Abstract 1100: A phase 1b study of Plk1 inhibitor onvansertib in combination with paclitaxel in metastatic triple-negative breast cancer (mTNBC) patients.

Antonio Giordano[1], Qingchun Jin[1], Eileen Wrabel[1], Raechel Davis[1], Alexander Schubert[1], Michael Stalteri[1], Zachary Smolar[1], Sarah L. Sammons[1], Nancy U. Lin[1], Kristina Fanucci[1], Filipa Lynce[1], Adrienne Gropper Waks[1], Maya Ridinger[2], Erica L. Mayer[1], Fairooz Kabbinavar[2], Stephen P Ethier[3], Nabihah Tayob[1], Geoffrey I. Shapiro[1], Steven J. Isakoff[4], Sara M. Tolaney[1].

1. Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA; 2. Cardiff Oncology, San Diego, CA; 3. Medical University of South Carolina, Charleston, SC; 4. Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA

Background

Onvansertib is an oral polo-like kinase 1 (PLK1) ATP-competitive inhibitor with preclinical data showing synergy when combined with paclitaxel (P) in TNBC models. Here, we report safety and outcome data for subjects enrolled in a phase 1b clinical trial of onvansertib and paclitaxel for patients (pts) with mTNBC

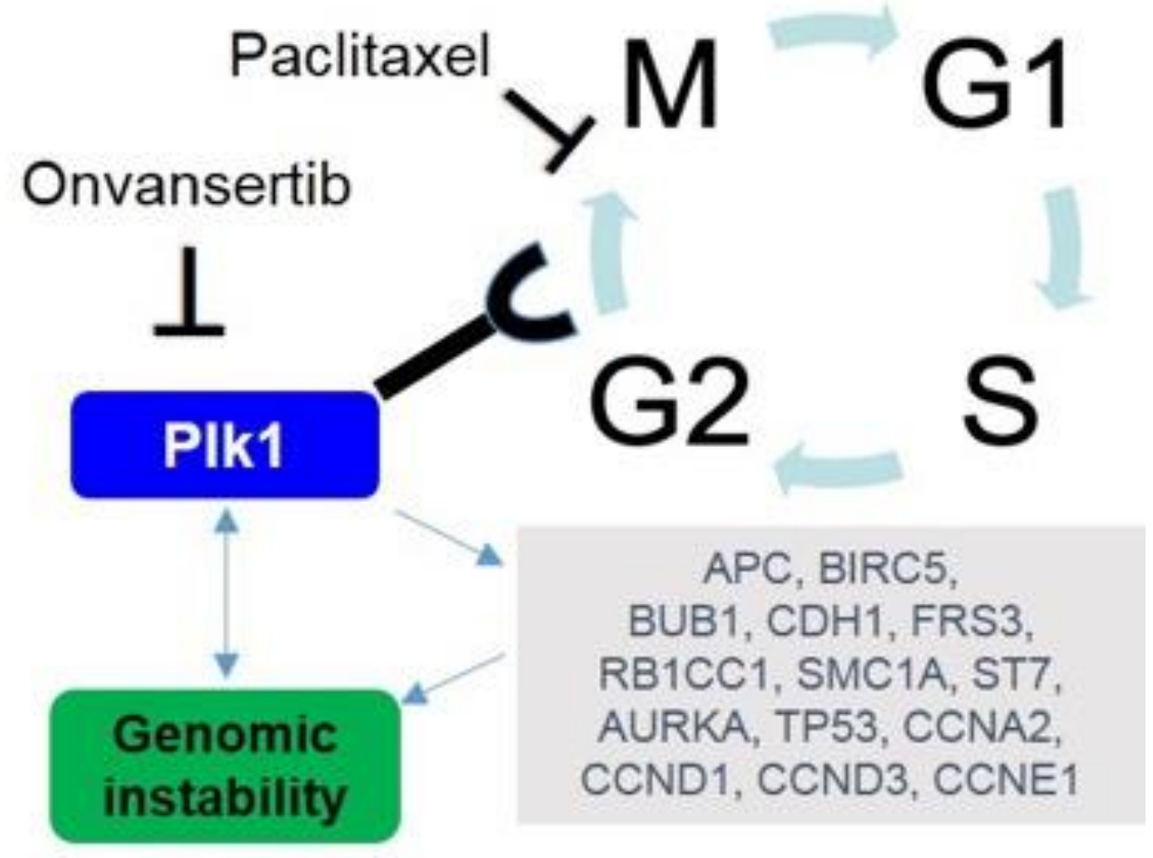
Paclitaxel
Onvansertib
M
G1
G2
S
Plk1
Genomic instability
APC, BIRC5, BUB1, CDH1, FRS3, RB1CC1, SMC1A, ST7, AURKA, TP53, CCNA2, CCND1, CCND3, CCNE1

This study tested a new drug combination for triple-negative breast cancer. We combined **onvansertib, an oral polo-like kinase 1 inhibitor, with weekly paclitaxel** to see if it was safe and effective. We found that the **combination was generally safe**, with some side effects like anemia and fatigue. The most effective **dose of onvansertib was 18 mg/m²** (corresponding to a flat dose of 30 mg). About 20% of patients treated at this dose saw their cancer shrink. The results are promising, suggesting further research is needed for this aggressive subtype of breast cancer.

Study Design

STUDY PRIMARY OBJECTIVES

- Safety, characterization of DLTs (End points: adverse event (AE) rate, DLT observation rate) of onvansertib plus paclitaxel
- Determination of recommended phase 2 dose (RP2D) of onvansertib plus paclitaxel

SECONDARY OBJECTIVES

- Describe Pharmacokinetics (PK) of onvansertib (End points: Cmax, AUC) in combination with paclitaxel
- ctDNA, PBMC, and baseline tissue NGS to predict response to paclitaxel plus onvansertib

Phase 1 Dose Escalation Plan – BOIN Design

- Patients enrolled in cohorts of 3 (max number of patients: 24)
- Target DLT rate: ≤30%

Table 1. Planned Dose Levels

Dose Level (DL)	Onvansertib Dose (QD)[A]	Paclitaxel Dose[B]
-1	6 mg/m^2	80 mg/m^2
0 (Starting DL)	9 mg/m^2	80 mg/m^2
1	12 mg/m^2	80 mg/m^2
2*	18 mg/m^2	80 mg/m^2

A. Onvansertib was given orally, once daily (QD) for 21 consecutive days, followed by 7 days off, cycle of 28 days.
B. Paclitaxel was given IV once on days 1, 8, and 15 of every 28-day cycle.
* DL2 was added with amendment 2 on 4/13/2023

Tissue collection

- Archival tissue was collected from prior biopsy, when available
- Blood for PBMCs and ctDNA was collected; PBMC collected at C1D1, C1D15, and at the off-treatment visit; ctDNA was collected on Cycle 1 Day 1, at the clinic visit immediately following restaging scans, and at the off-treatment visit.

- Blood samples for PK analysis was obtained during Cycle 1 on Days 1, 8, and 15, and Cycle 2 Day 1

Trial enrolled at Dana-Farber Cancer Institute and Massachusetts General Hospital from September 2022 to August 2024

Results

Table 1. Baseline Patient Characteristics

Characteristic	All (n=17)
	No. of Patients (%)
Age at registration, Years	
Median (range)	49 (35 – 71)
Race	
White	14 (82.4%)
Black or African-American	2 (11.8%)
Other	1 (5.9%)
ECOG PS at Baseline	
0	11 (64.7%)
1	6 (35.3%)
Disease-free interval from early breast cancer	
≤ 2 years	5 (29.4%)
> 2 years	8 (47.1%)
Stage IV disease at diagnosis	4 (23.5%)
Adjuvant or neoadjuvant chemotherapy	
Adjuvant or neoadjuvant anthracycline	12 (70.6%)
Adjuvant or neoadjuvant taxane	12 (70.6%)
Other Adjuvant or neoadjuvant therapy	10 (58.8%)
Lines of Chemotherapy in Metastatic setting	
1	3 (17.6%)
2	3 (17.6%)
>2	11 (64.7%)
Prior Taxane in metastatic setting	
Yes	3 (17.6%)
No	14 (82.4%)
Prior taxane in either setting	
Yes	14 (82.4%)
No	3 (17.6%)
Prior Immunotherapy	
Yes	7 (41.2%)
No	10 (58.8%)

Table 2. DLT Evaluable Patients

Dose cohort	DLT n (%)
9 mg/m2 (n = 3)	0 (0.0%)
12 mg/m2 (n = 3*)	0 (0.0%)
18 mg/m2 (n = 9*)	3 (30.0%)

*One patient treated at dose level 1 was not DLT evaluable and replaced;
*One patient treated at dose level 2 was not DLT evaluable and replaced

Table 3. Adverse Events (>20% and regardless of attribution to drugs)

	All Grade >= 2	Grade 2	Grade 3	Grade 4
Toxicity	**N (%)**	**N (%)**	**N (%)**	**N (%)**
Anemia	8 (47.1)	6 (35.3)	2 (11.8)	-
Neutrophil count decreased	8 (47.1)	4 (23.5)	1 (5.9)	3 (17.7)
Fatigue	5 (29.4)	4 (23.5)	1 (5.9)	-
Abdominal pain	4 (23.5)	3 (17.7)	1 (5.9)	-

Peripheral neuropathy was seen in 2 patients, both G2 (11.8%)

Median number of cycle was 2 (1-6); number of patients with dose delay 5 (29.4%) and dose reduction 7 (41.2%). One pt in DL2 remains on treatment, and 16 are off treatment (11 pts discontinued due to disease progression (PD) per RECIST 1.1; 3 due to clinical PD; 1 due to unacceptable toxicity; 1 death unrelated to the study drug).

Figure 1. Swimmer plot for all pts with best overall response

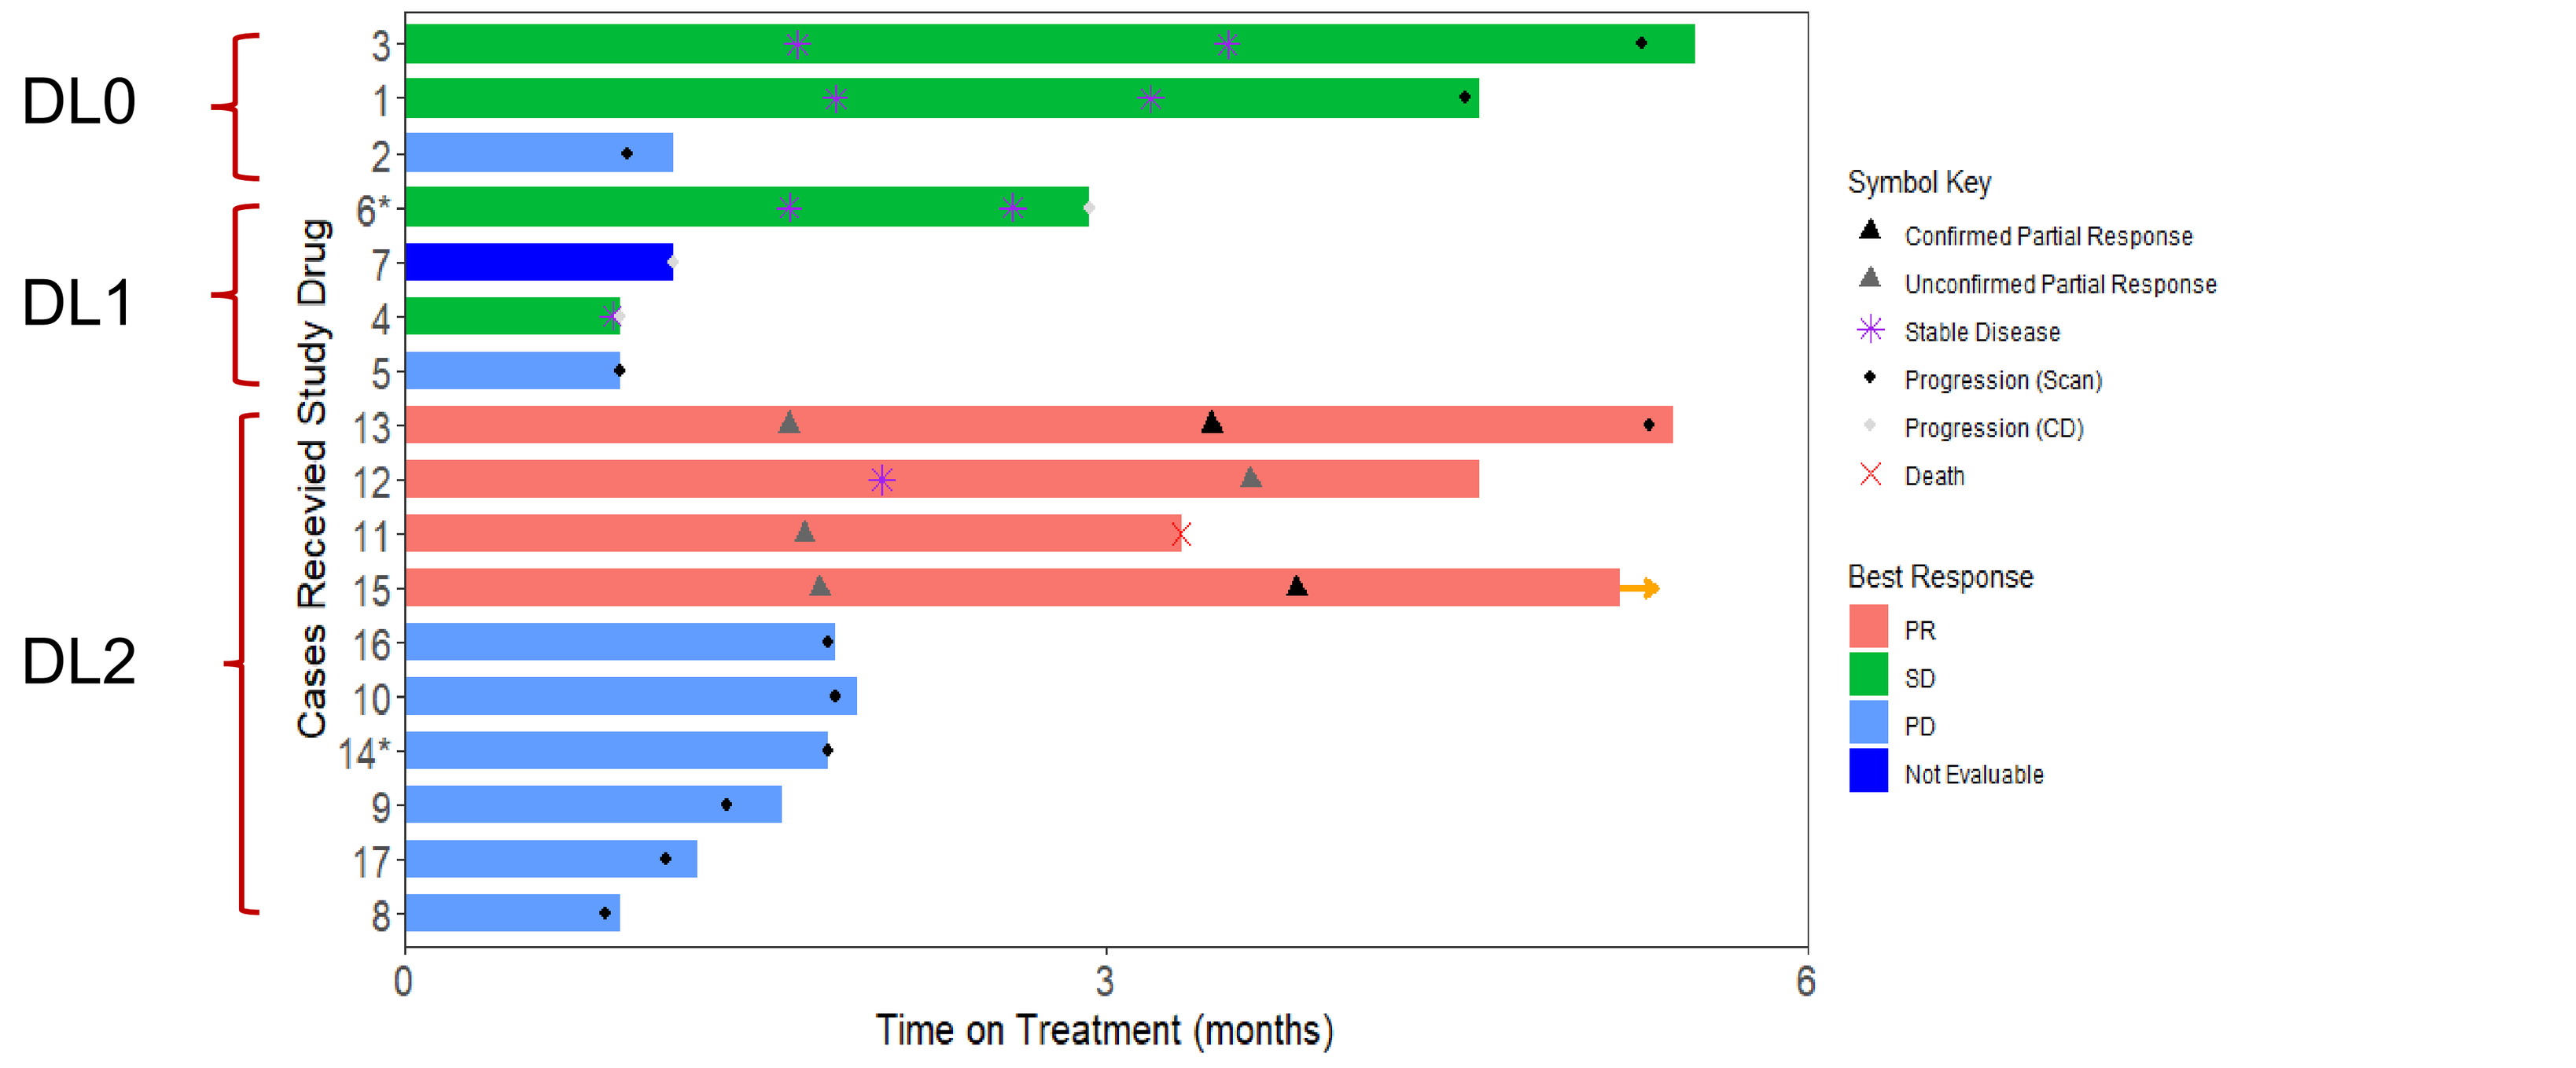
DL0
DL1
DL2
3
1
2
6*
7
4
5
13
12
11
15
16
10
14*
9
17
8
Cases Recevied Study Drug
0
3
6
Time on Treatment (months)
Symbol Key
Confirmed Partial Response
Unconfirmed Partial Response
Stable Disease
Progression (Scan)
Progression (CD)
Death
Best Response
PR
SD
PD
Not Evaluable

*Patient 6 at DL1 is not DLT evaluable, patient 14 at DL2 is not DLT evaluable
*Non evaluable patient 7 went off treatment because of clinical progression

- The RP2D of onvansertib in combination with paclitaxel is 18 mg/m² (corresponding to a flat dose of 30 mg). All 4 responders were treated in DL2 (18mg/m^2), 3/4 pts received prior P (2/4 in mTNBC setting) and IO (all in mTNBC), 2/4 received an ADC, 3/4 received < 3 lines of chemotherapy. In a total of 10 pts treated at RP2D, we report two (**20%**, case #13 and #15) **confirmed PR**, and two unconfirmed PR.

- Case #13 entered study after progression on 1st line paclitaxel plus pembrolizumab. Case #15 received paclitaxel in the adjuvant setting (3 years prior), one line in the metastatic setting including TDXd and durvalumab; patient is still enrolled and receiving treatment with resolution of target lesions (-100%)

Table 4. Summary Table of Best Response by RECIST 1.1

Response	N = 17 (%)
Partial Response (PR)	4 (23.5%)
Confirmed PR	2 (11.8%)
Unconfirmed PR	2 (11.8%)
SD ≥ 12 wks	2 (11.8%)
SD < 12 wks	2 (11.8%)
Progression of disease (PD)	
By RECIST 1.1	8 (47.1%)
Clinical PD	1 (5.9%)

Summary

- The combination of onvansertib and paclitaxel demonstrated a safe and manageable toxicity profile, with myelosuppression being the most common AE

- Toxicity, as well as activity, of the combination was dose dependent. Best responses were seen at the **RP2D 18 mg/m^2** (flat dose of 30 mg) **of onvansertib days 1-21 in combination with paclitaxel 80 mg/m^2 day 1,8,15 of a 28-day cycle**

- The results of our phase 1 trial showed activity of onvansertib plus paclitaxel in patients with TNBC. PKs and other biomarkers will be presented in the future. This combination warrants further exploration of the combination at the RP2D.

Acknowledgements

- NIH/NCI K12 Paul Calabresi Career Development for Clinical & Translational Oncology Program, K12 CA157688
- METAvivor Early Career Investigator Award
- Cardiff Oncology (San Diego, CA)

Contact: antonio_giordano@dfci.harvard.edu 𝕏 @antgiorda