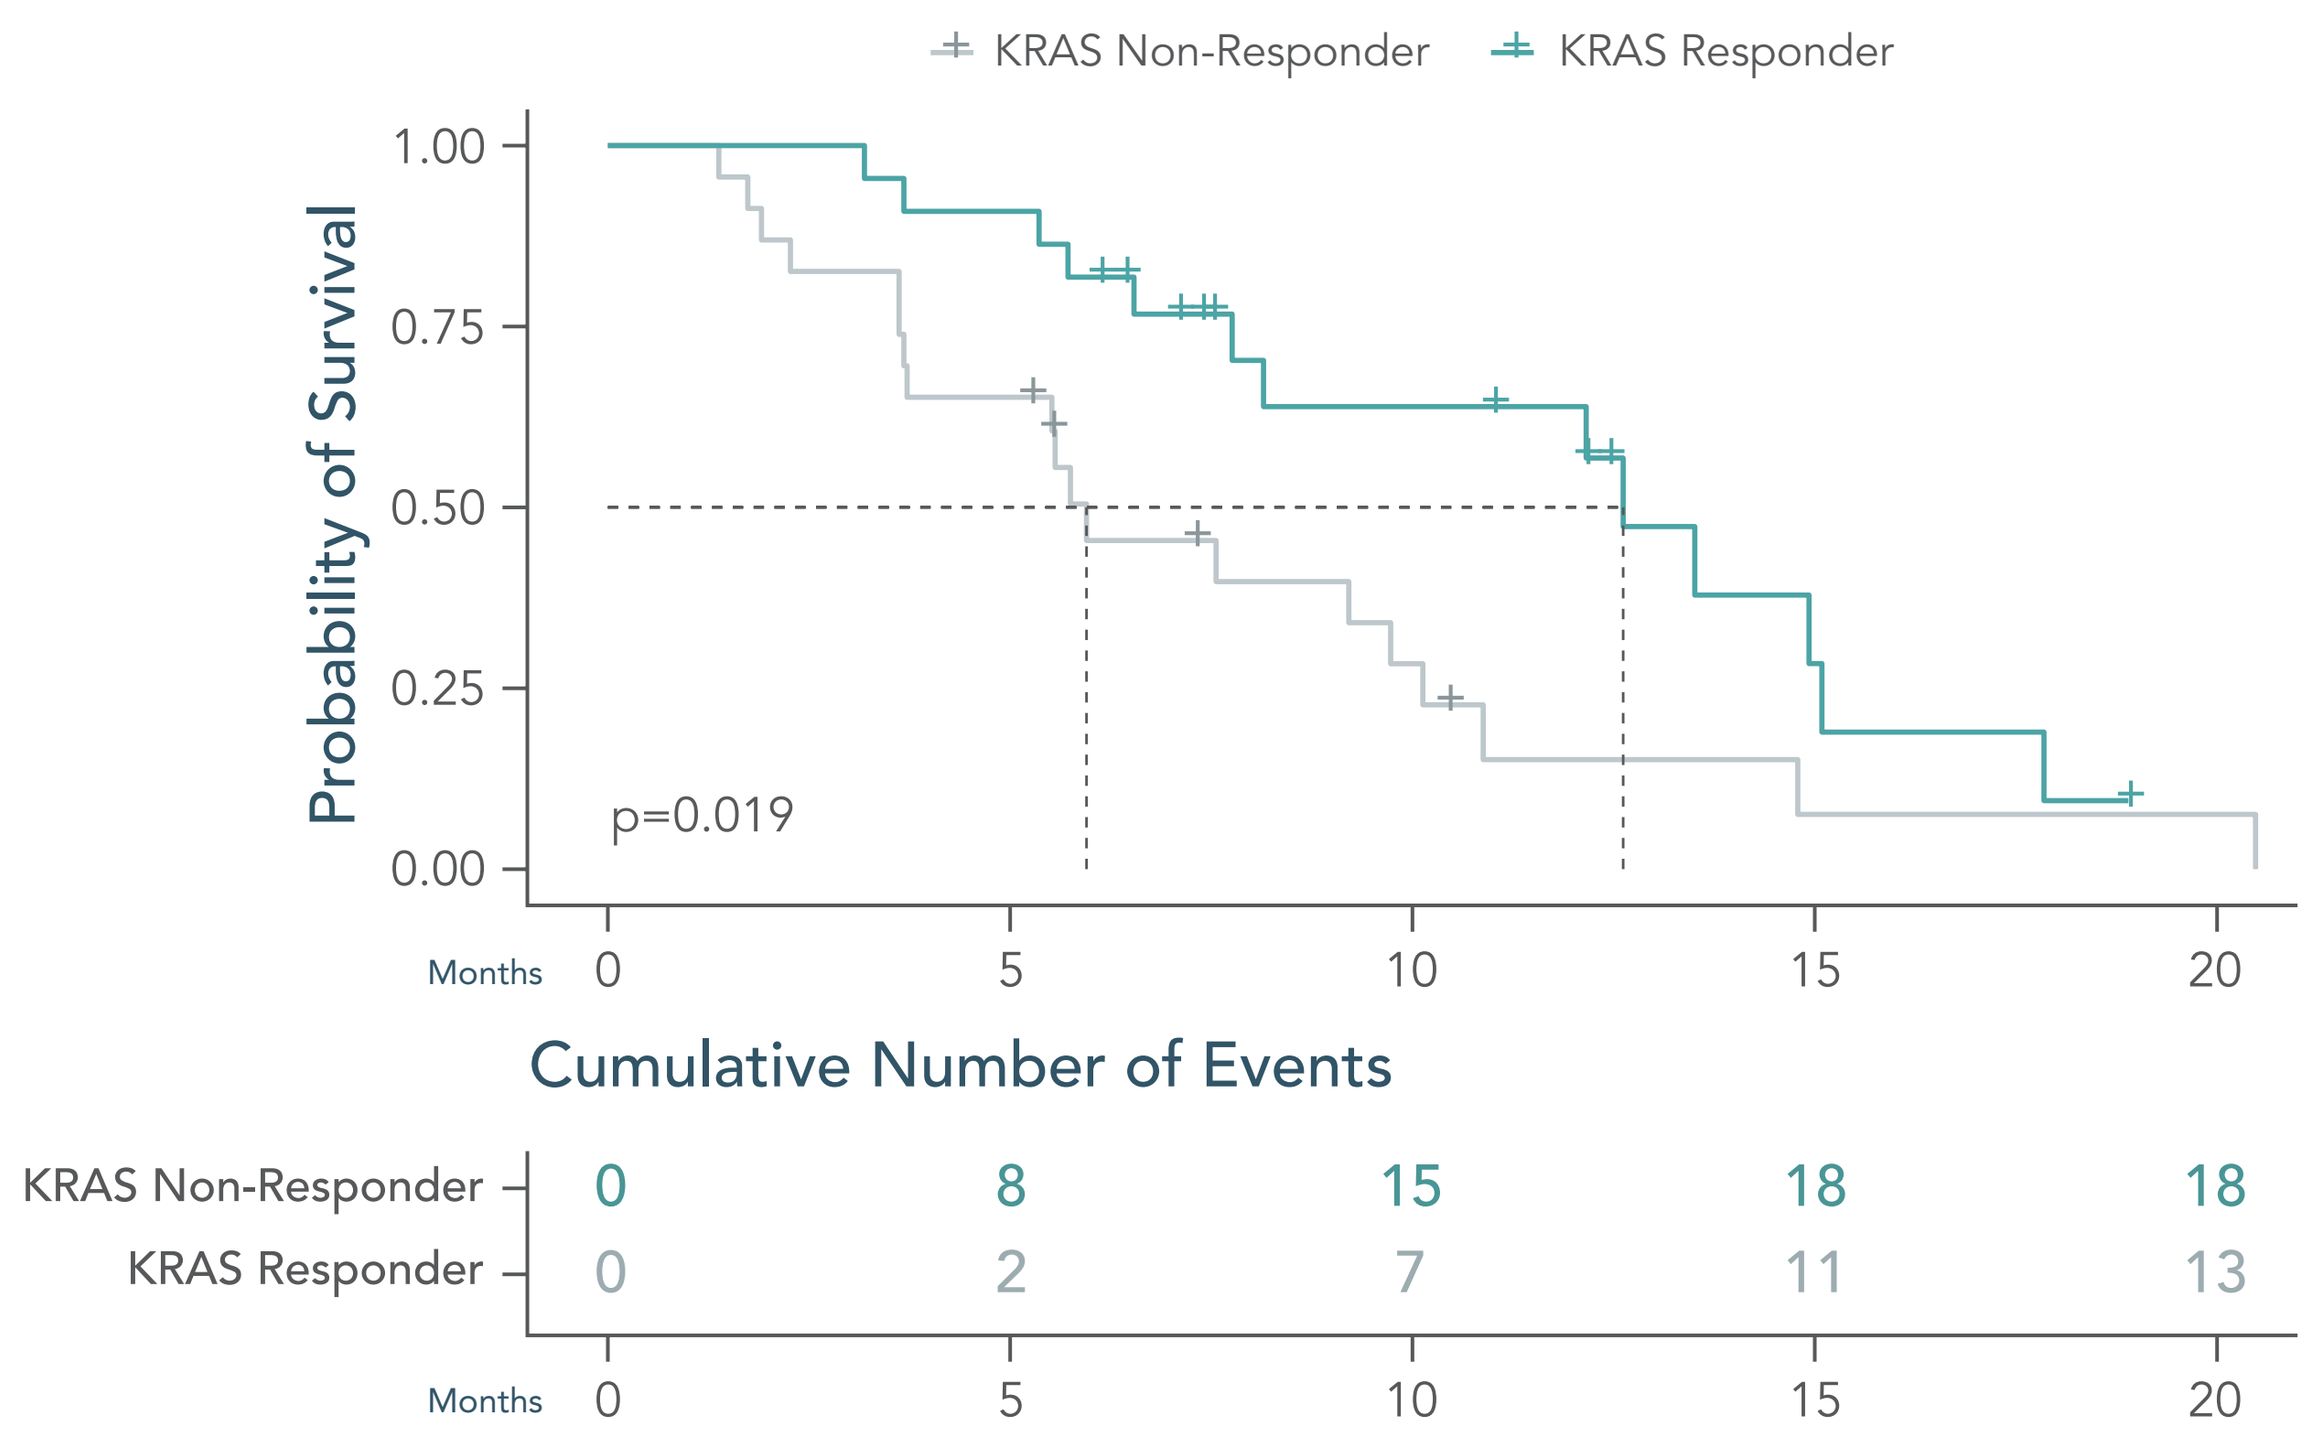

KRAS Non-Responder
KRAS Responder
Probability of Survival
p=0.019
Months 0 5 10 15 20
Cumulative Number of Events
KRAS Non-Responder 0 8 15 18 18
KRAS Responder 0 2 7 11 13

Changes in MAF Using Guardant360® Assay Predicts Clinical Response

- 29 patients of the Phase1b/2, with available C1D1 and C2D1 plasma samples, were evaluated for ctDNA changes using Guardant360®.
- **Molecular Responders were defined as patients with a ≥90% decrease in mean MAF** after 1 cycle of treatment.
- 16 (55%) patients were determined to be Molecular Responders.
- **Molecular Responders showed significantly higher ORR and longer PFS than Molecular Non-responders (Table 3):**
 - ORR of 56.3% versus 7.7% (OR=12.7 [CI 1.8-366.0], p=0.0082).
 - PFS of 12.2 months versus 5.6 (p=0.0015).

TABLE 3. EFFICACY OF ONVANSERTIB + FOLFIRI/BEV IN MOLECULAR RESPONDERS AND NON-RESPONDERS

	Molecular Responder	Molecular Non-Responder
Patients (n)	16 (55%)	13 (45%)
CR+PR (n)	9	1
ORR (%)	56.3	7.7
Median PFS [CI] (months)	12.2 [8.2-NR]	5.6 [3.6-NR]

Conclusions

- Onvansertib in combination with FOLFIRI/bev shows promising efficacy compared to historical control of FOLFIRI/bev for 2L KRAS mutant mCRC, with an ORR of 35% and PFS of 9.3 months as of 25-JUL-2022. DoR was 11.7 months, supporting no immediate acquired resistance.
- The efficacy of the combination was observed across KRAS variants.
- Stratification of patients based on MAF changes in ctDNA after 1 treatment cycle identified a subset of patients (~50%) with increased clinical benefit (ORR and PFS):
 - Patients with a ≥90% decrease in MAF of a single target (KRAS, ddPCR) or multi-genes (74-gene panel, NGS) had significantly higher ORR and longer PFS.
- This data supports the use of MAF changes in liquid biopsies as a response biomarker to onvansertib+FOLFIRI/bev in future clinical studies.

Study Contact: Heinz-Josef Lenz, MD, Lenz@USC.edu

Presented at: ESMO 2022, Poster Session 8